PPARδ modulation partially restores mitochondrial defects resulting from dystrophin loss of function

Eric Bell, Robert Shine, Peter Dwyer, Lyndsay Olson, Jennifer Truong, Ross Fredenburg, Matthew Goddeeris, Effie Tozzo

Mitobridge, Inc. Cambridge, MA 02138

Abstract

Duchenne muscular dystrophy (DMD) is a recessive, fatal X-linked disease that is characterized by progressive skeletal muscle wasting due to absence of dystrophin, which is an essential protein that bridges the inner cytoskeleton and extra-cellular matrix. The *mdx* mouse has a point mutation in the dystrophin gene that results in a similar muscle wasting observed in DMD patients. It has been previously demonstrated that *mdx* muscle has mitochondrial-related deficits including decreased spare respiratory capacity[1], complex 1 mediated ATP production[2], and mitochondrial biomass[3]. However, it was unclear when these deficits manifest in the muscle and what the underlying defect(s) in mitochondrial biology might be.

This study set out to characterize the mitochondria in primary muscle satellite cell derived myoblasts and myotubes from *mdx* mice and wild type control mice. Compared to background-matched healthy cells isolated from wild type mouse muscle, *mdx* derived cells have reduced mitochondrial bioenergetics. Moreover, *mdx* derived cells show reduced levels of mitochondria, which may partially explain the reduction in bioenergetics. These *mdx* cells also have increased levels of the nuclear hormone receptor PPARδ without increased levels of putative target genes such as *Angptl4* and *Pdk4*. Here, we demonstrate that a novel PPARδ modulator improves mitochondrial function in the *mdx* model, which supports the notion that modulating PPARδ may be therapeutically beneficial in DMD patients.

