PPARδ modulation improves the bioenergetic defect in mitochondrial myopathy

Eric Bell[1], Peter Dwyer[1], Robert Shine[1], Lyndsay Olson[1], Erzsebet Polyak[2], Yi Cheng[2], Julian Ostrovksy[2], Marni J. Falk[2, 3], Robert Taylor[4], Dominique Stickens[1], Effie Tozzo[1]

[1] Mitobridge, Inc, 1030 Massachusetts Avenue, Cambridge MA 02138; [2] Division of Human Genetics, Department of Pediatrics, The Children's Hospital of Philadelphia, Philadelphia, PA 19104, USA; [3] Department of Pediatrics, University of Pennsylvania Perelman School of Medicine, Philadelphia, PA, 19104 USA; [4] Welcome Trust Centre for Mitochondrial Research Newcastle University, Newcastle UK

Abstract

Mitochondrial myopathies are a heterogeneous group of neuromuscular diseases. Despite extensive variation in the clinical phenotype, skeletal muscle weakness and fatigue are common hallmarks of mitochondrial myopathies leading to reduced mobility and endurance. Skeletal muscle, which accounts for much of the body's energy consumption, generally oxidizes both fats and carbohydrates; however, in periods of prolonged activity fat becomes the primary source of energy. Fatty acid oxidation capacity may be impaired in primary mitochondrial respiratory chain diseases. Here, we evaluated whether fibroblasts from mitochondrial myopathy human subjects, with either a mutation in the *MT-TL1* gene (MELAS), 5 Kb common-deletion in Kearns-Sayre syndrome (KSS), or mutations in *MT-ND4* and *MT-ND6* genes (Leigh/LHON) effectively utilized fat as a substrate to support oxidative phosphorylation. Both MELAS and KSS lines exhibited markedly reduced mitochondrial respiration when fatty acids were used as the carbon source. Application of a novel modulator of the PPARδ nuclear receptor (MA-0211 aka MTB-1, ASP0367) to MELAS, KSS, and Leigh/LHON fibroblasts increased the expression of genes that regulate fatty acid oxidation. MA-0211 also increased mitochondrial respiratory capacity when fatty acids were used as the carbon source. Moreover, treating aged, diet-induced obese mice with MA-0211 significantly decreased their fatigue in an exercise endurance test. Together these results suggest that increasing fatty acid-mediated respiration may offer a valuable therapeutic approach to relieve fatigue and increase endurance in mitochondrial myopathy.

Introduction

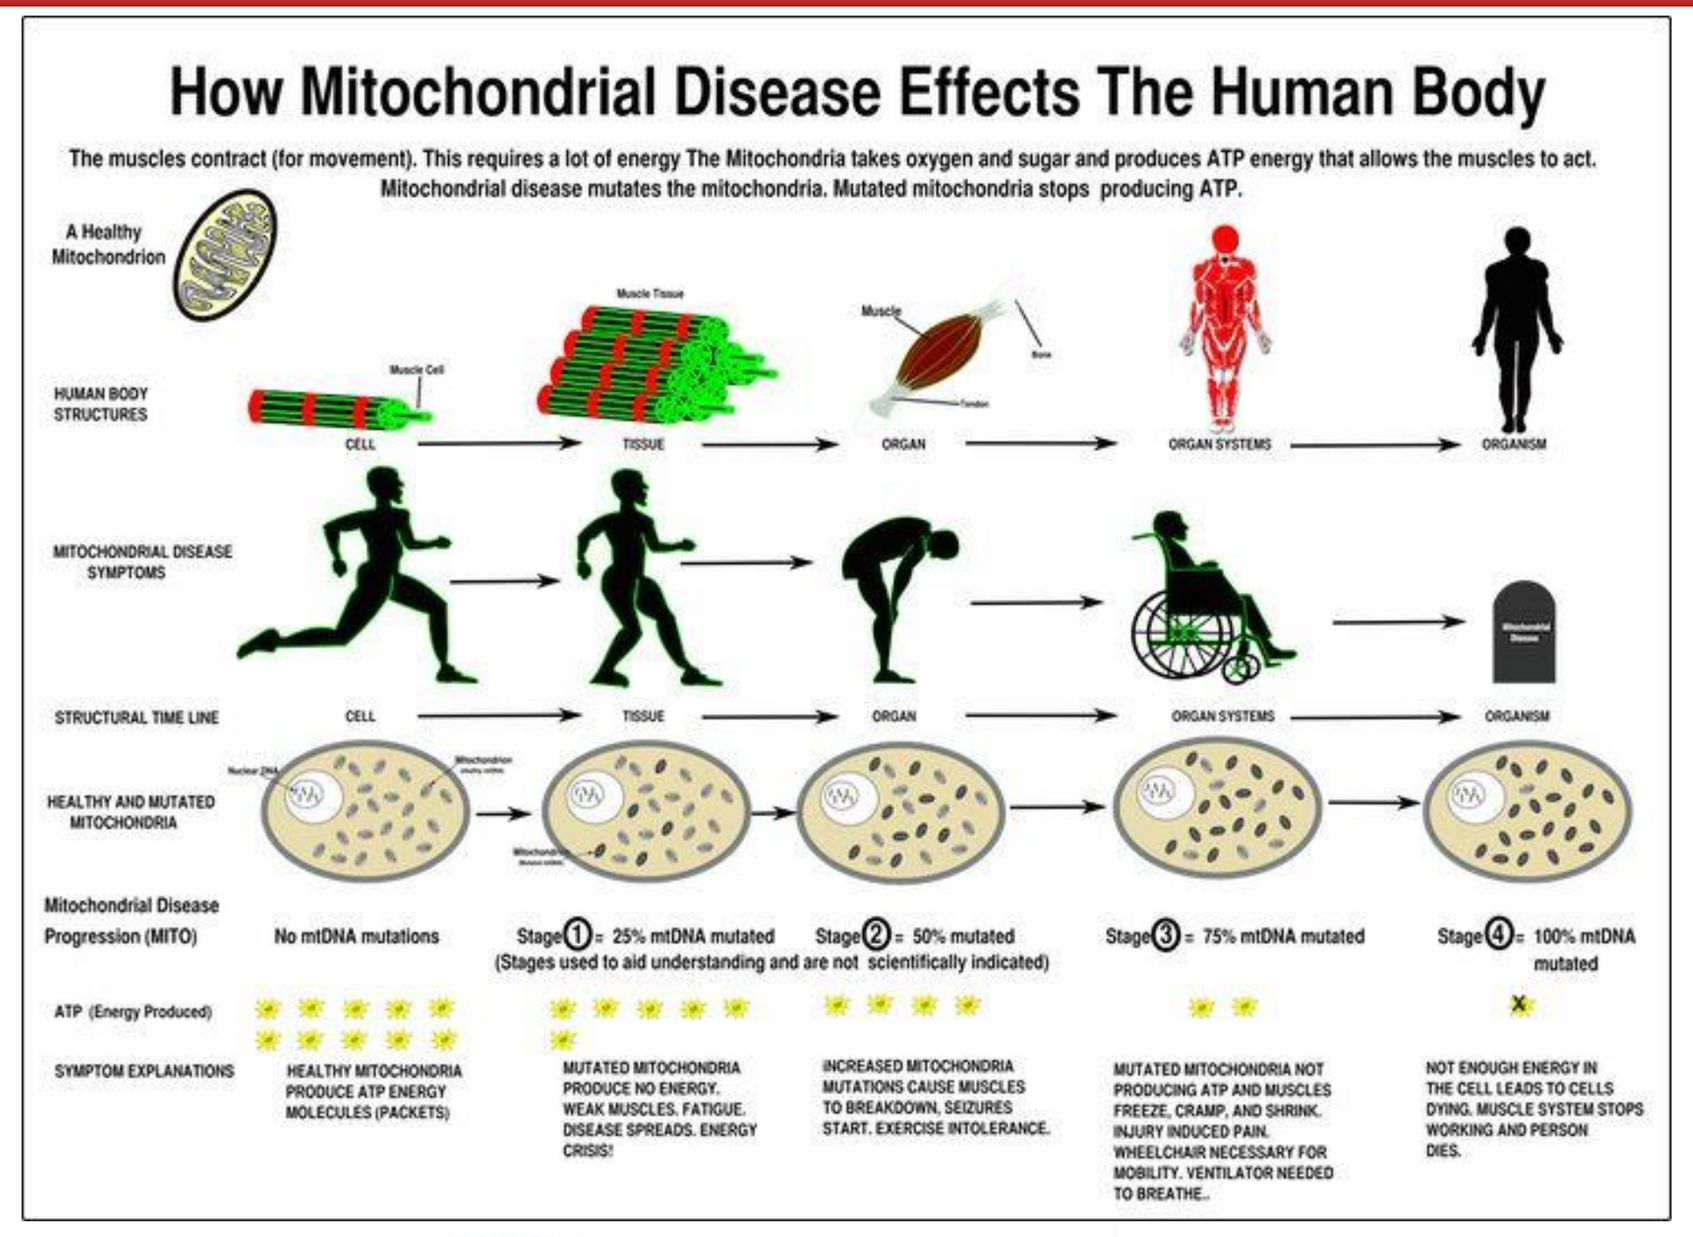
How Mitochondrial Disease Effects The Human Body
The muscles contract (for movement). This requires a lot of energy The Mitochondria takes oxygen and sugar and produces ATP energy that allows the muscles to act. Mitochondrial disease mutates the mitochondria. Mutated mitochondria stops producing ATP.

