PPARδ modulator MTB-2 enhances FAO in vitro and attenuates IR-induced gene expression changes in vivo, 48 hours and 14 days post AKI

Christina Bracken, Jeff Stanwix, Hien Hoang, Eric Bell, Effie Tozzo
Mitobridge, Inc. Cambridge, MA 02138

Background

Ischemic acute kidney injury (AKI) is characterized by persistent proximal tubule mitochondrial dysfunction. Due to their highly oxidative metabolism, proximal tubule cells utilize fatty acids to generate the energy required for their specialized function. We hypothesized that enhancing fatty acid oxidation with a potent and highly selective PPARδ modulator will restore mitochondrial function, offering a potential therapeutic treatment for AKI.

Methods and Analysis

In vitro assays: Human hTERT RPTECs were treated with 500nM MTB-2 for 24 hours and analyzed for gene expression by qPCR and their ability to utilize palmitate to drive mitochondrial oxygen consumption by Seahorse. **Animal model:** Sprague-Dawley (SD) rats underwent 45 minutes of bilateral renal pedicle ischemia followed by reperfusion. Post-reperfusion, MTB-2 or vehicle was dosed IV QD for 2 days; termination was at either 48 hours or 14 days. Renal function was measured by plasma creatinine and GFR estimated by creatinine clearance. **Kidney cortex gene expression** was analyzed by either qPCR or nanostring.

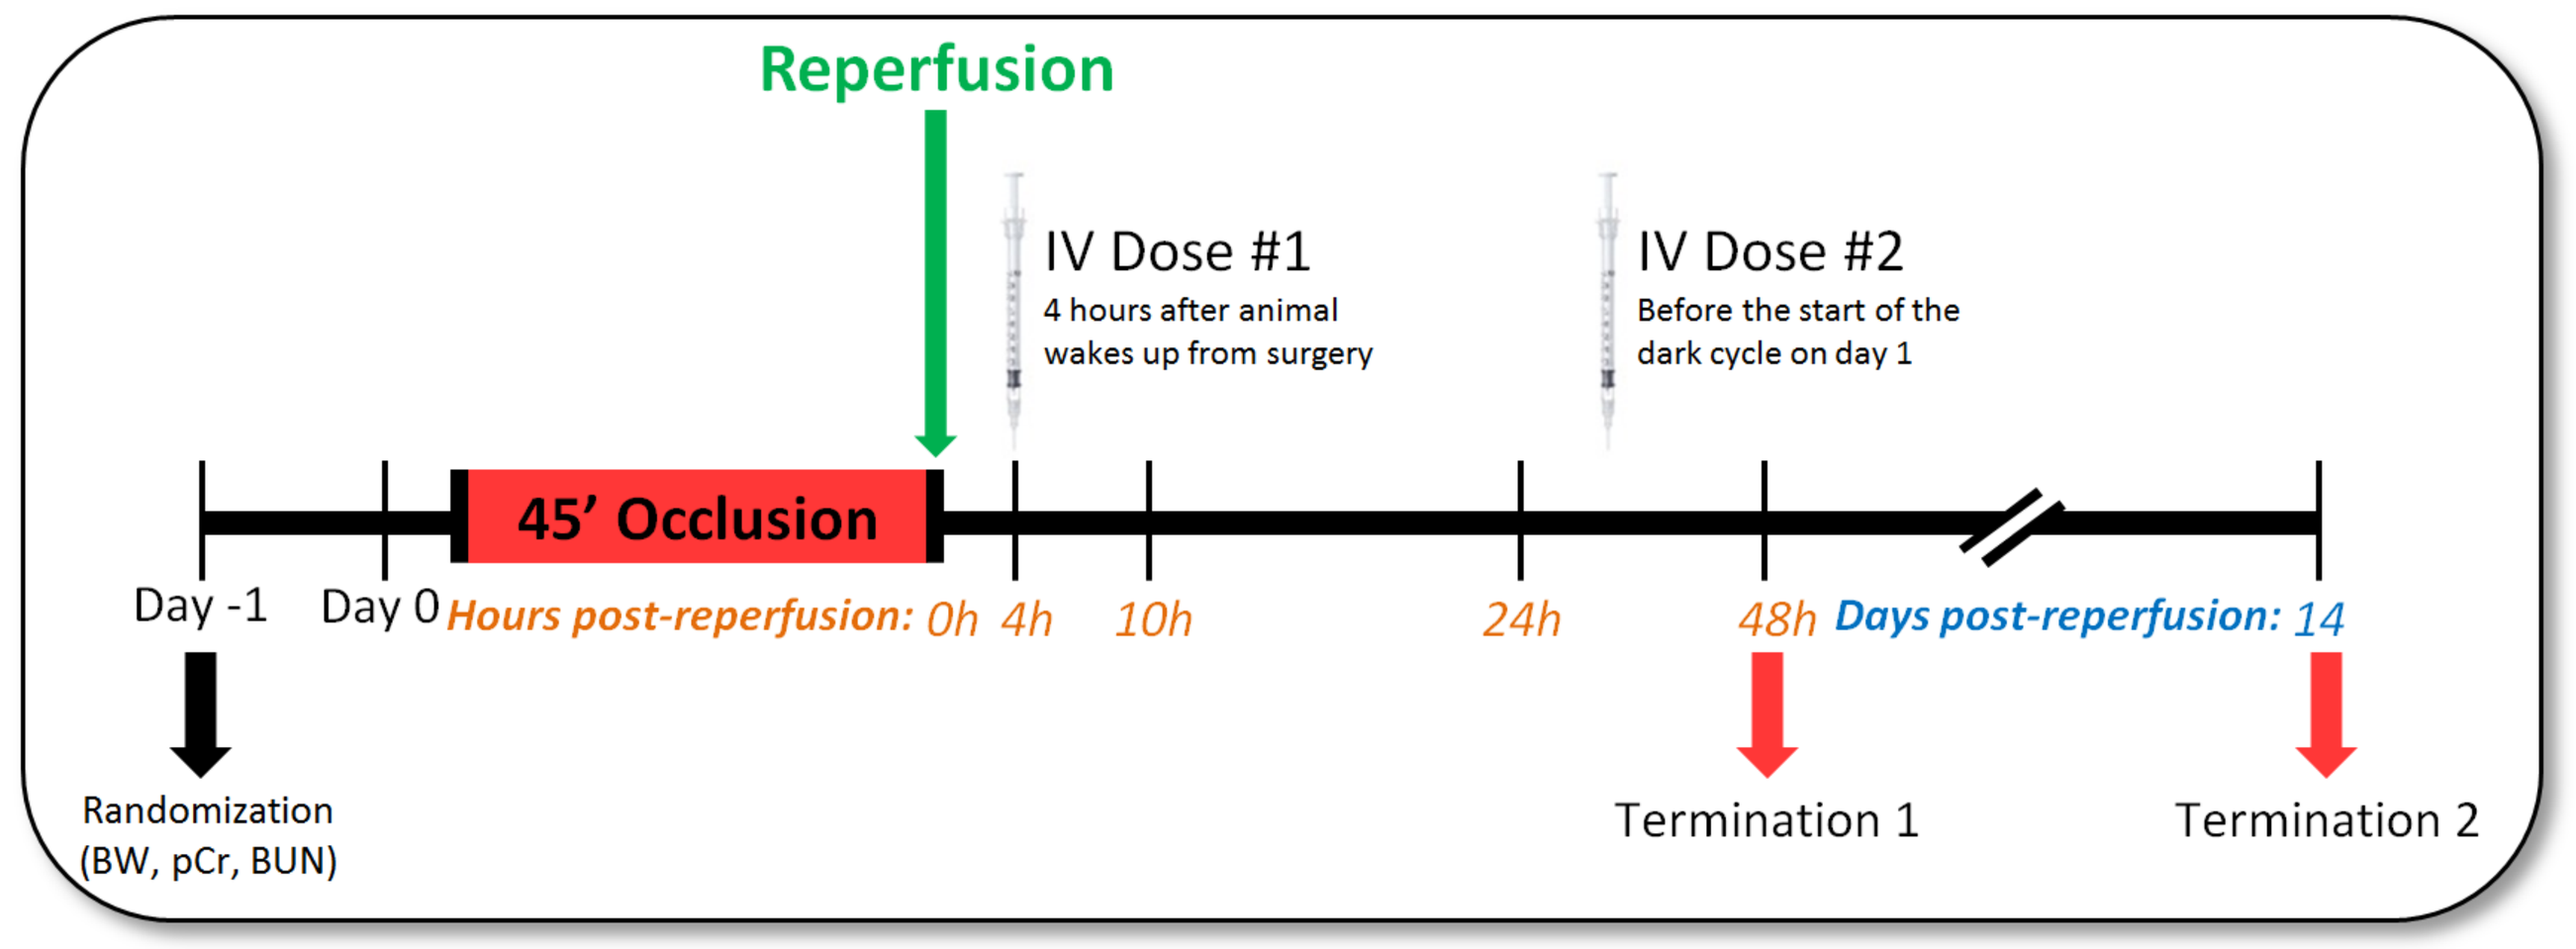
Reperfusion
IV Dose #1
4 hours after animal wakes up from surgery
IV Dose #2
Before the start of the dark cycle on day 1
45' Occlusion
Day -1
Day 0
Hours post-reperfusion: 0h 4h 10h 24h 48h
Days post-reperfusion: 14
Randomization (BW, pCr, BUN)
Termination 1
Termination 2