Materials and Analysis

Wild type (WT) and *mdx* myoblast isolation and culture: Quadricep and gastrocnemius muscles from a single mouse were pooled and subjected to a mechanical/collagenase digestion. Isolated myoblasts were cultured on Matrigel-coated cultureware in DMEM/F-12 with 10% FBS and 20ng/mL bFGF. Myoblast homogeneity was determined by a co-staining for vimentin and desmin (data not shown).

Myotube differentiation: Confluent myoblasts were changed to DMEM + 2%FBS + insulin-selenium-transferrin for 5 days with the assay performed Day 6.

Steady state ATP measurement: Cells were treated with DMSO (basal) or 100nM oligomycin for 24h. Steady state ATP was then measured using the CellTiter-Glo assay (Promega).

Fatty Acid Oxidation: Myoblasts were starved for 18h in low glucose media with carnitine. KHB plus BSA or BSA-palmitate was added and oxygen consumption was analyzed using a Seahorse Metabolic Analyzer. The palmitate response was determined by calculating a ratio of the FCCP-induced BSA-palmitate respiration/FCCP-induced BSA respiration.

Gene expression: RNA was isolated using a NucleoSpin RNA kit (Macherey-Nagle) and converted to cDNA with the High Capacity cDNA Reverse Transcriptase Kit (Thermo). Gene expression was then determined using the SmartChip Real-Time PCR system (Wafergen) and analyzed with Qbase (BioGazelle).

Statistical analysis: Data was graphed as boxplots and statistical significance determined using GraphPad Prism. Comparisons were performed between WT and *mdx* in the various experimental conditions.*p<0.05; **p<0.01; ***p<0.001; ****p<0.0001

Results

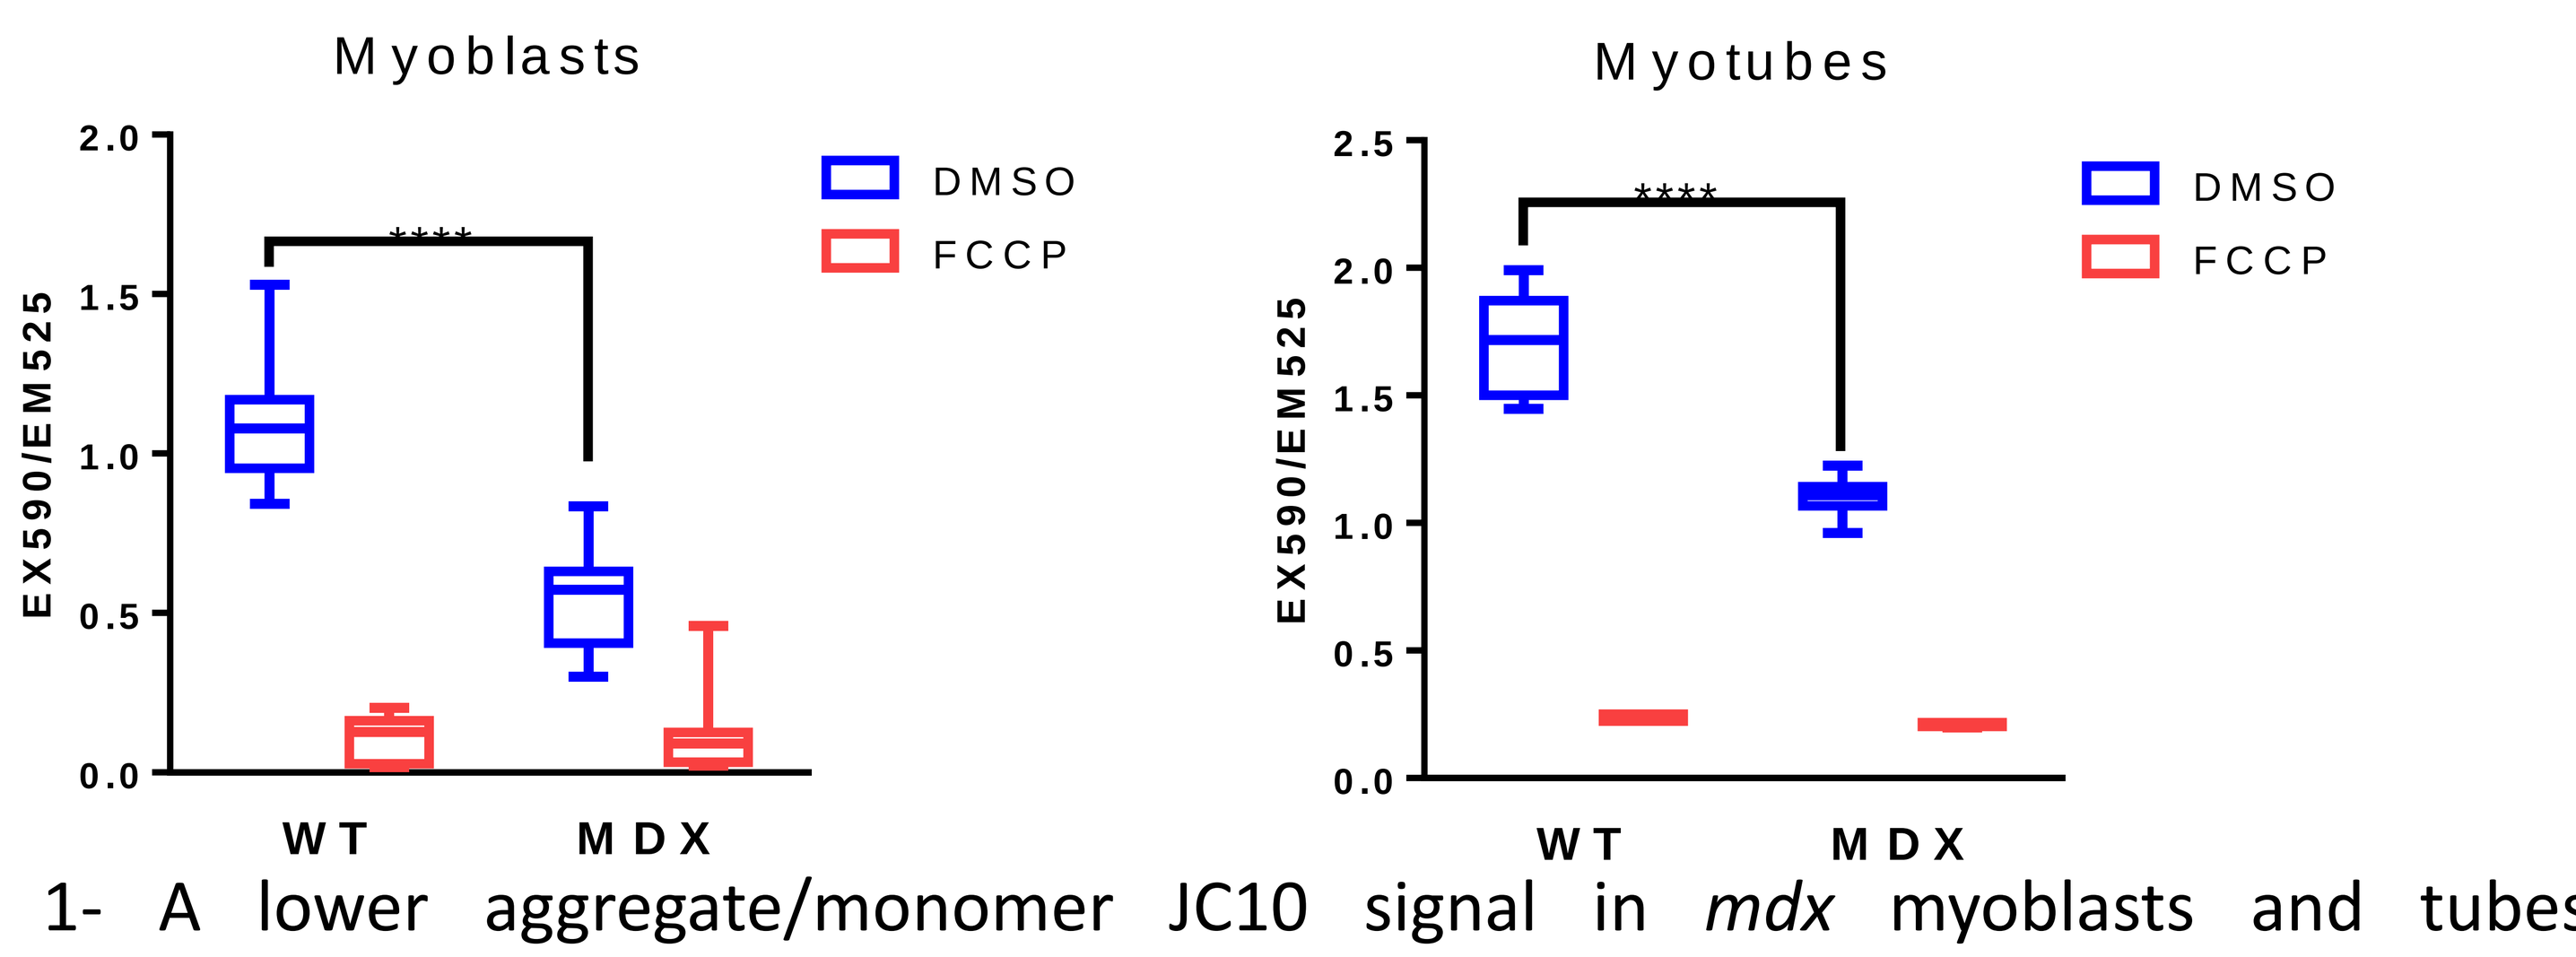
Myoblasts
Myotubes
EX590/EM525
DMSO
FCCP