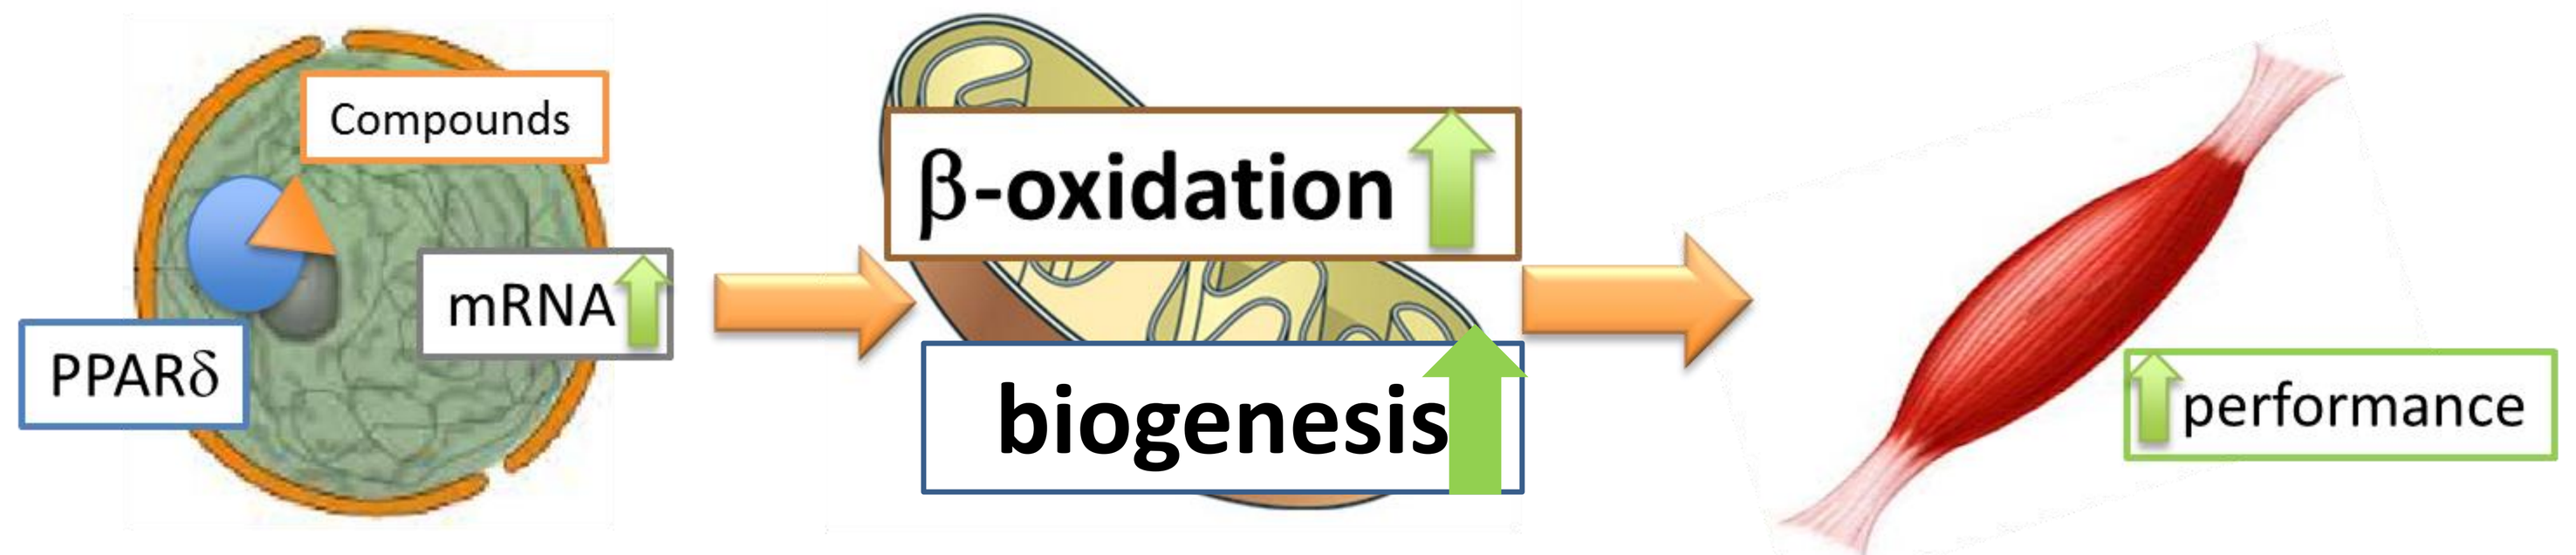
Compounds
PPARδ
mRNA
β-oxidation
biogenesis
performance