MTB-2 PK/PD Relationship

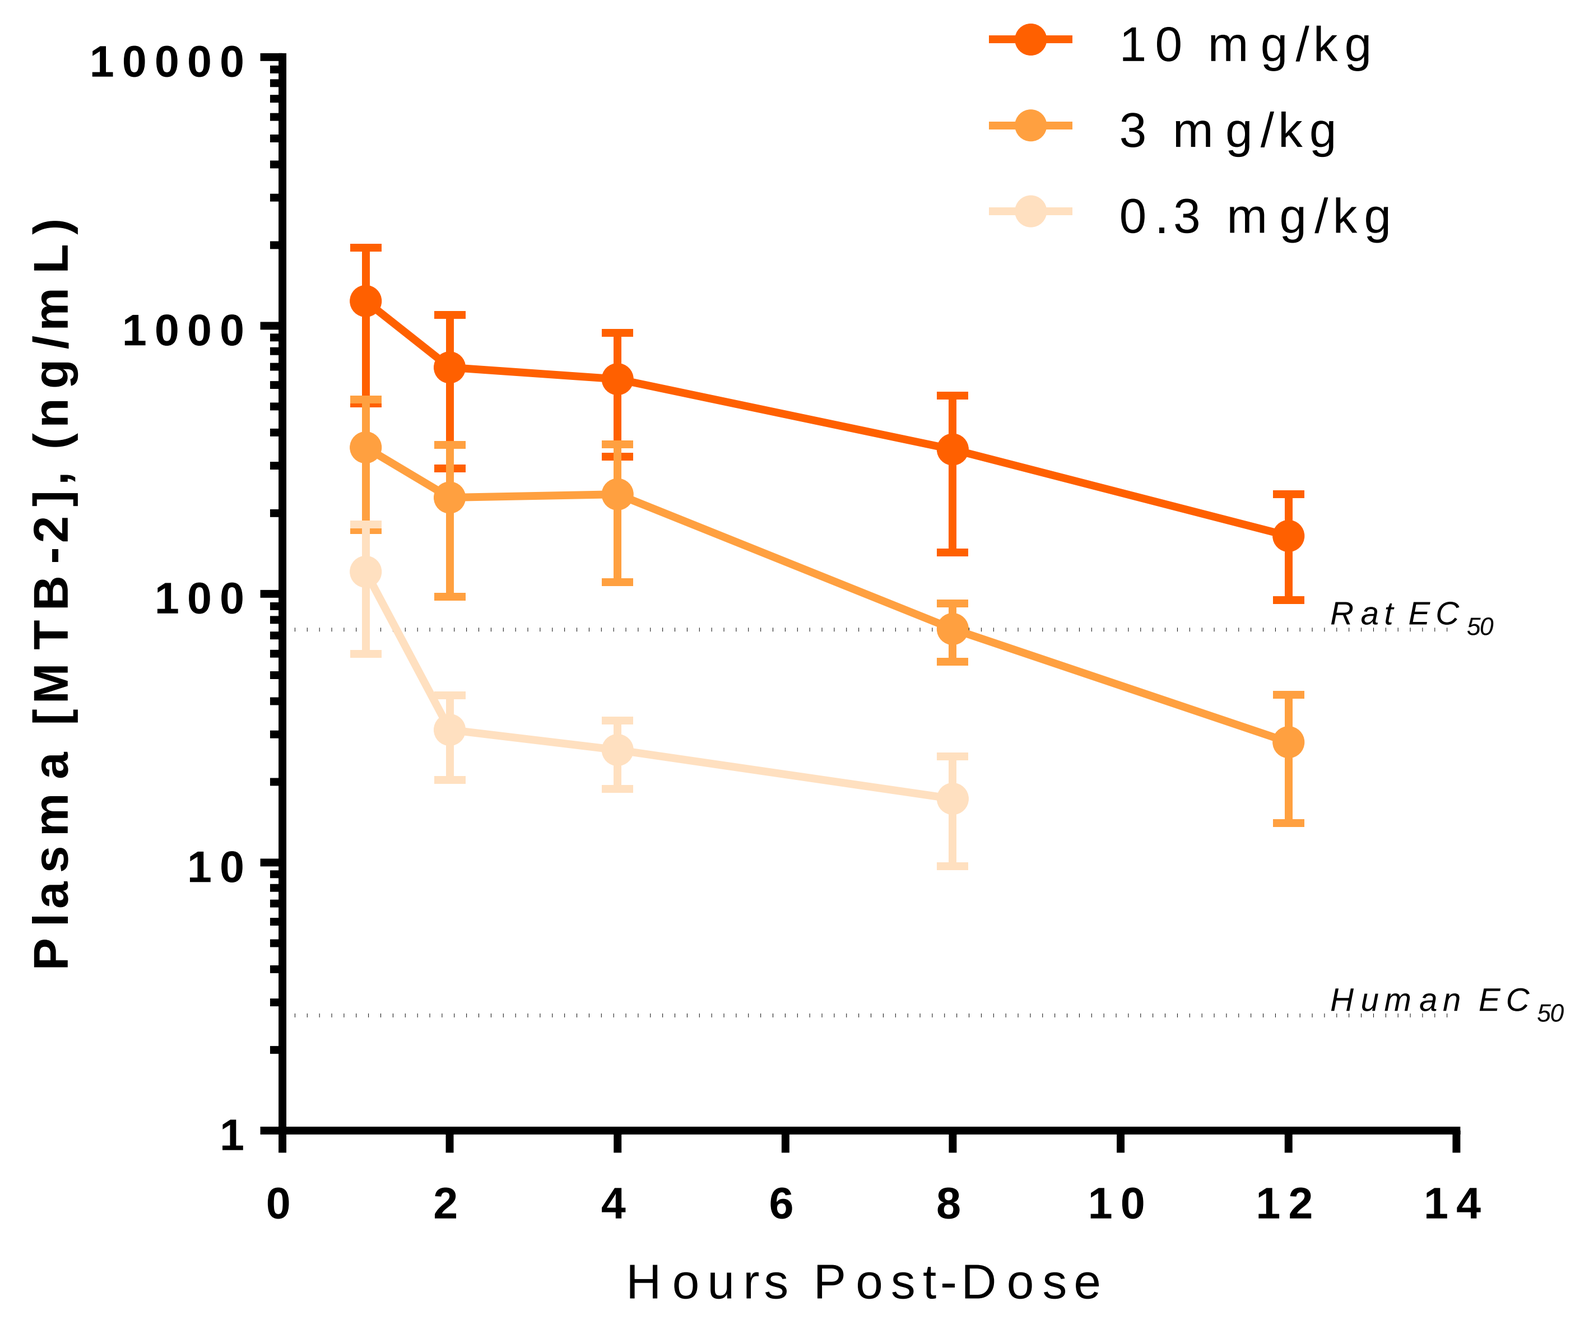
10 mg/kg
3 mg/kg
0.3 mg/kg
Plasma [MTB-2], (ng/mL)
Rat EC50
Human EC50
Hours Post-Dose

Drug concentration and target engagement were measured in normal rats administered a single IV dose of MTB-2. Exposure of MTB-2 in plasma is above Human EC_{50} at all doses tested for up to 8 hours. Kidney exposure is equivalent to plasma (not shown). This translates to target engagement for up to 12 hours post dose.