WT
MDX

Figure 1- A lower aggregate/monomer JC10 signal in *mdx* myoblasts and tubes demonstrates a lower membrane potential

Results

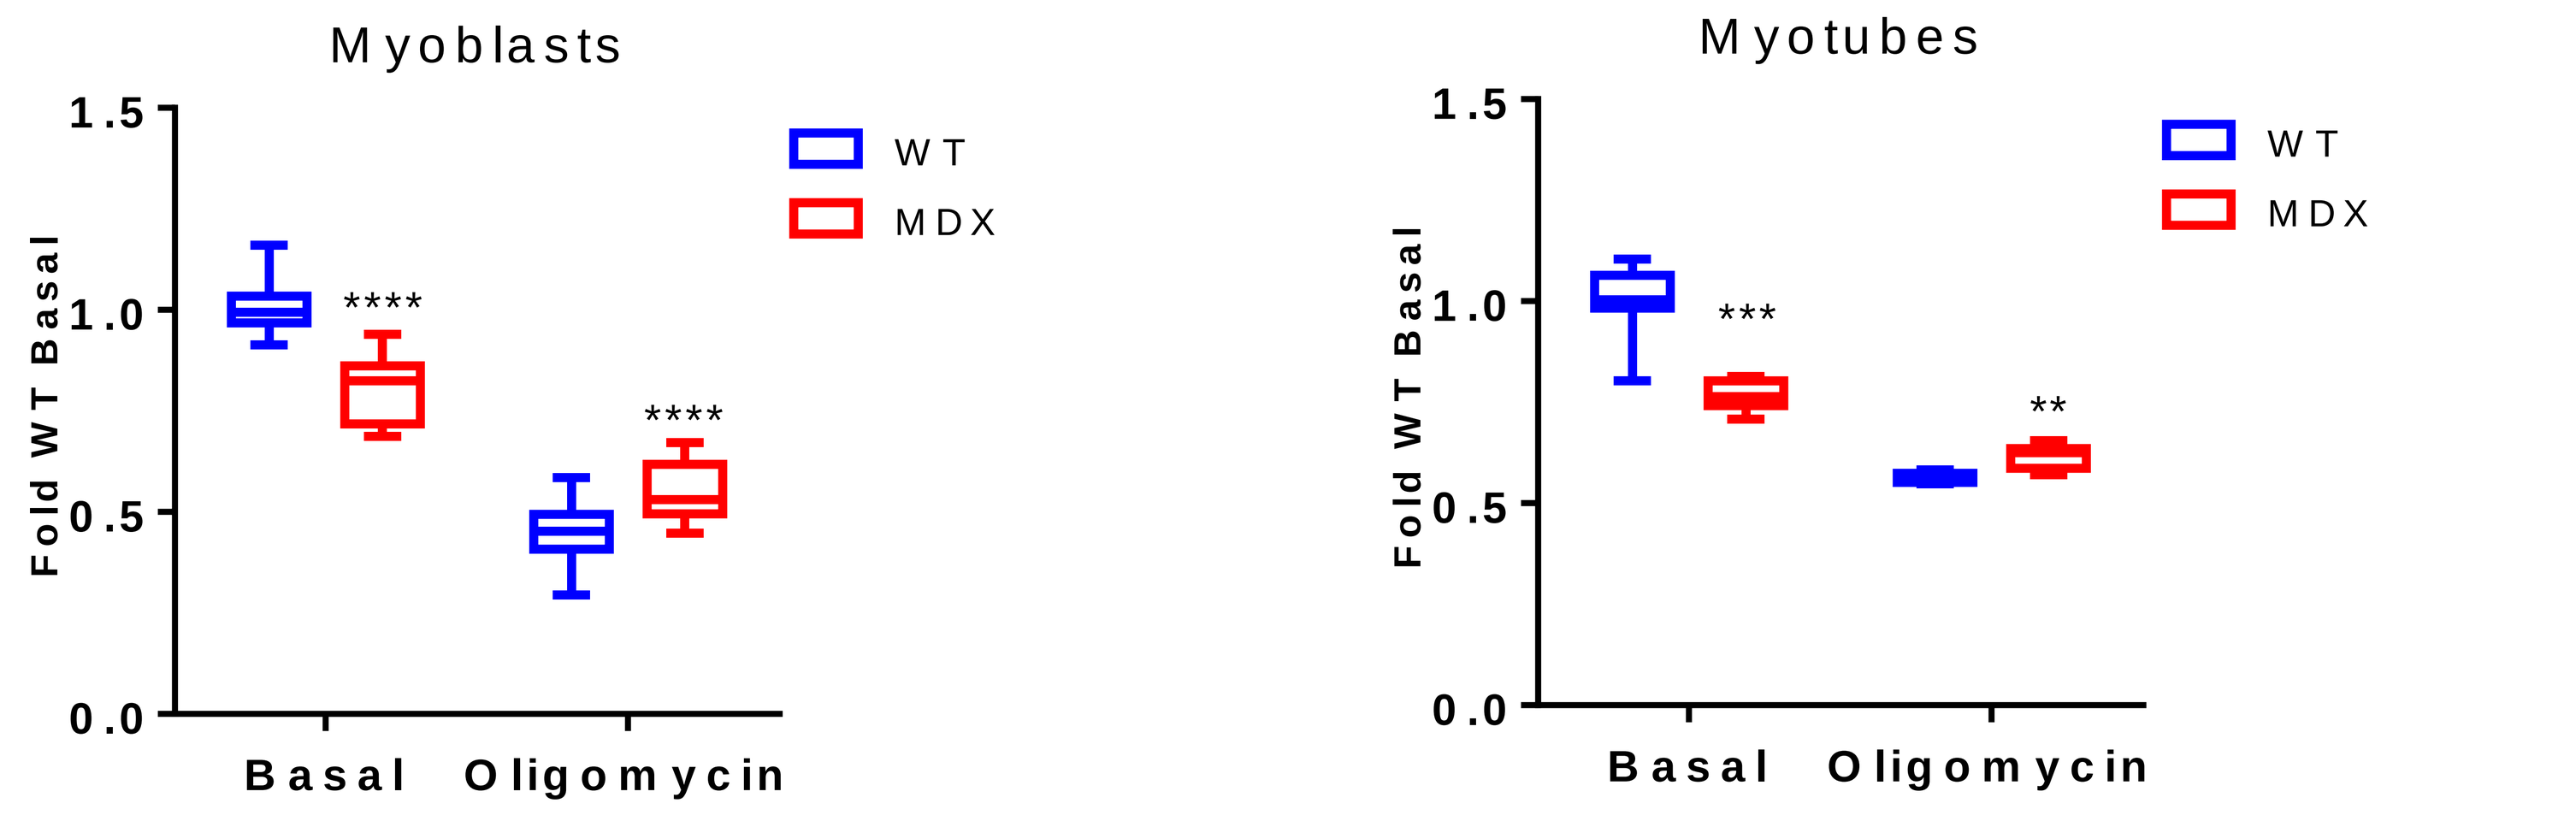
Myoblasts
Myotubes
Fold WT Basal
WT
MDX
Basal
Oligomycin