MA-0211: Leigh/LHON patient derived fibroblasts

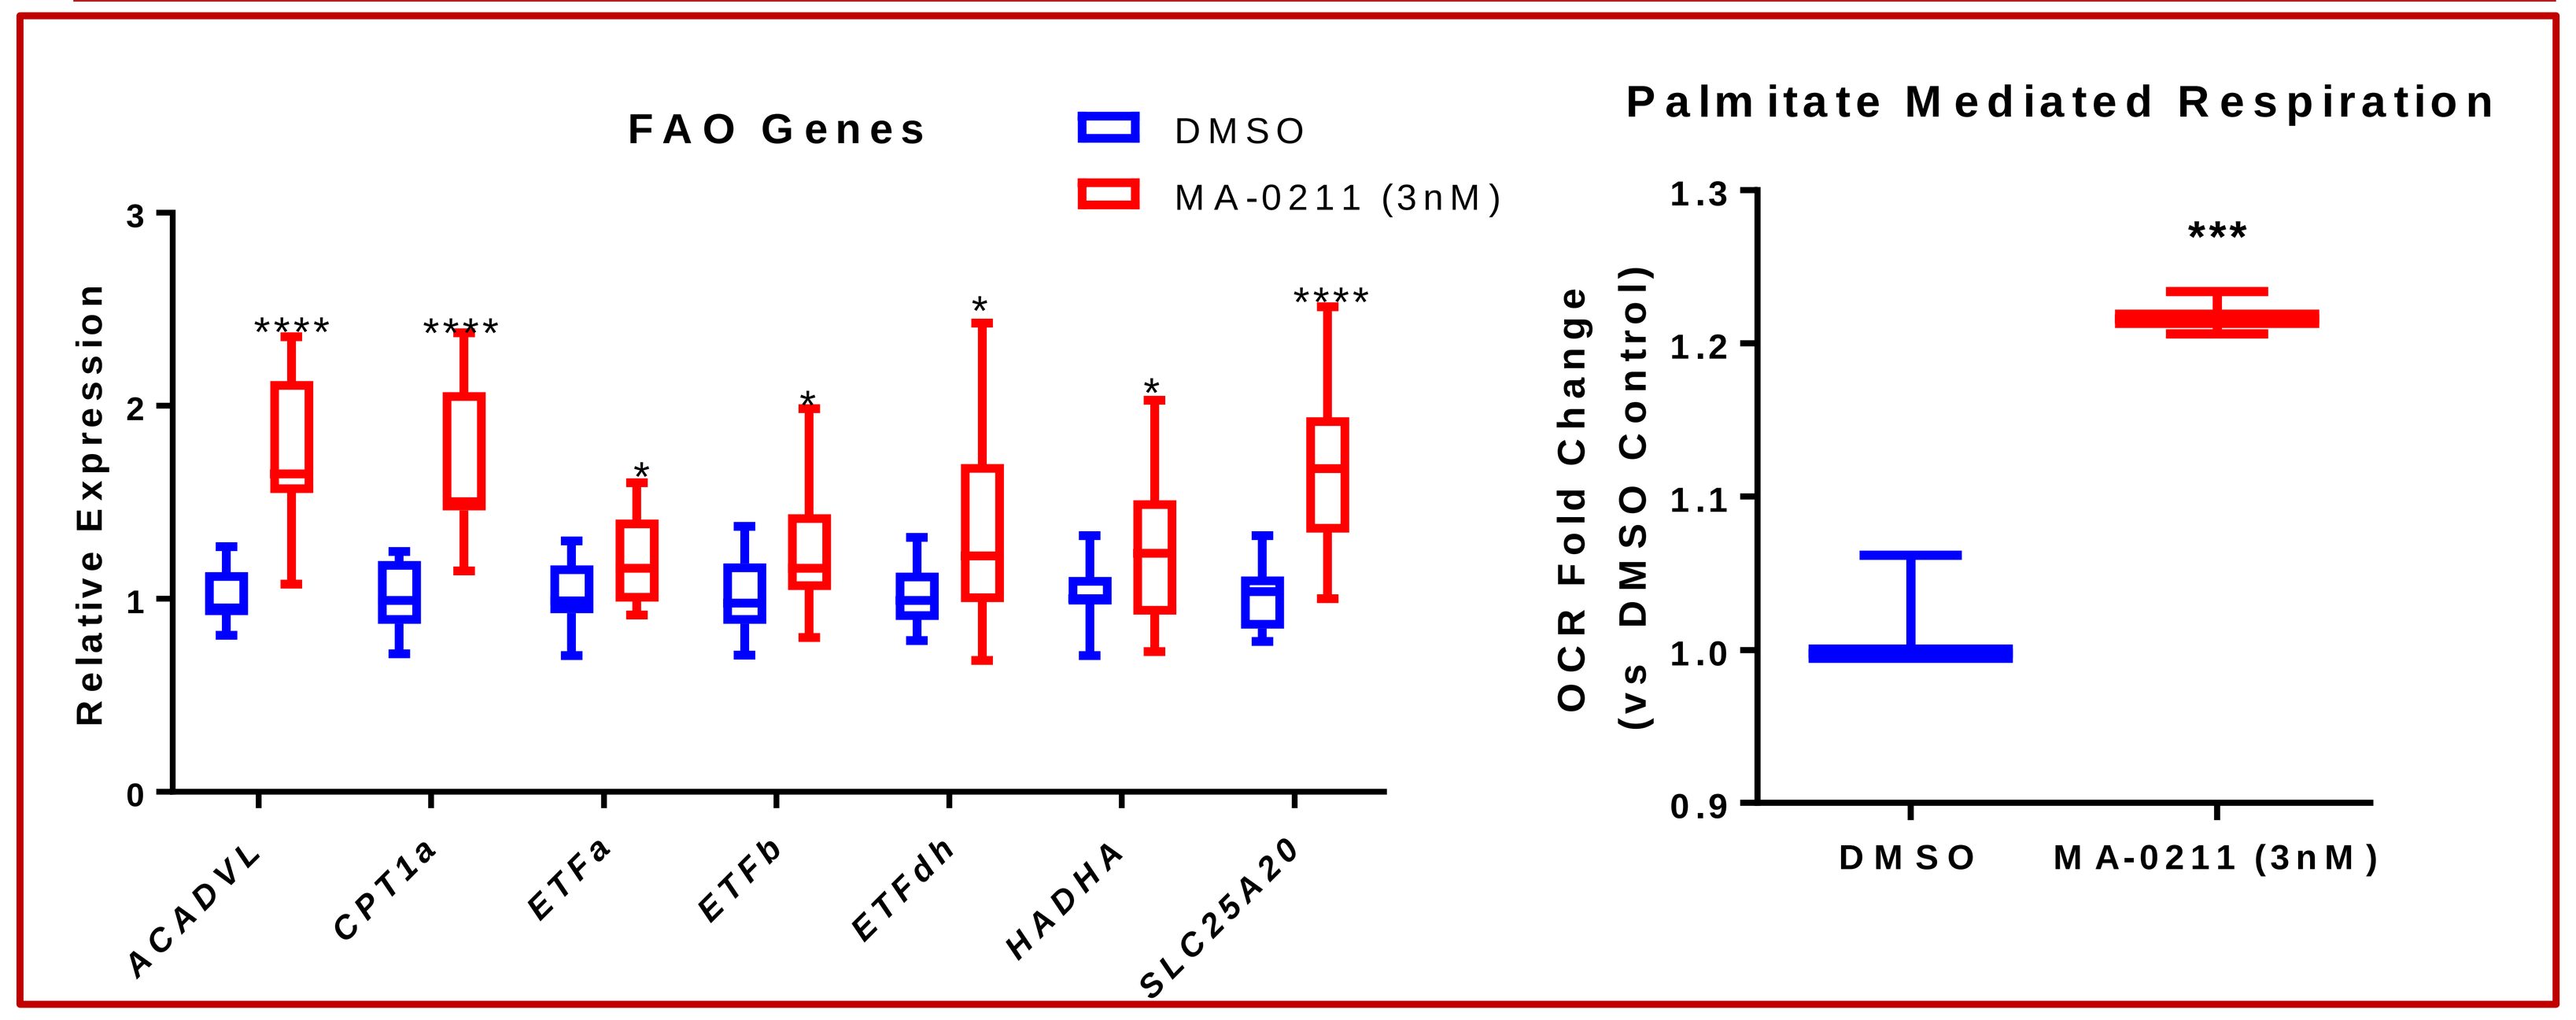
FAO Genes
DMSO
MA-0211 (3nM)
Relative Expression
ACADVL
CPT1a
ETFa
ETFb
ETFdh
HADHA
SLC25A20
Palmitate Mediated Respiration
OCR Fold Change (vs DMSO Control)
DMSO
MA-0211 (3nM)

Data generated in Leigh/LHON patient derived fibroblasts harboring mutations in both ND4 and ND6. *p<0.05, ***p<0.001, ****p<0.0001 unpaired two-tailed t-test. Data is compiled from 3 independent experiments each with one biological replicate.