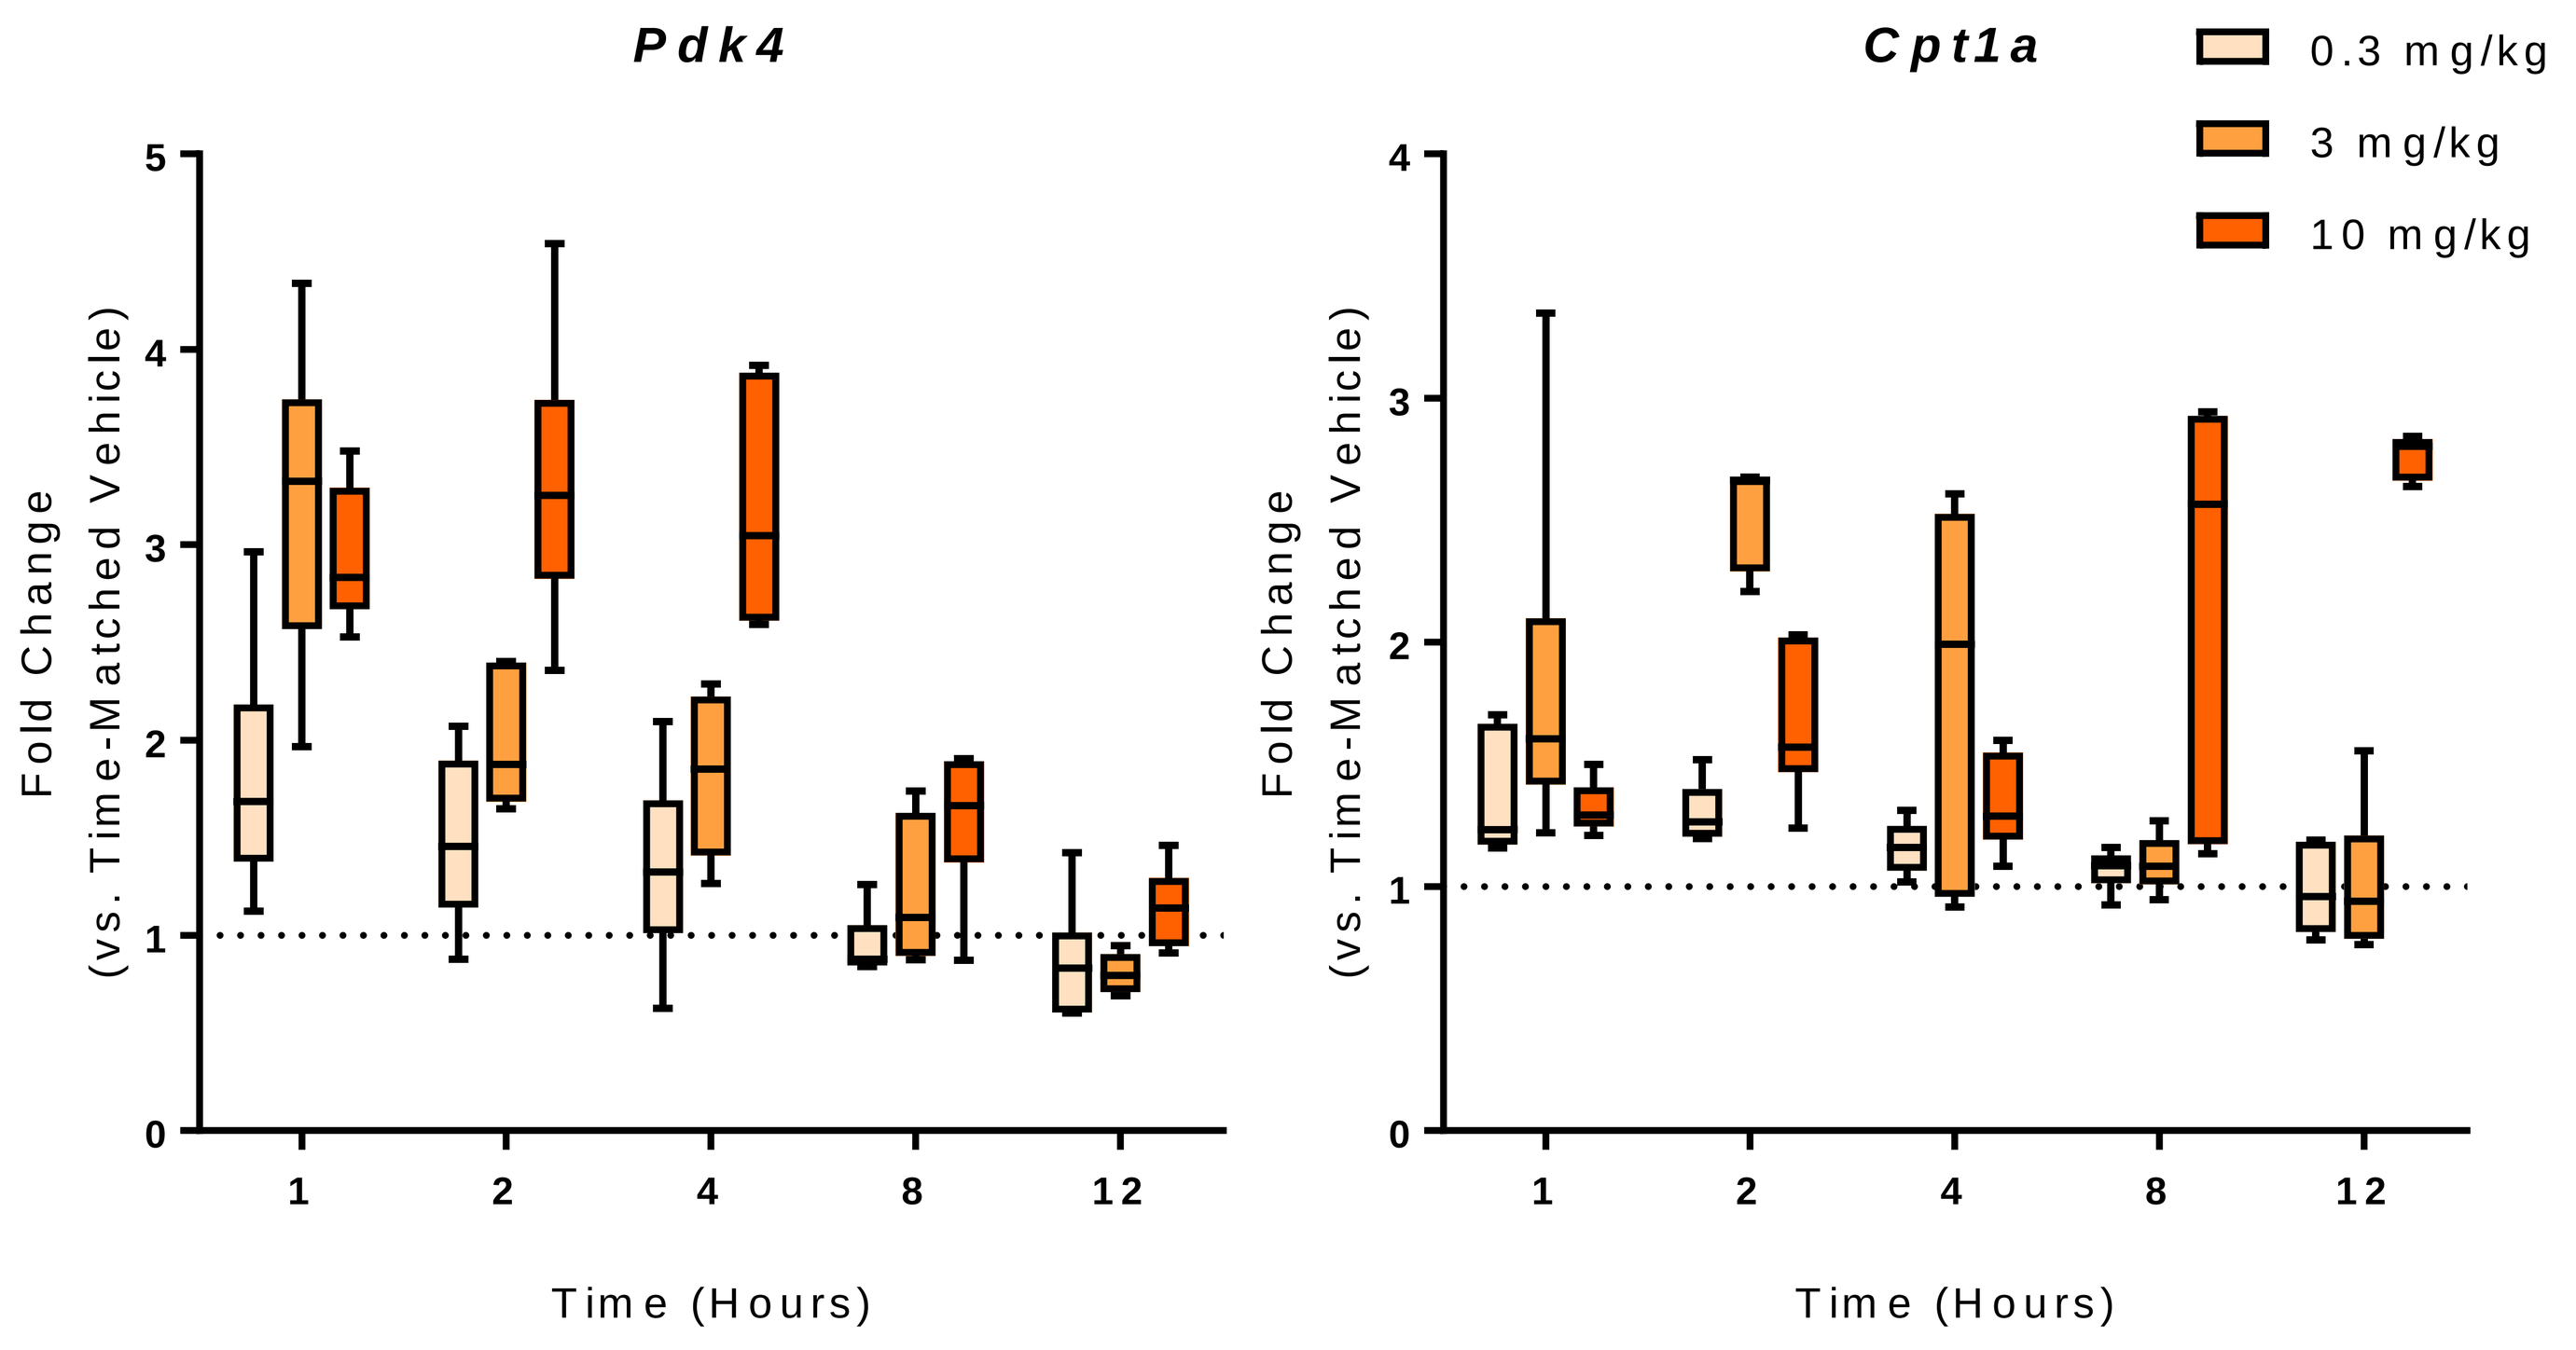
Pdk4
Cpt1a
0.3 mg/kg
3 mg/kg
10 mg/kg
Fold Change (vs. Time-Matched Vehicle)
Time (Hours)