Figure 2- *mdx* myoblasts and myotubes have lower steady state levels of ATP and are less sensitive to oligomycin treatment indicating decreased mitochondrial ATP production.

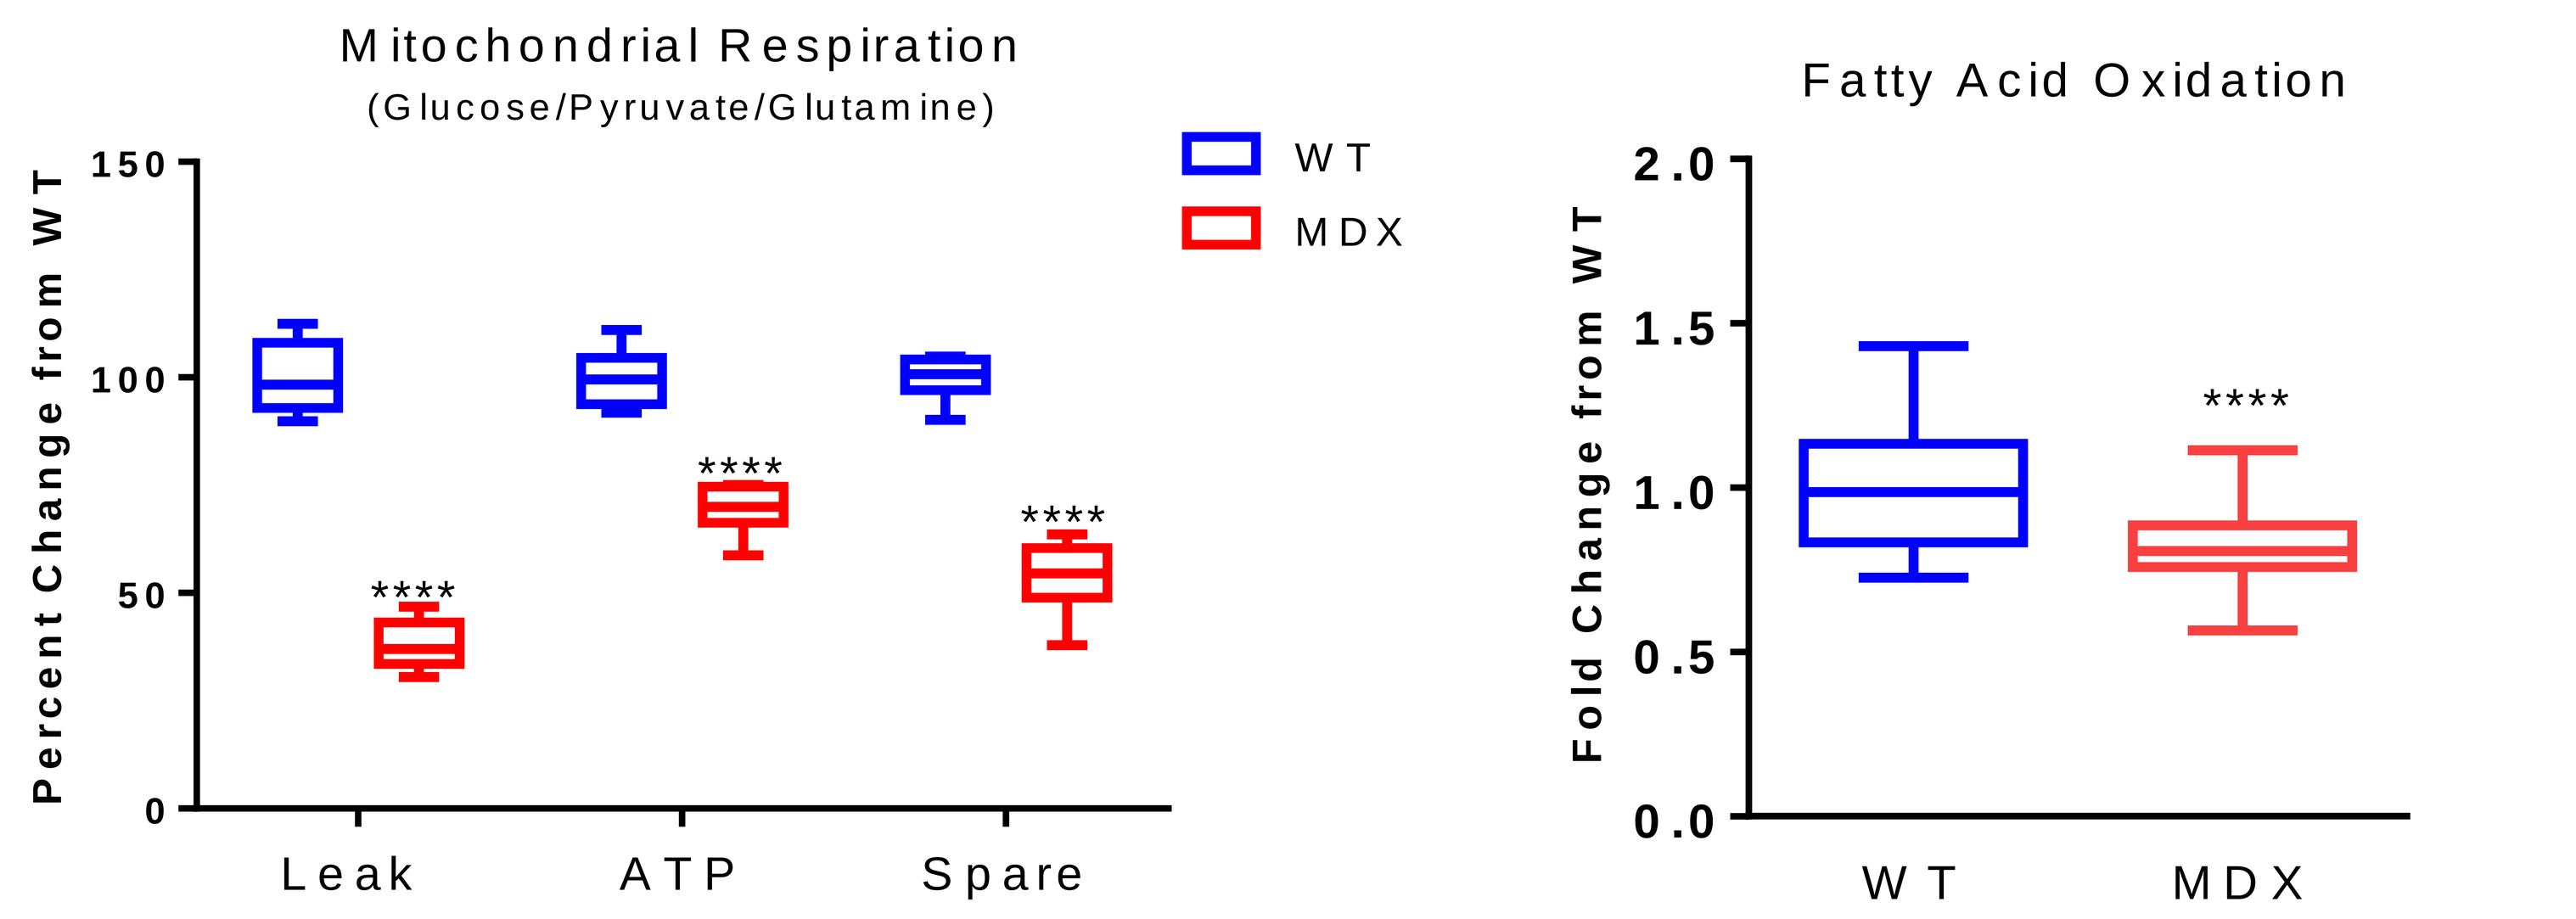
Mitochondrial Respiration
(Glucose/Pyruvate/Glutamine)
Percent Change from WT
Leak
ATP
Spare
Fatty Acid Oxidation
Fold Change from WT
WT
MDX