MA-0211: KSS and MELAS patient derived Fibroblasts

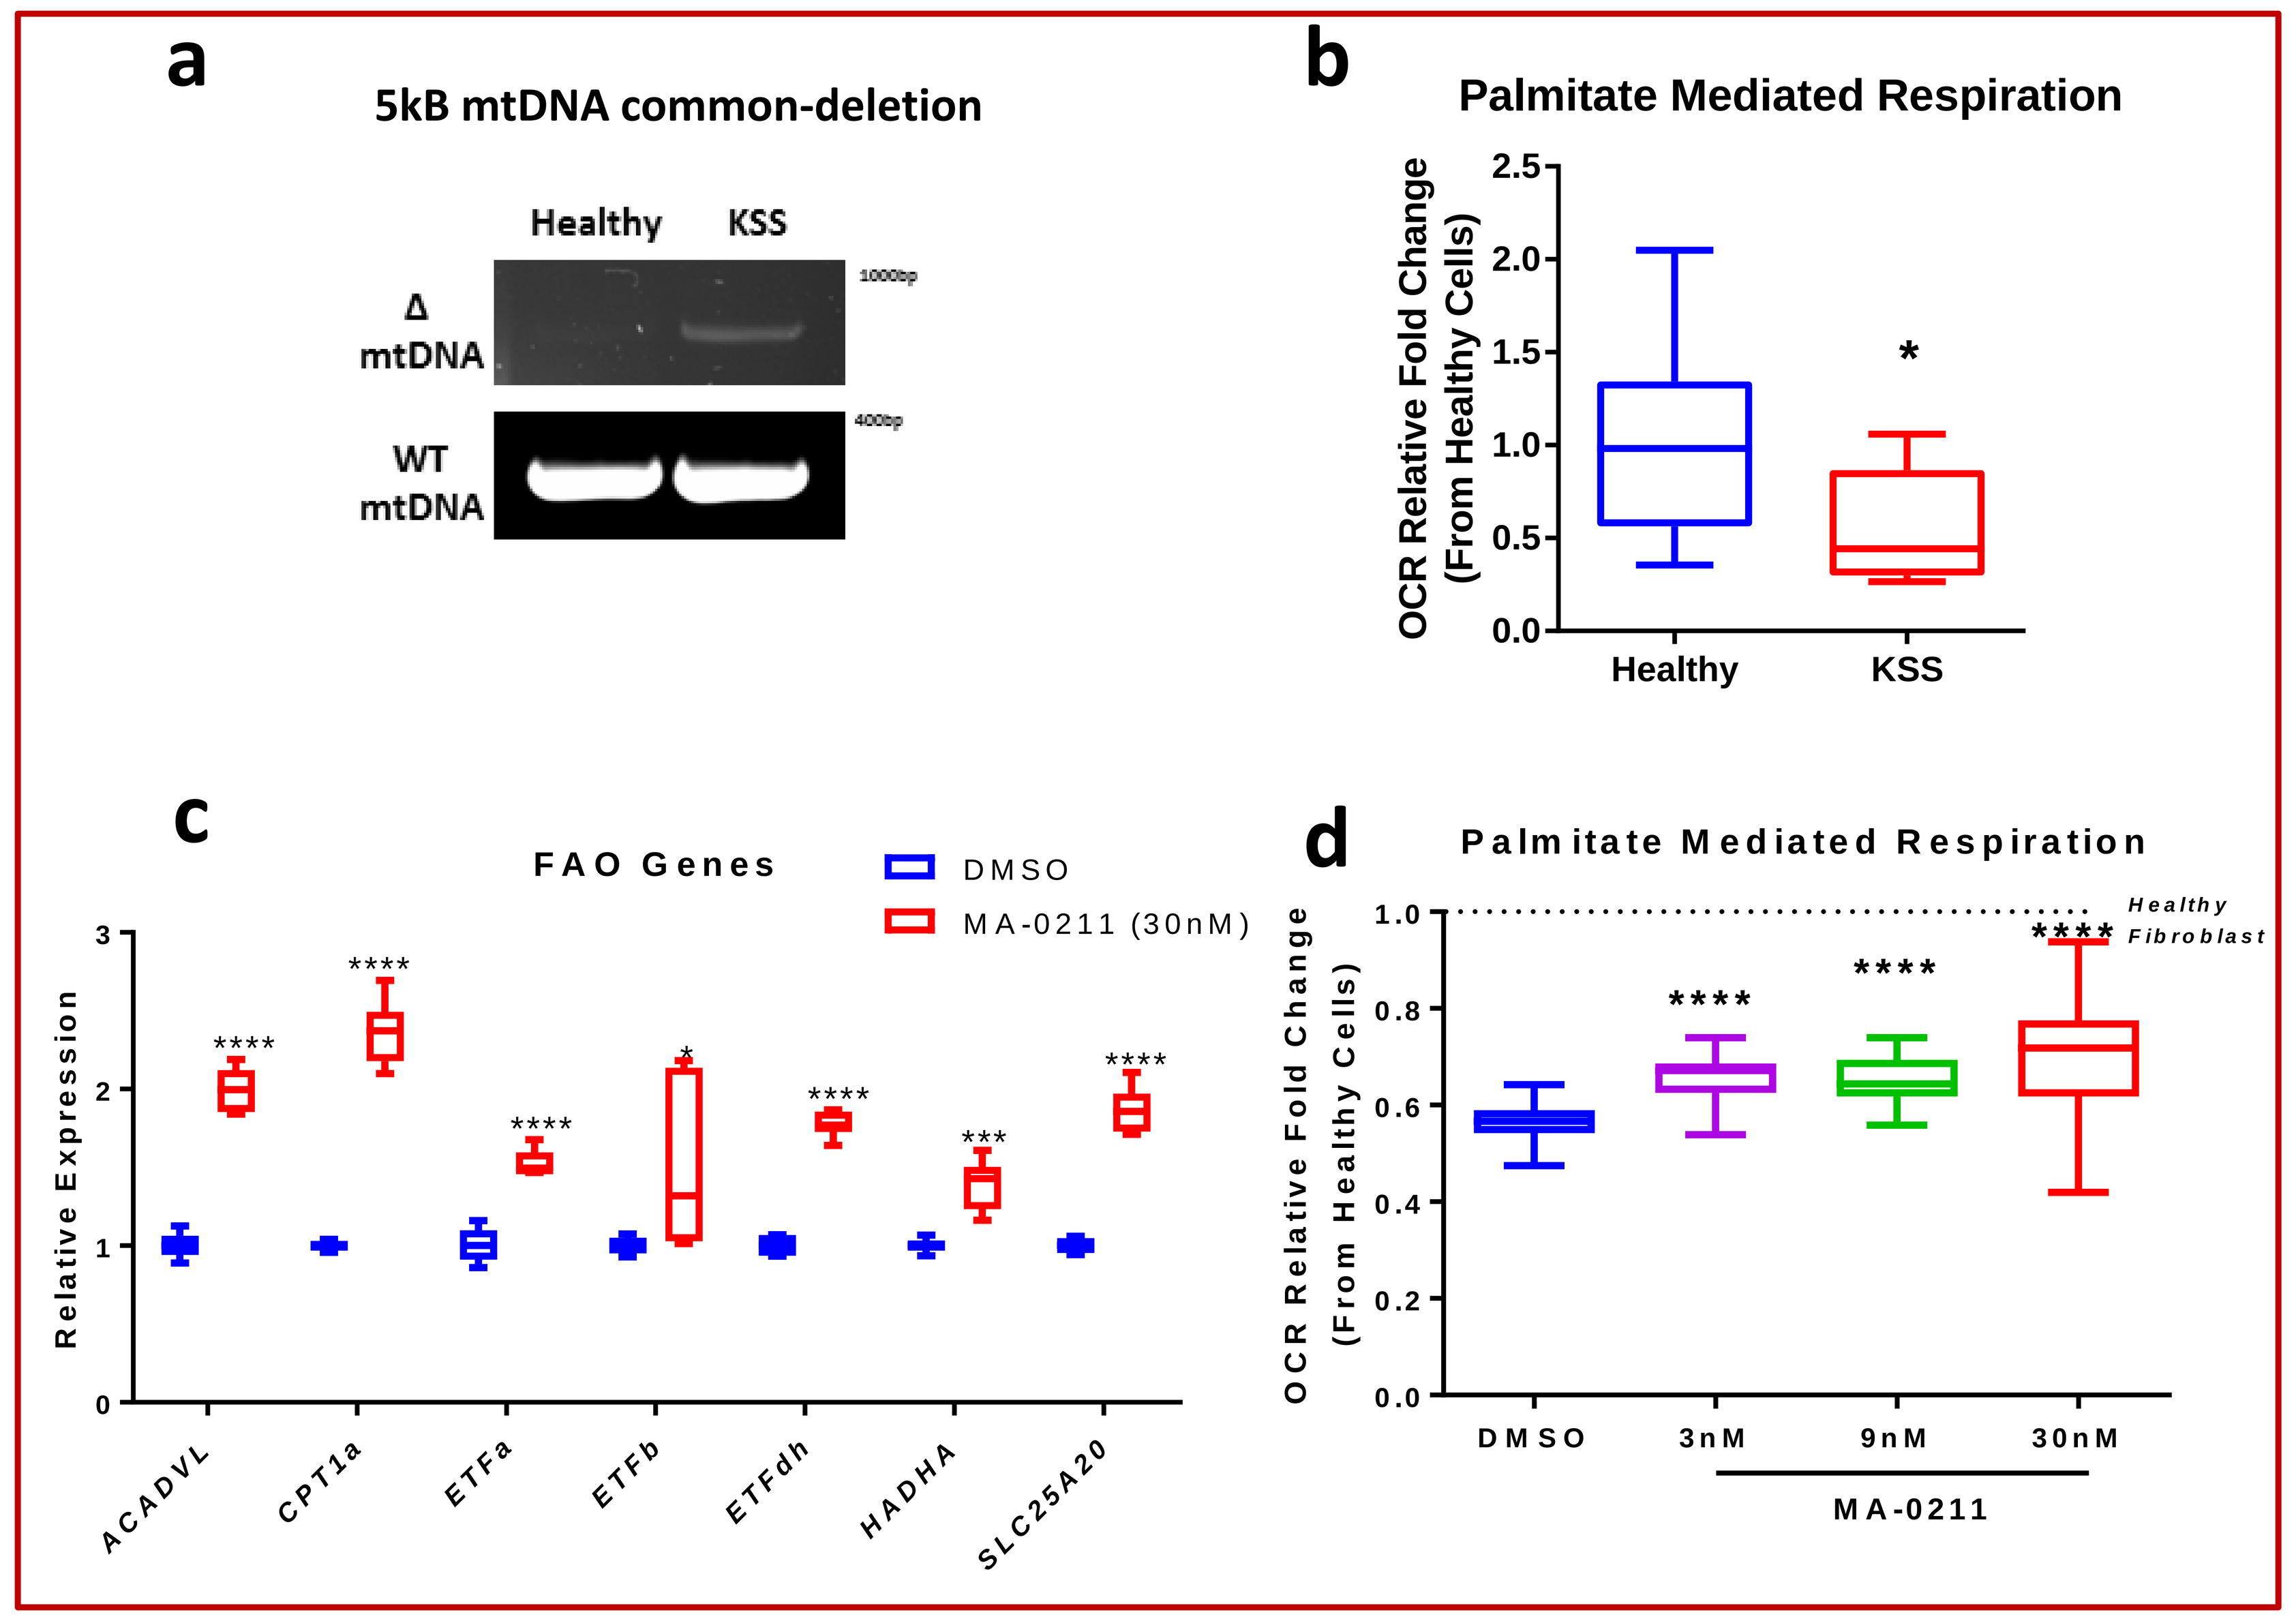
a
5kB mtDNA common-deletion
Healthy
KSS
Δ mtDNA
WT mtDNA
1000bp
400bp
b
Palmitate Mediated Respiration
OCR Relative Fold Change (From Healthy Cells)
Healthy
KSS
c
FAO Genes
DMSO
MA-0211 (30nM)
Relative Expression
ACADVL
CPT1a
ETFa
ETFb
ETFdh
HADHA
SLC25A20
d
Palmitate Mediated Respiration
OCR Relative Fold Change (From Healthy Cells)
Healthy Fibroblast
DMSO
3nM
9nM
30nM
MA-0211