MTB-2 enhances fatty acid oxidation

FAO Gene Expression

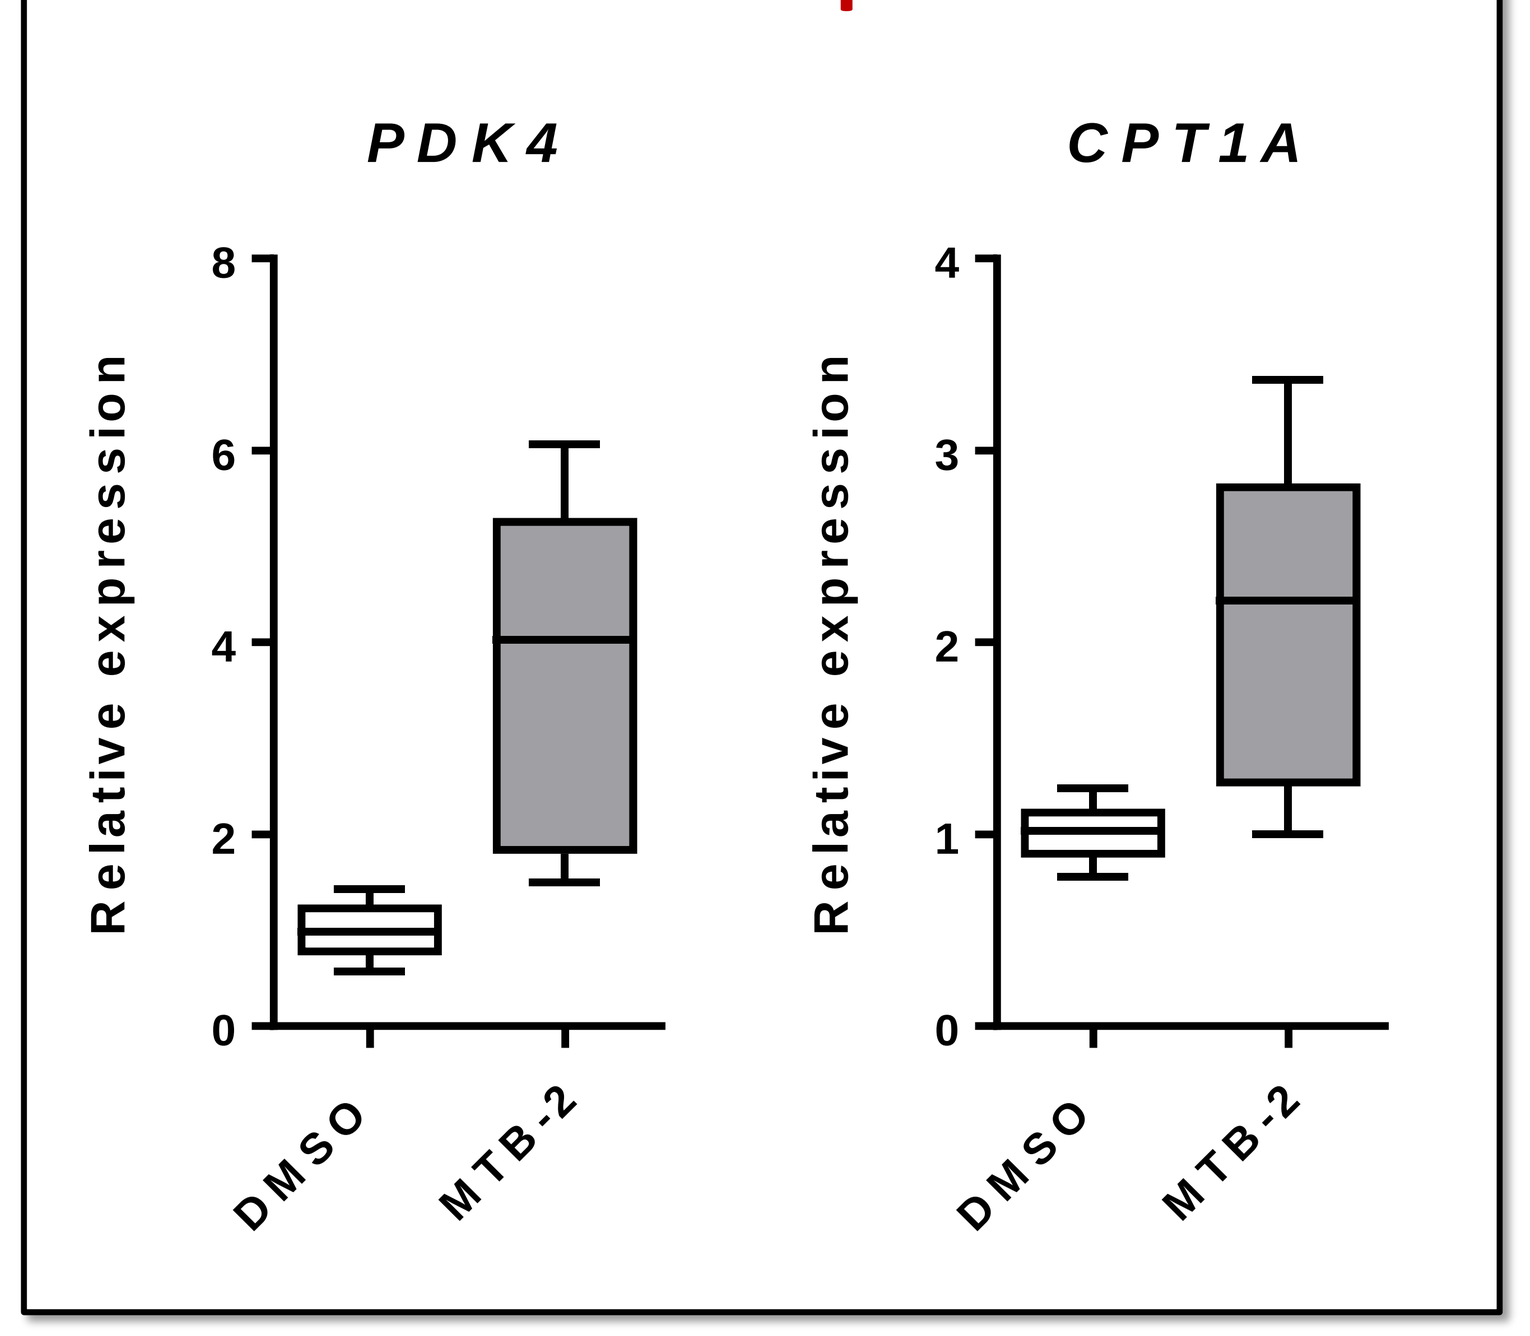
PDK4
CPT1A
Relative expression
DMSO
MTB-2