Figure 3- Oxidation of multiple substrates is decreased in the context of dystrophin mutation.

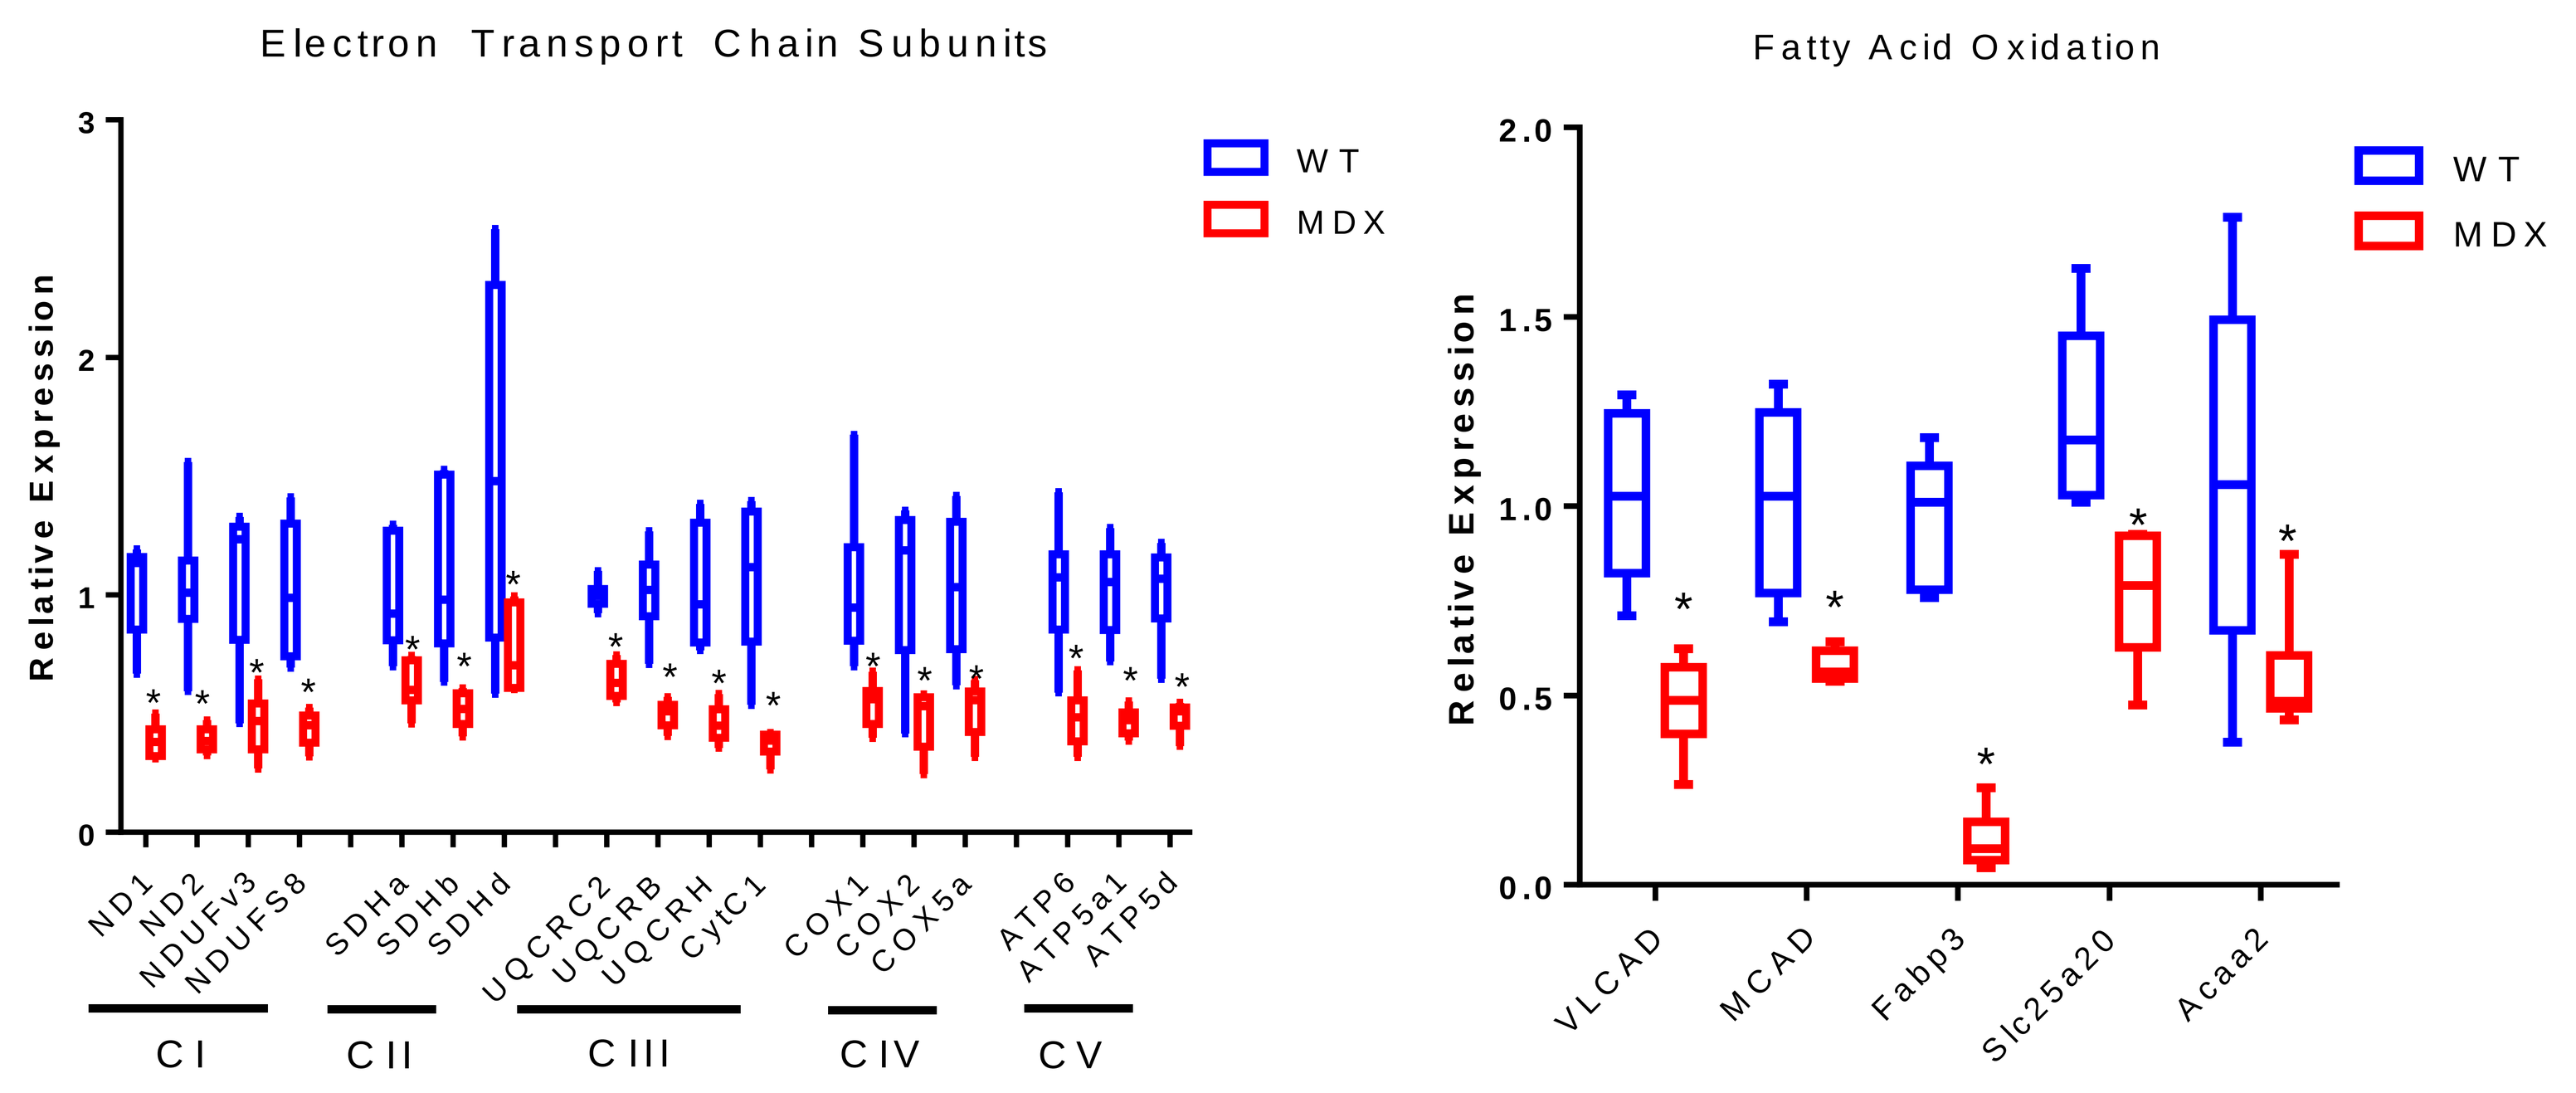
Electron Transport Chain Subunits
Relative Expression
WT
MDX
ND1
ND2
NDUFv3
NDUFS8
SDHa
SDHb
SDHd
UQCRC2
UQCRB
UQCRH
CytC1
COX1
COX2
COX5a
ATP6
ATP5a1
ATP5d
C I
C II
C III
C IV
C V
Fatty Acid Oxidation
VLCAD
MCAD
Fabp3
Slc25a20
Acaa2