*p<0.05, ***p<0.001, ****p<0.0001; (b) Mann-Whitney test; (c) unpaired two-tailed t-test; (d) Kruskal-Wallis test.

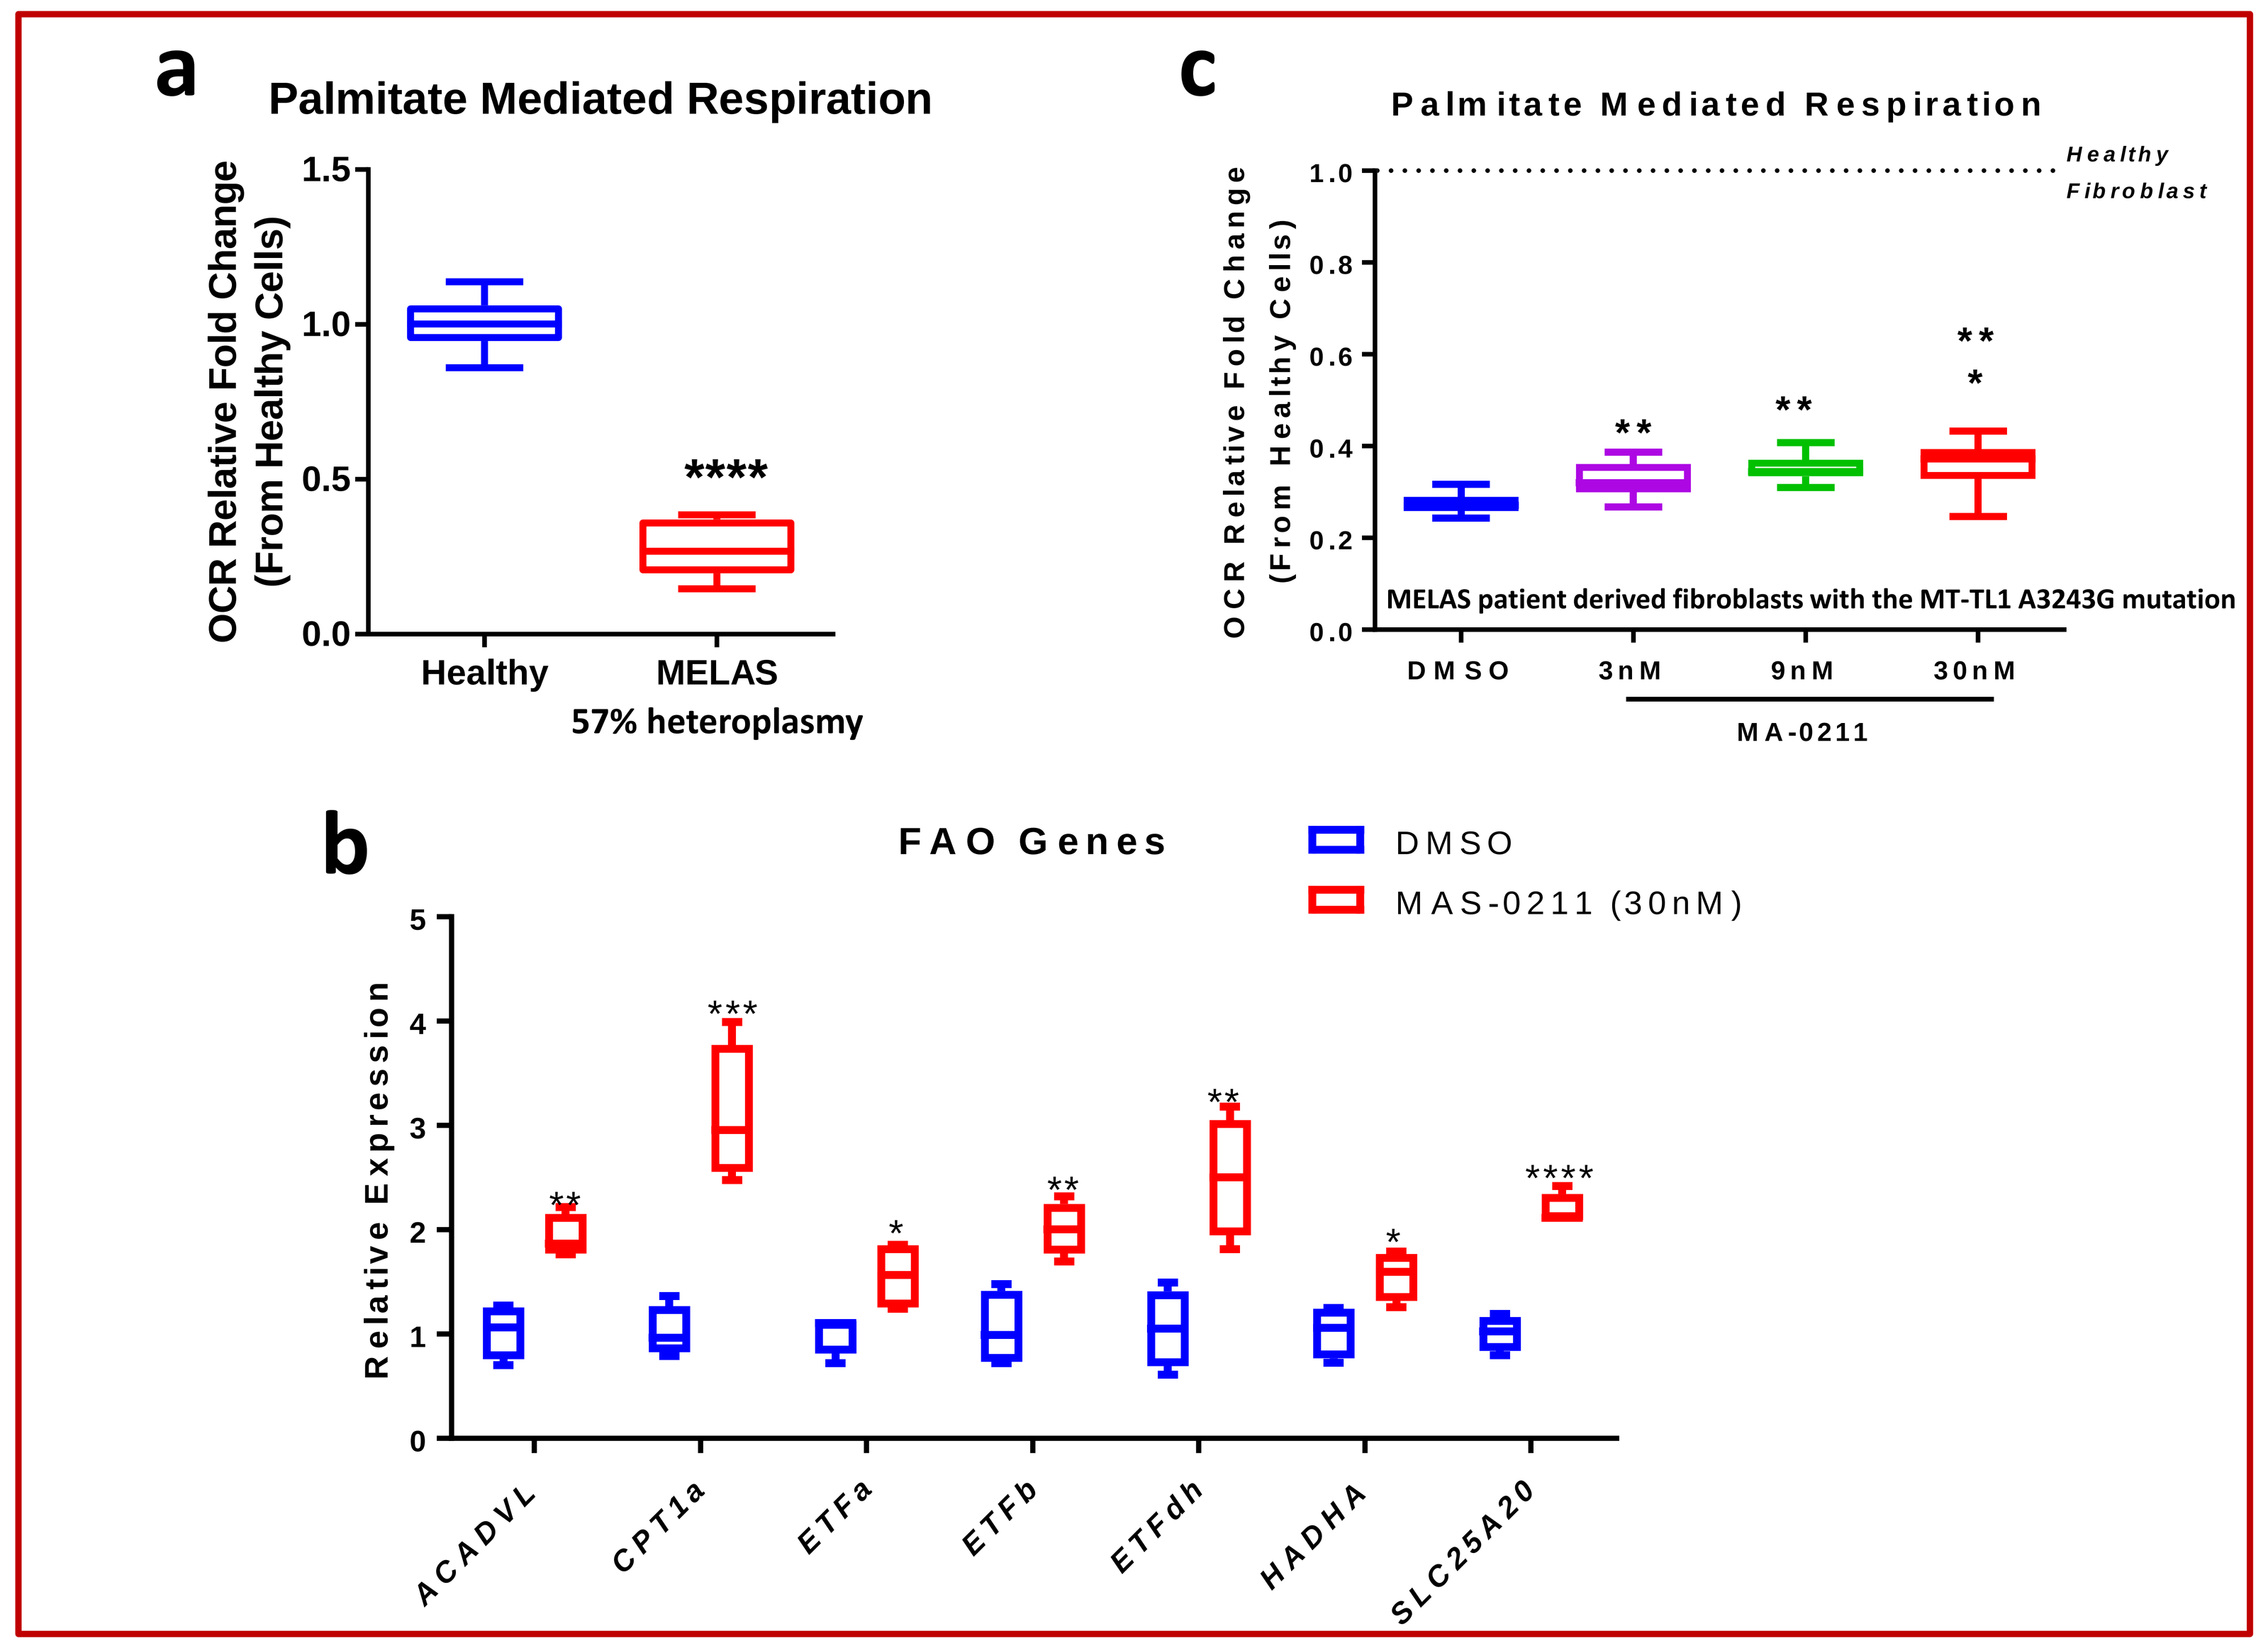
a
Palmitate Mediated Respiration
OCR Relative Fold Change (From Healthy Cells)
Healthy
MELAS
57% heteroplasmy
c
Palmitate Mediated Respiration
OCR Relative Fold Change (From Healthy Cells)
Healthy Fibroblast
MELAS patient derived fibroblasts with the MT-TL1 A3243G mutation
DMSO
3nM
9nM
30nM
MA-0211
b
FAO Genes
DMSO
MAS-0211 (30nM)
Relative Expression
ACADVL
CPT1a
ETFa
ETFb
ETFdh
HADHA
SLC25A20

*p<0.05, **p<0.01, ***p<0.001, ****p<0.0001; (a) Mann-Whitney test ; (b) unpaired two-tailed t-test; (c) One-Way ANOVA Tukey's correction.