Mitochondrial FAO

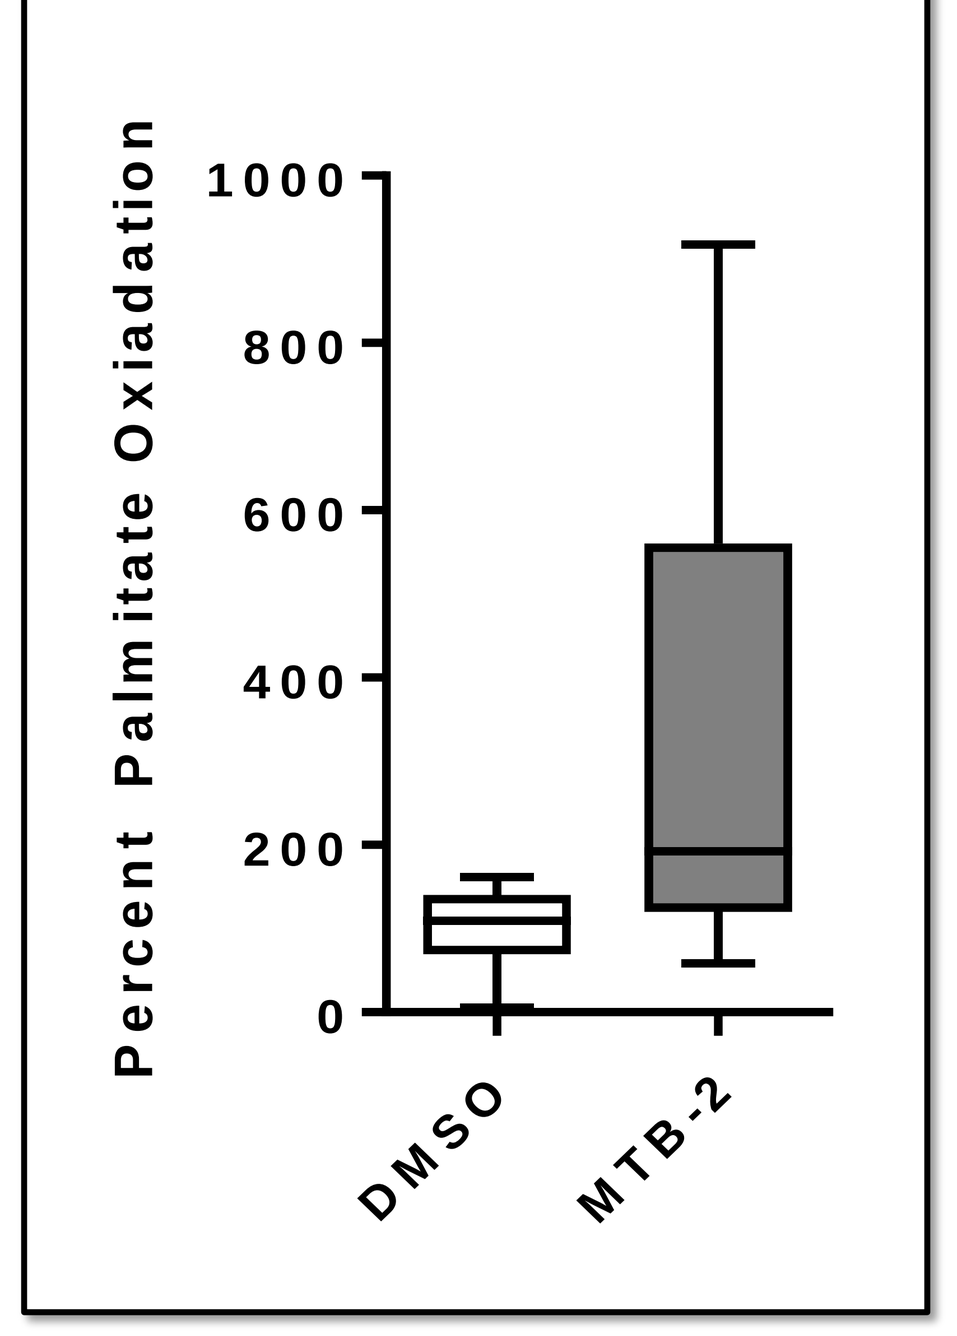
Percent Palmitate Oxiadation
DMSO
MTB-2

MTB-2 effectively attenuates AKI

Plasma Creatinine

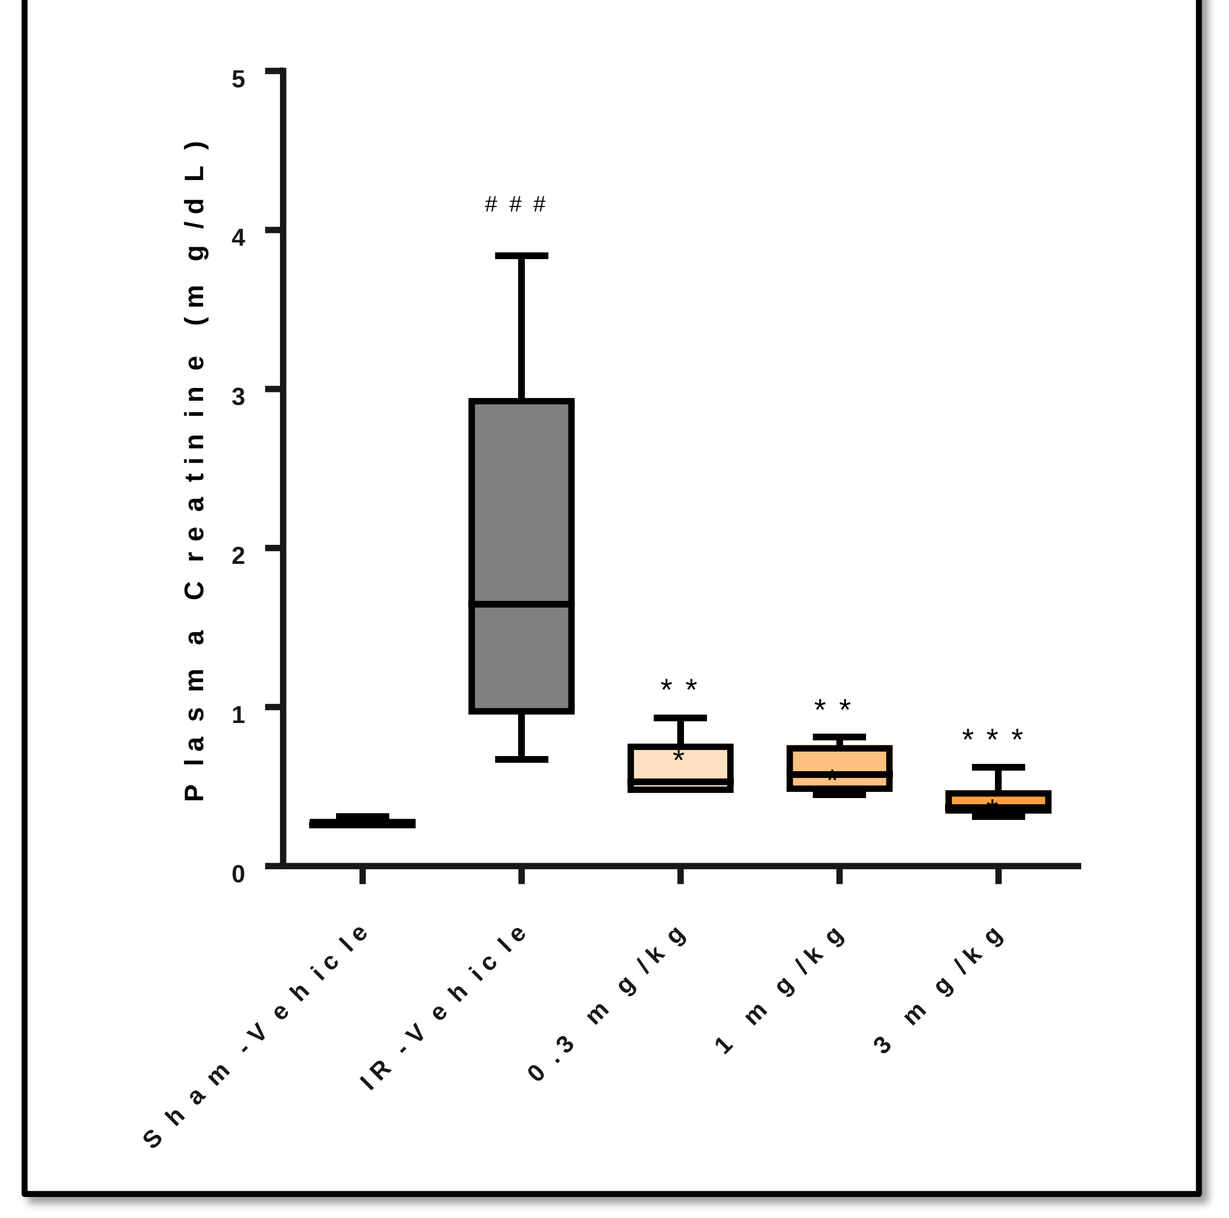
Plasma Creatinine (mg/dL)
Sham-Vehicle
IR-Vehicle
0.3 mg/kg
1 mg/kg
3 mg/kg