Figure 4- mdx myoblasts exhibit lower decreased expression of electron transport chain subunits and fatty acid oxidation genes.

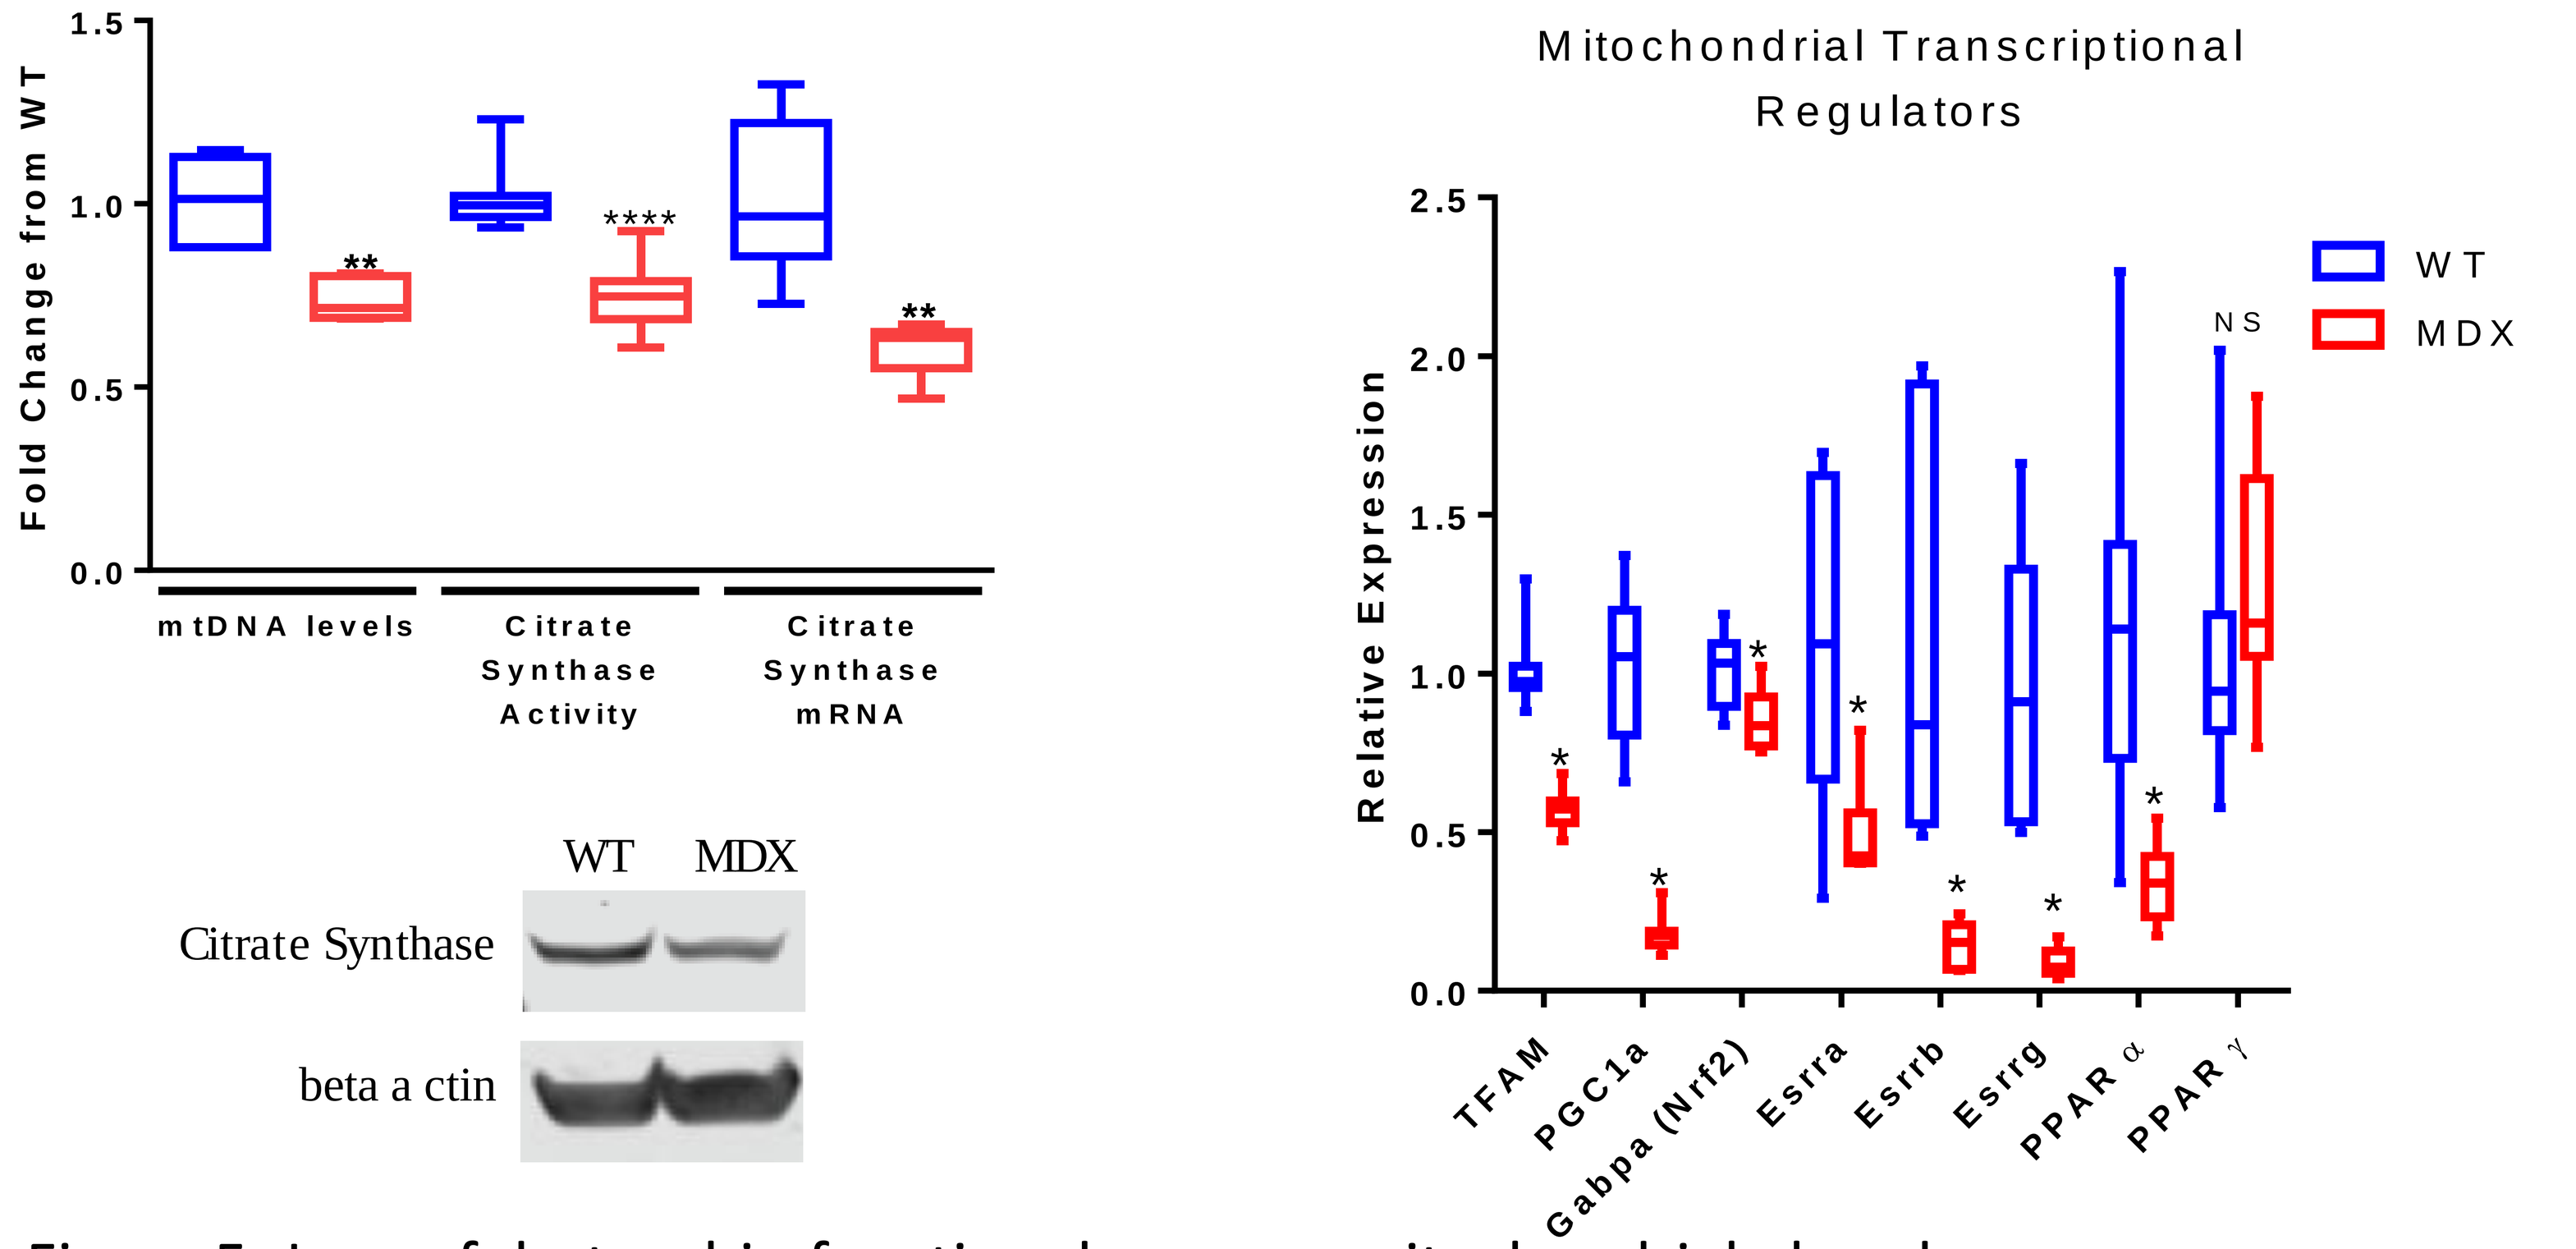
Fold Change from WT
mtDNA levels
Citrate Synthase Activity
Citrate Synthase mRNA
WT
MDX
Citrate Synthase
beta a ctin
Mitochondrial Transcriptional Regulators
Relative Expression
NS
TFAM
PGC1a
Gabpa (Nrf2)
Esrra
Esrrb
Esrrg
PPAR α
PPAR γ