MA-0211: MERRF cybrids

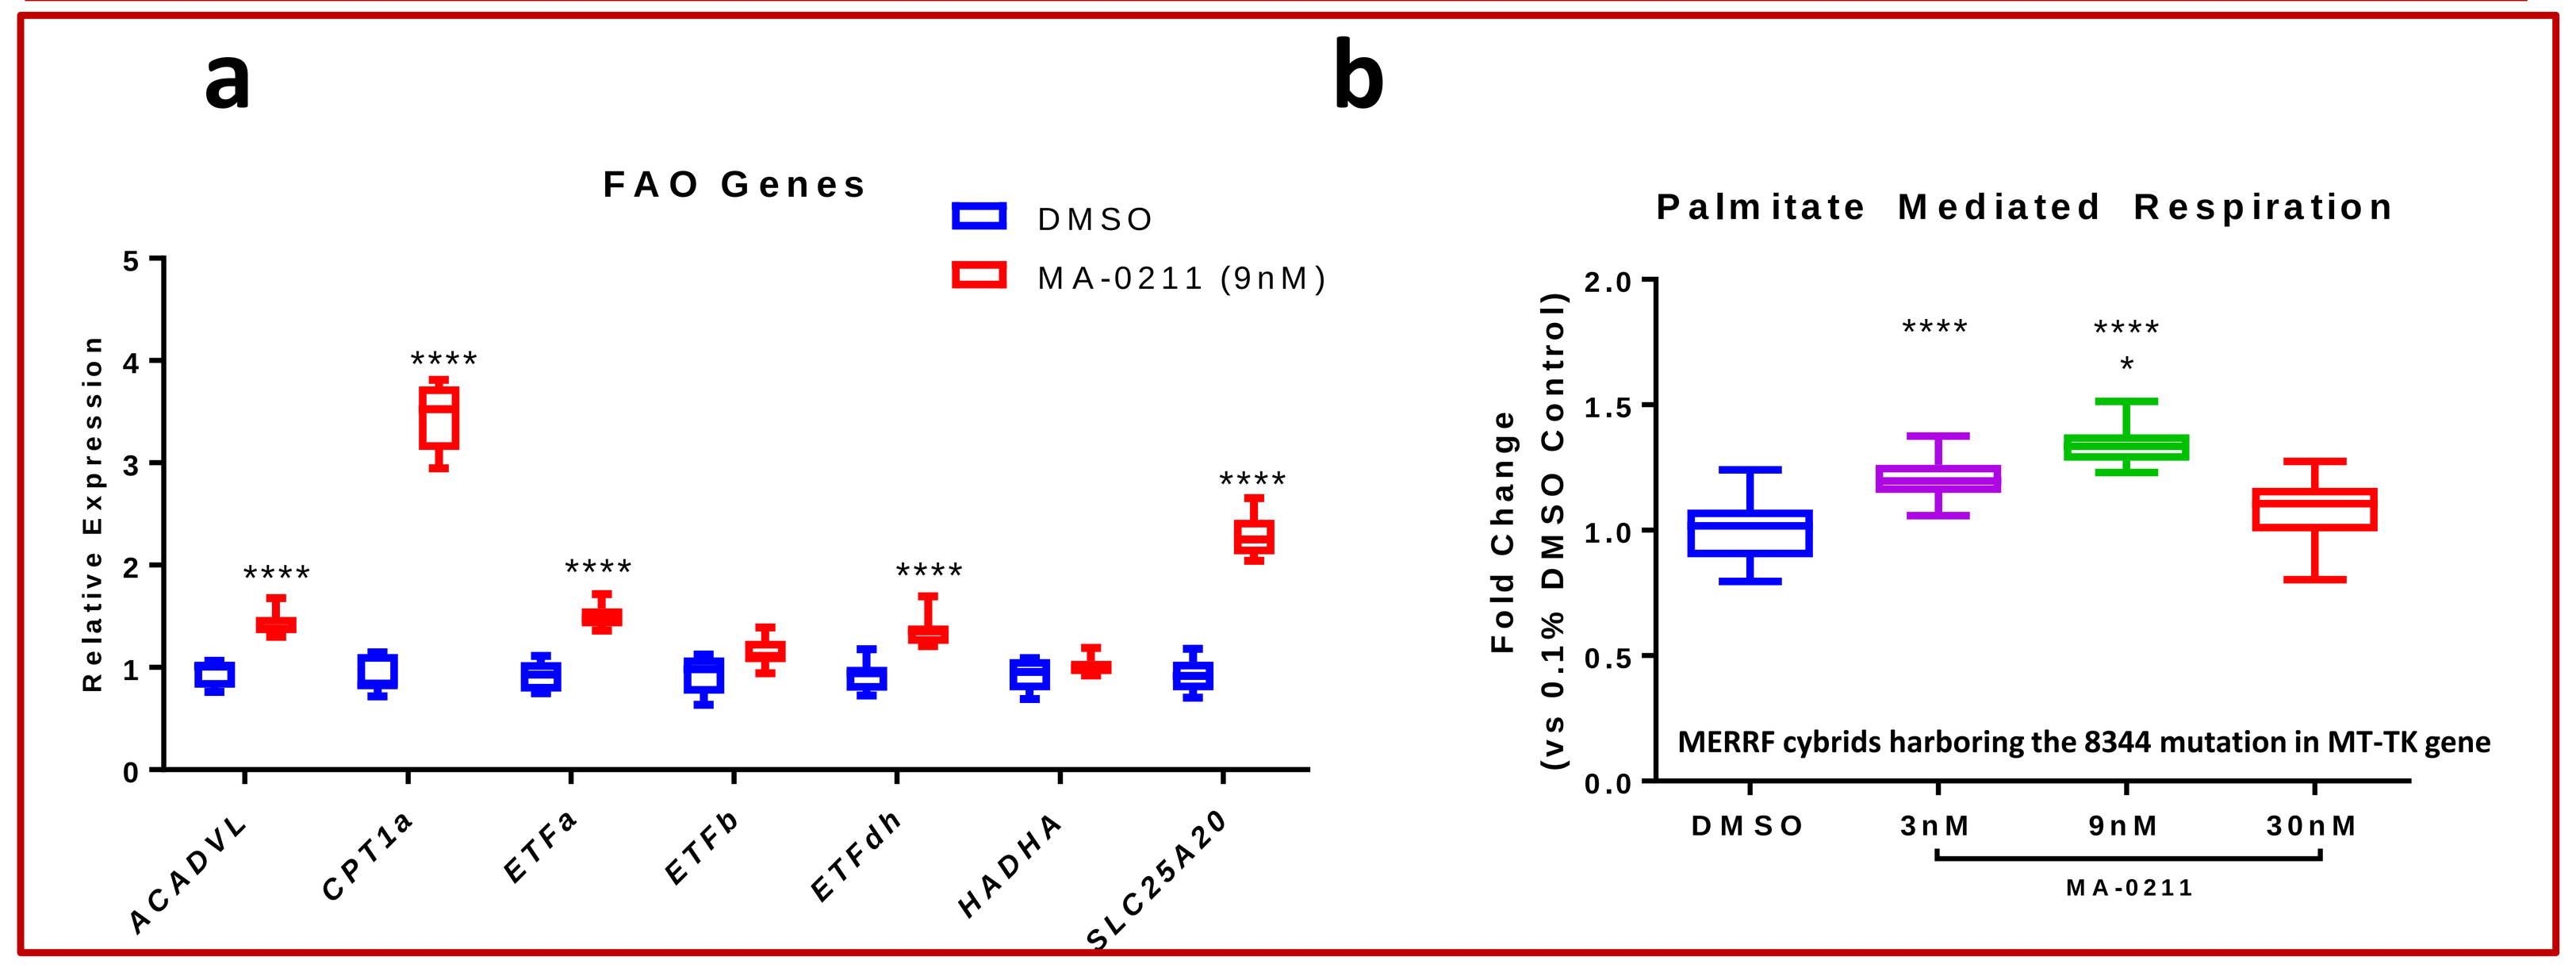
a
FAO Genes
DMSO
MA-0211 (9nM)
Relative Expression
ACADVL
CPT1a
ETFa
ETFb
ETFdh
HADHA
SLC25A20
b
Palmitate Mediated Respiration
Fold Change (vs 0.1% DMSO Control)
MERRF cybrids harboring the 8344 mutation in MT-TK gene
DMSO
3nM
9nM
30nM
MA-0211

*p<0.05, ****p<0.0001; (a) unpaired two-tailed t-test; (b) One-Way ANOVA Tukey's correction.