Figure 5- Loss of dystrophin function decreases mitochondrial abundance

Results

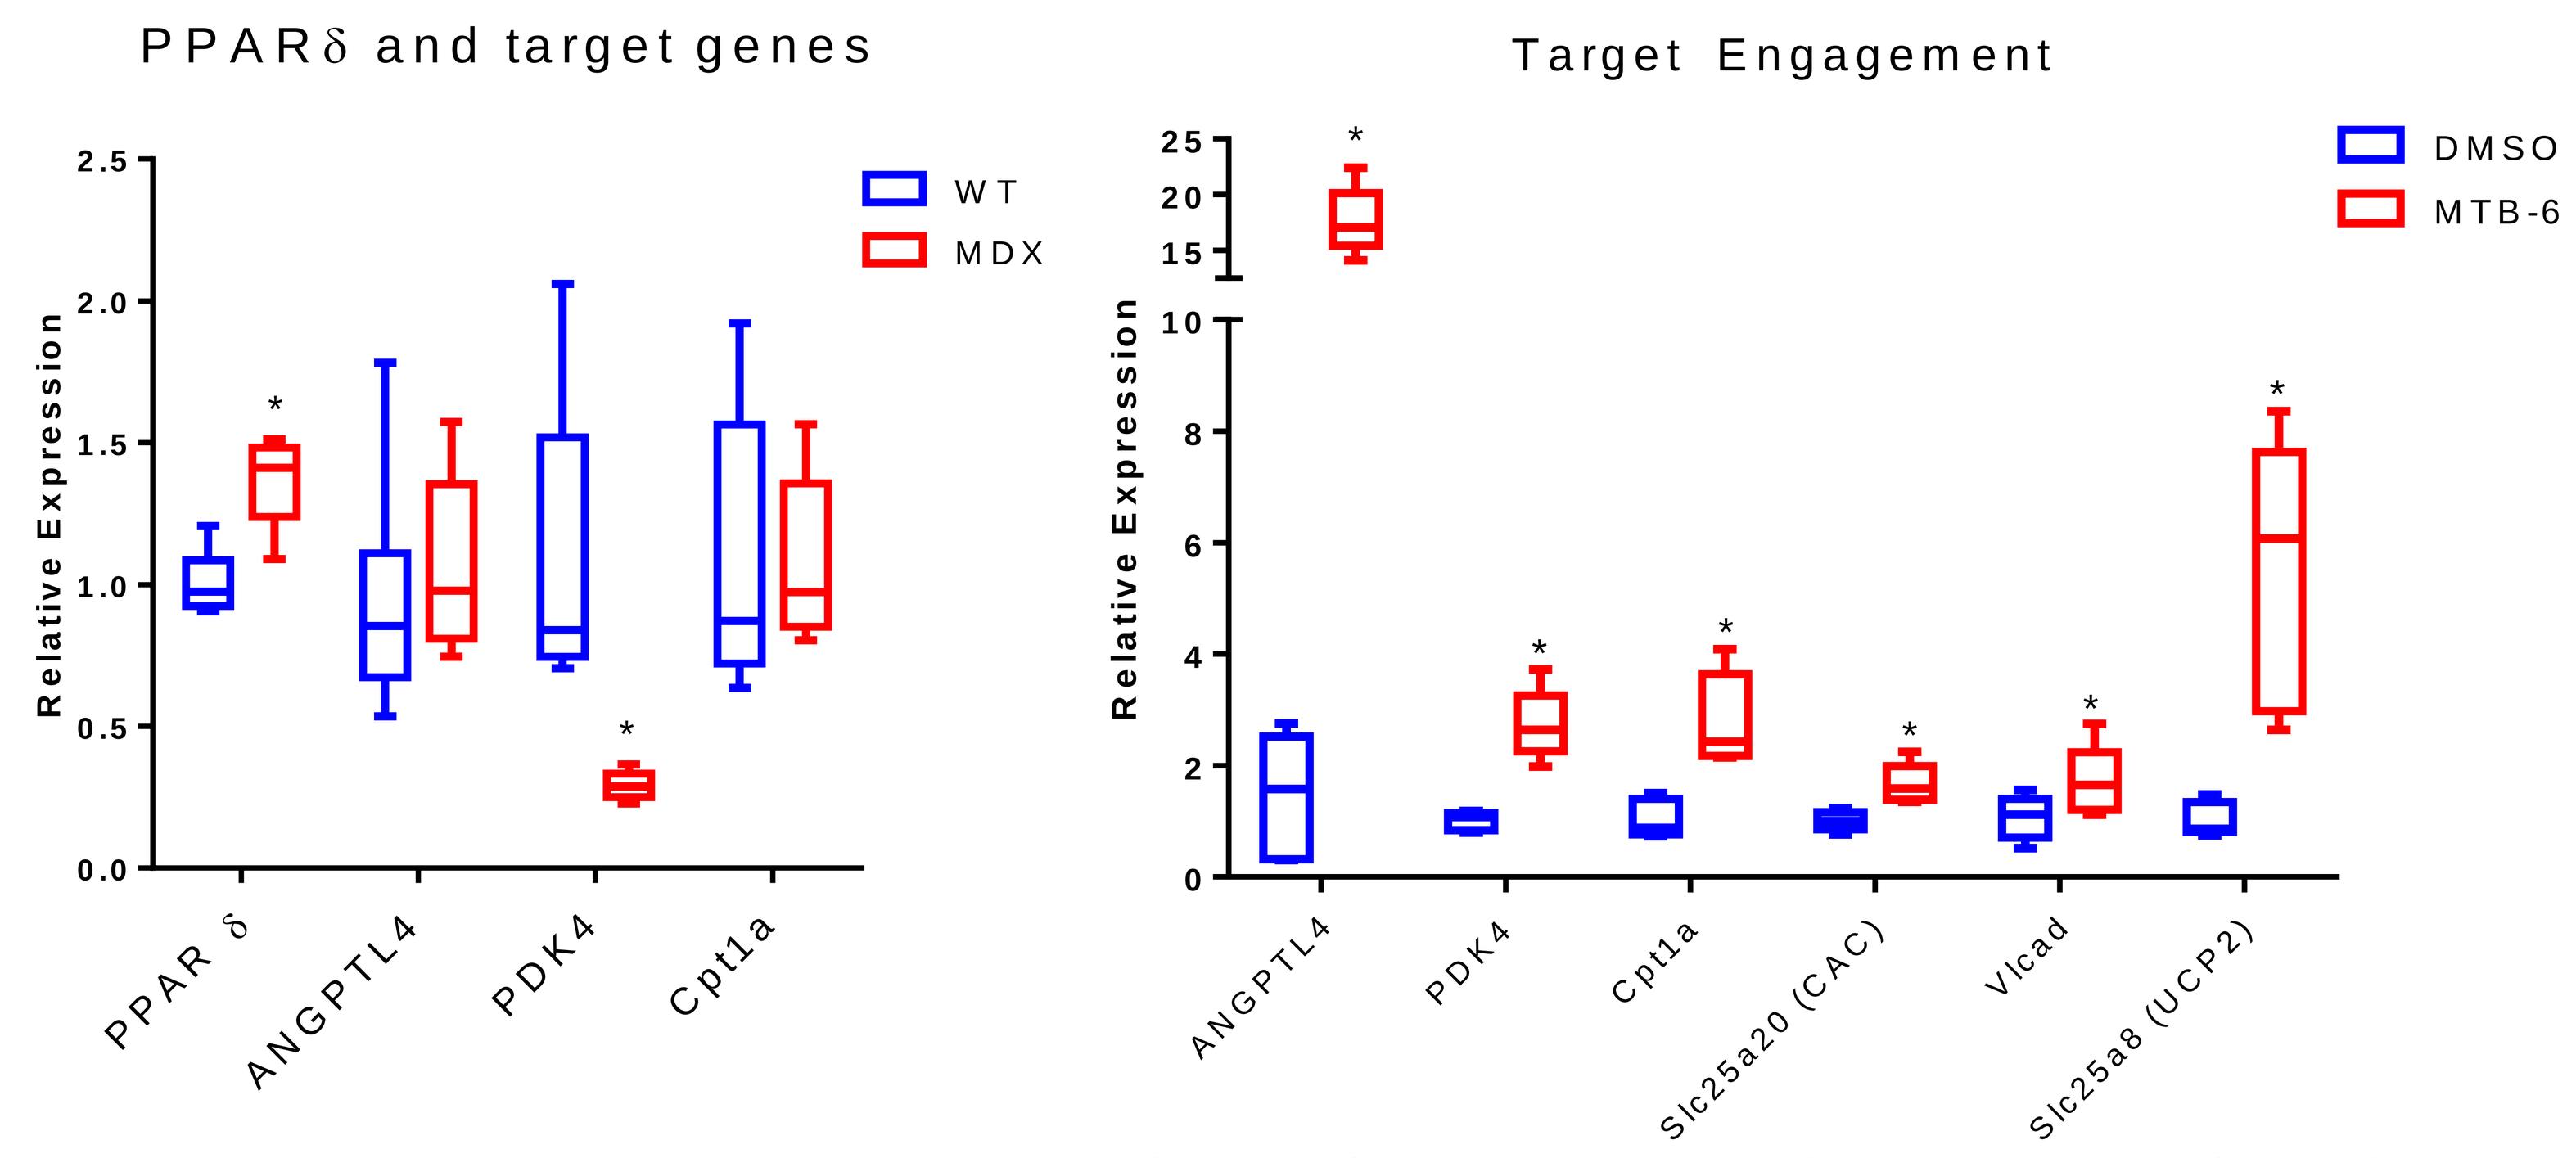
PPARδ and target genes
Relative Expression
WT
MDX
PPAR δ
ANGPTL4
PDK4
Cpt1a
Target Engagement
DMSO
MTB-6
Slc25a20 (CAC)
Vlcad
Slc25a8 (UCP2)

Figure 6- Expression of PPARδ is increased in *mdx* myoblasts without increased levels of putative target genes; treatment with a PPARδ modulator increases PPARδ-target gene expression.

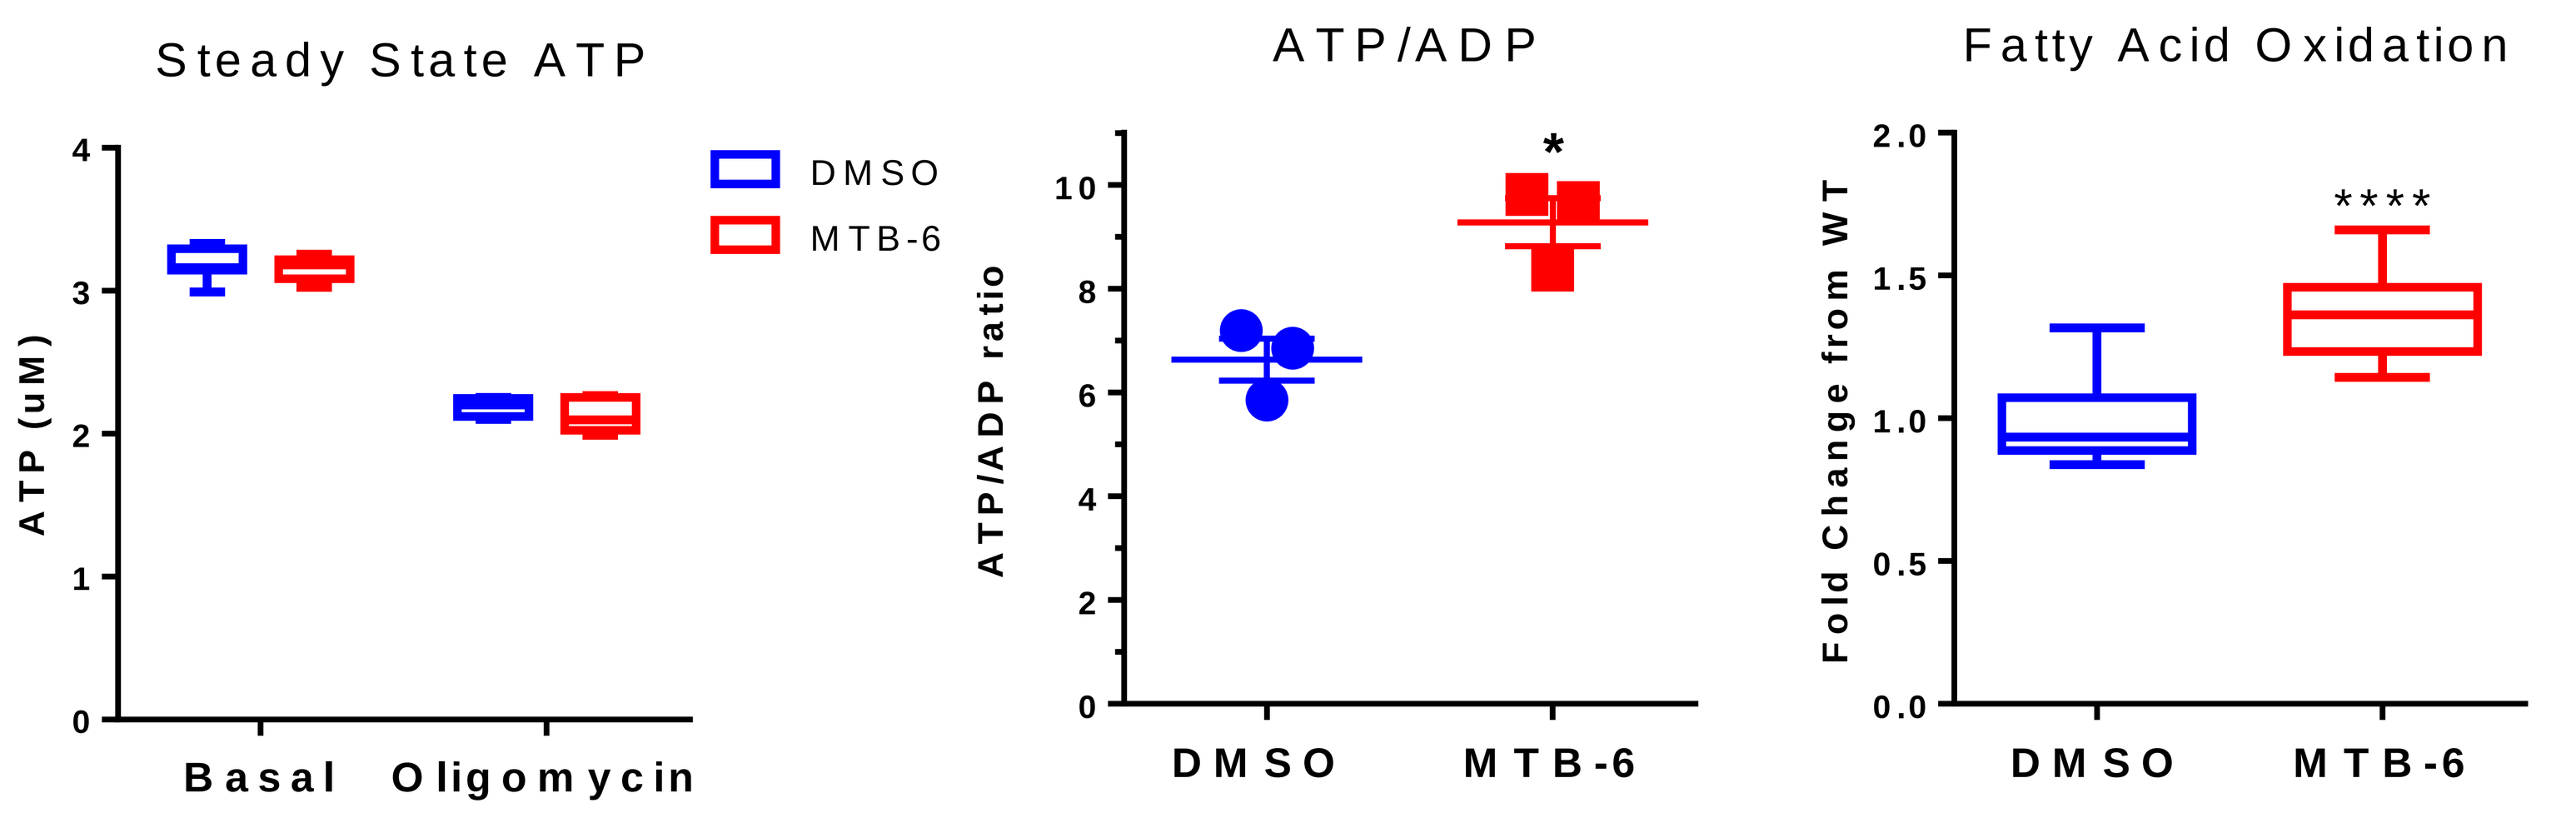
Steady State ATP
ATP (uM)
DMSO
MTB-6
Basal
Oligomycin
ATP/ADP
ATP/ADP ratio
Fatty Acid Oxidation
Fold Change from WT

Figure 7- Treatment of mdx myoblasts with the PPARδ modulator MTB-6 improves mitochondrial defects.

Conclusions and Future Directions

We have demonstrated that the mdx derived muscle cells exhibit significant deficit in mitochondrial function, which are apparent in myoblasts, before myogenesis has completed. These defects present as decreased membrane potential, reduced cellular ATP, and inefficient oxidation of various substrates. Mitochondrial abundance is decreased in mdx derived muscle cells as are transcripts encoding mitochondrial electron transport chain subunits, fatty acid oxidation genes, and master regulators of mitochondrial biogenesis. The functional deficit in mitochondrial output could be due to reduced mitochondrial biomass and/or a specific defect in the mitochondria present.

PPARδ expression in mdx derived muscle cells is increased; however, the putative target genes that are indicative of activity are not increased. Treatment of mdx myoblasts with a novel PPARδ modulator, MTB-6, increases the ATP/ADP ratio and fatty acid oxidation. Future experiments will focus on elucidating the mechanism for decreased functionality and assess the impact of mitochondrial deficits on the dystrophic muscle isolated from mice and humans.

References

[1]Schuh RA, et al. *Am J Physiol Regul Integr Comp Physiol.* 302(6):R712-9. 2012.
[2]Rybalka E, et al. *PLoS One.* 9(12):e115763. 2014.
[3]Godin R, et al. *J. Physiol.* 590(21):5487-502. 2012.

mitobridge