MA-0211: DIO mice

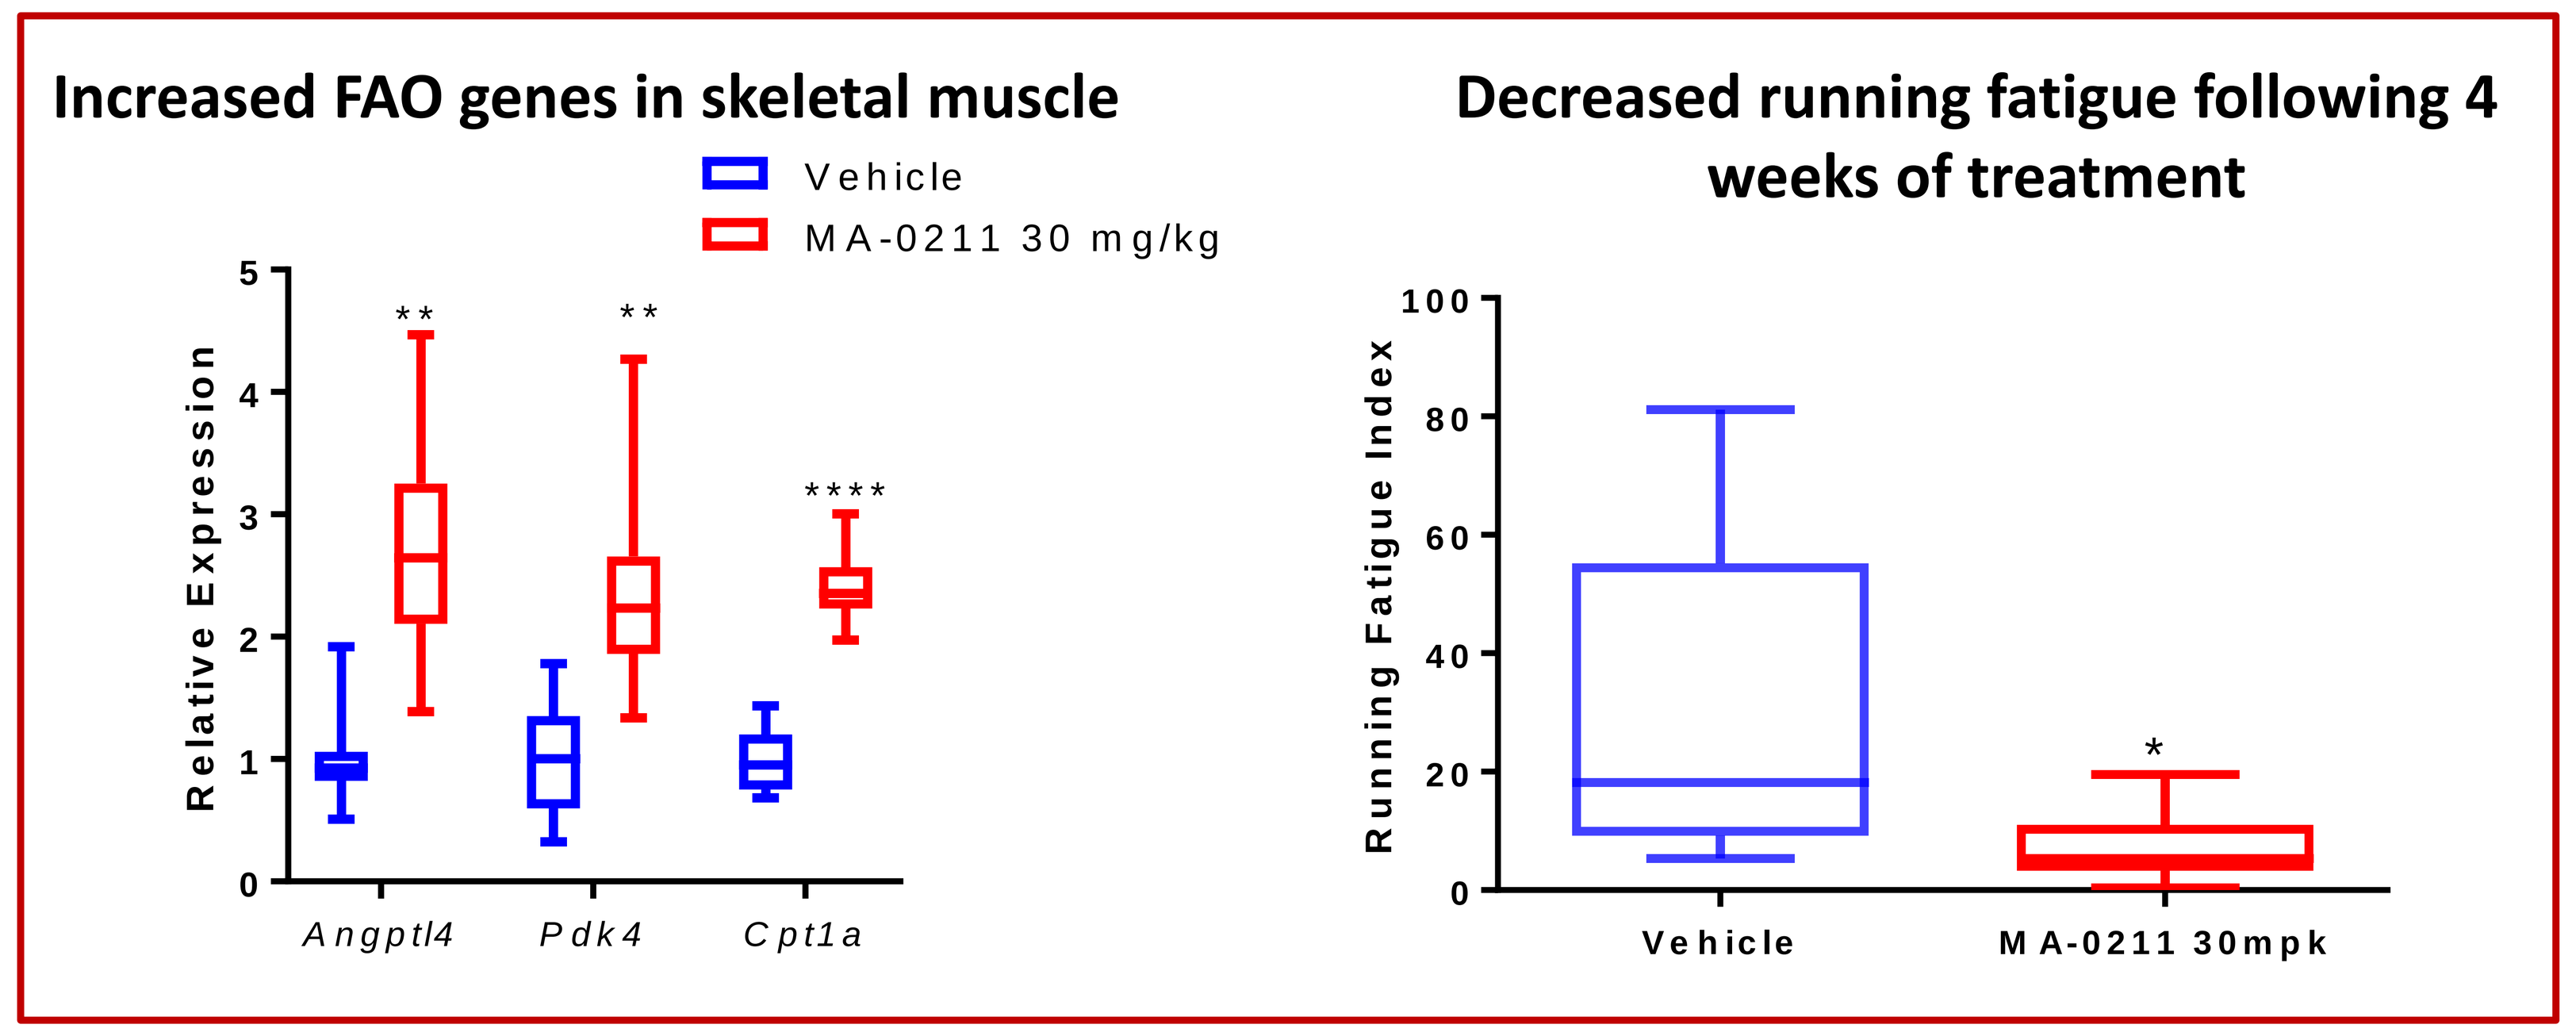
Increased FAO genes in skeletal muscle
Vehicle
MA-0211 30 mg/kg
Relative Expression
Angptl4
Pdk4
Cpt1a
Decreased running fatigue following 4 weeks of treatment
Running Fatigue Index
Vehicle
MA-0211 30mpk

*p<0.05, **p<0.01, ****p<0.0001 unpaired two-tailed t-test

Conclusions and Future Directions

Mitochondrial dysfunction has been increasingly recognized as an important contributor to an array of neuromuscular diseases. The current standard of care consists of a combination of nutritional supplements. A novel approach to treating mitochondrial myopathies (MM) is to identify therapeutics that can improve mitochondrial function in the muscle by increasing fuel utilization through fatty acid oxidation. We showed that MA-0211, a potent and selective modulator of PPARδ, can increase the expression of genes encoding proteins involved in OXPHOS in fibroblasts from MM patients. This leads to an increase in the utilization of fatty acids and an increase in mitochondrial respiration. Remarkably, an increase in mitochondrial respiration could be observed in fibroblasts from patients with a wide variety of mutations, indicating that PPARδ modulation could potentially benefit most MM patients. The in vitro effects of MA-0211 were also shown to translate in vivo by decreasing the fatigue index during running of DIO mice, indicating that the effect of increasing mitochondrial respiration is not just specific to fibroblasts but can also improve muscle function. In preclinical safety studies MA-0211 was shown to be safe and well tolerated. The next step will be to test this therapeutic in clinical trials.

Acknowledgments

We thank Carlos Moraes for providing the MERRF cybrids and for his advice on experimental conditions.

mitobridge