Creatinine Clearance

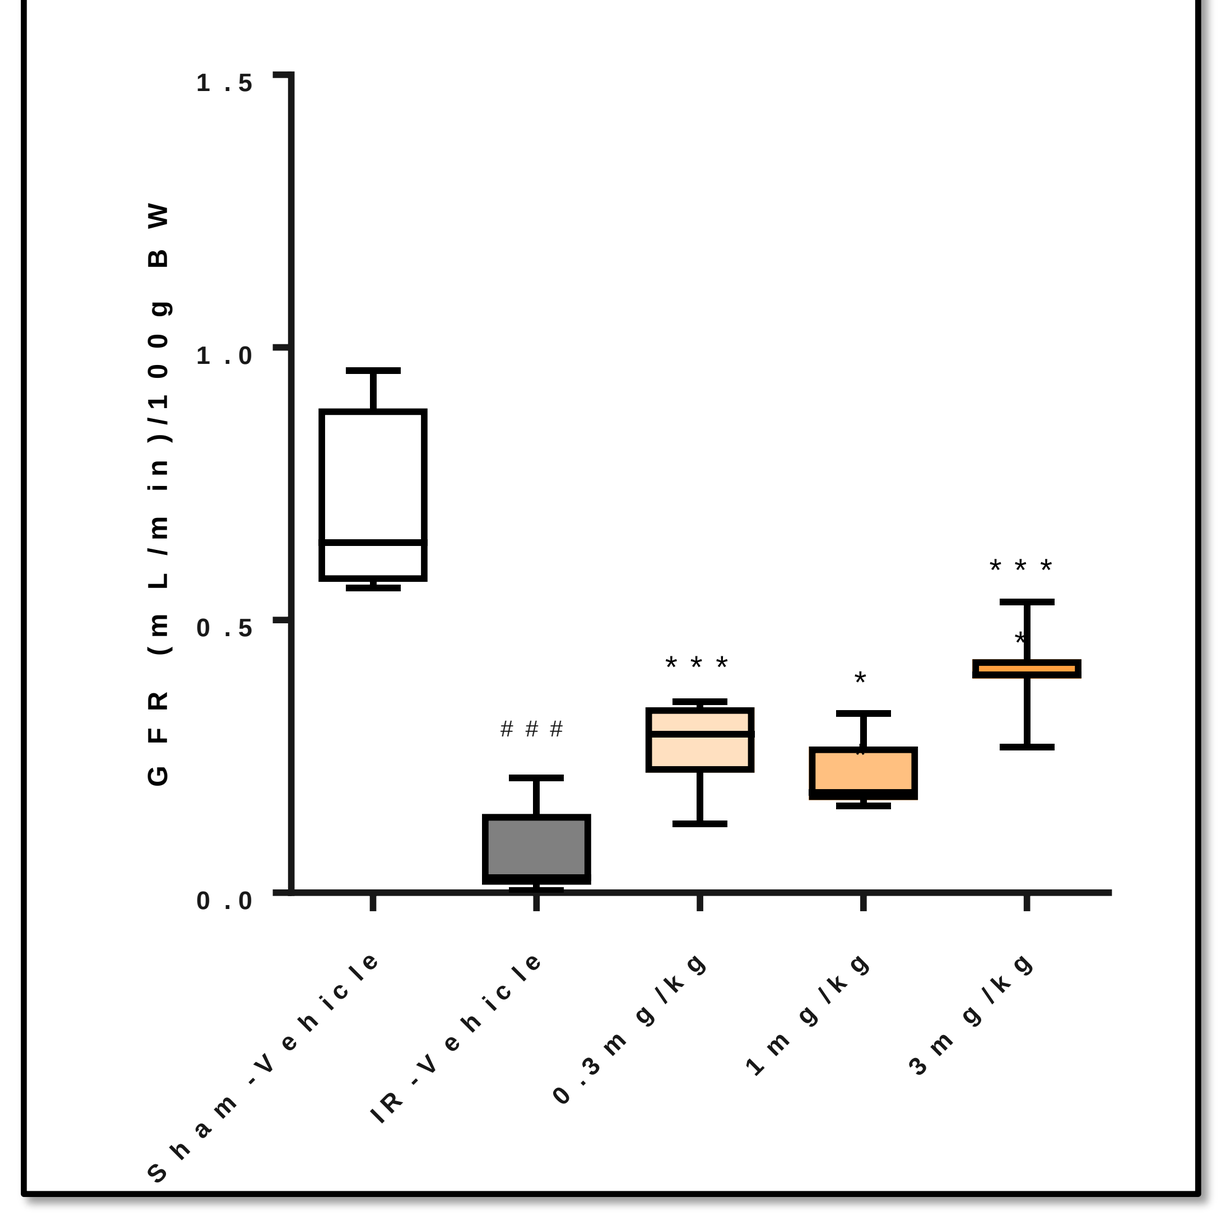
GFR (mL/min)/100g BW
Sham-Vehicle
IR-Vehicle
0.3 mg/kg
1 mg/kg
3 mg/kg

MTB-2 rescues loss of proximal tubule genes, reduces kidney injury genes

Proximal Tubule Abundance

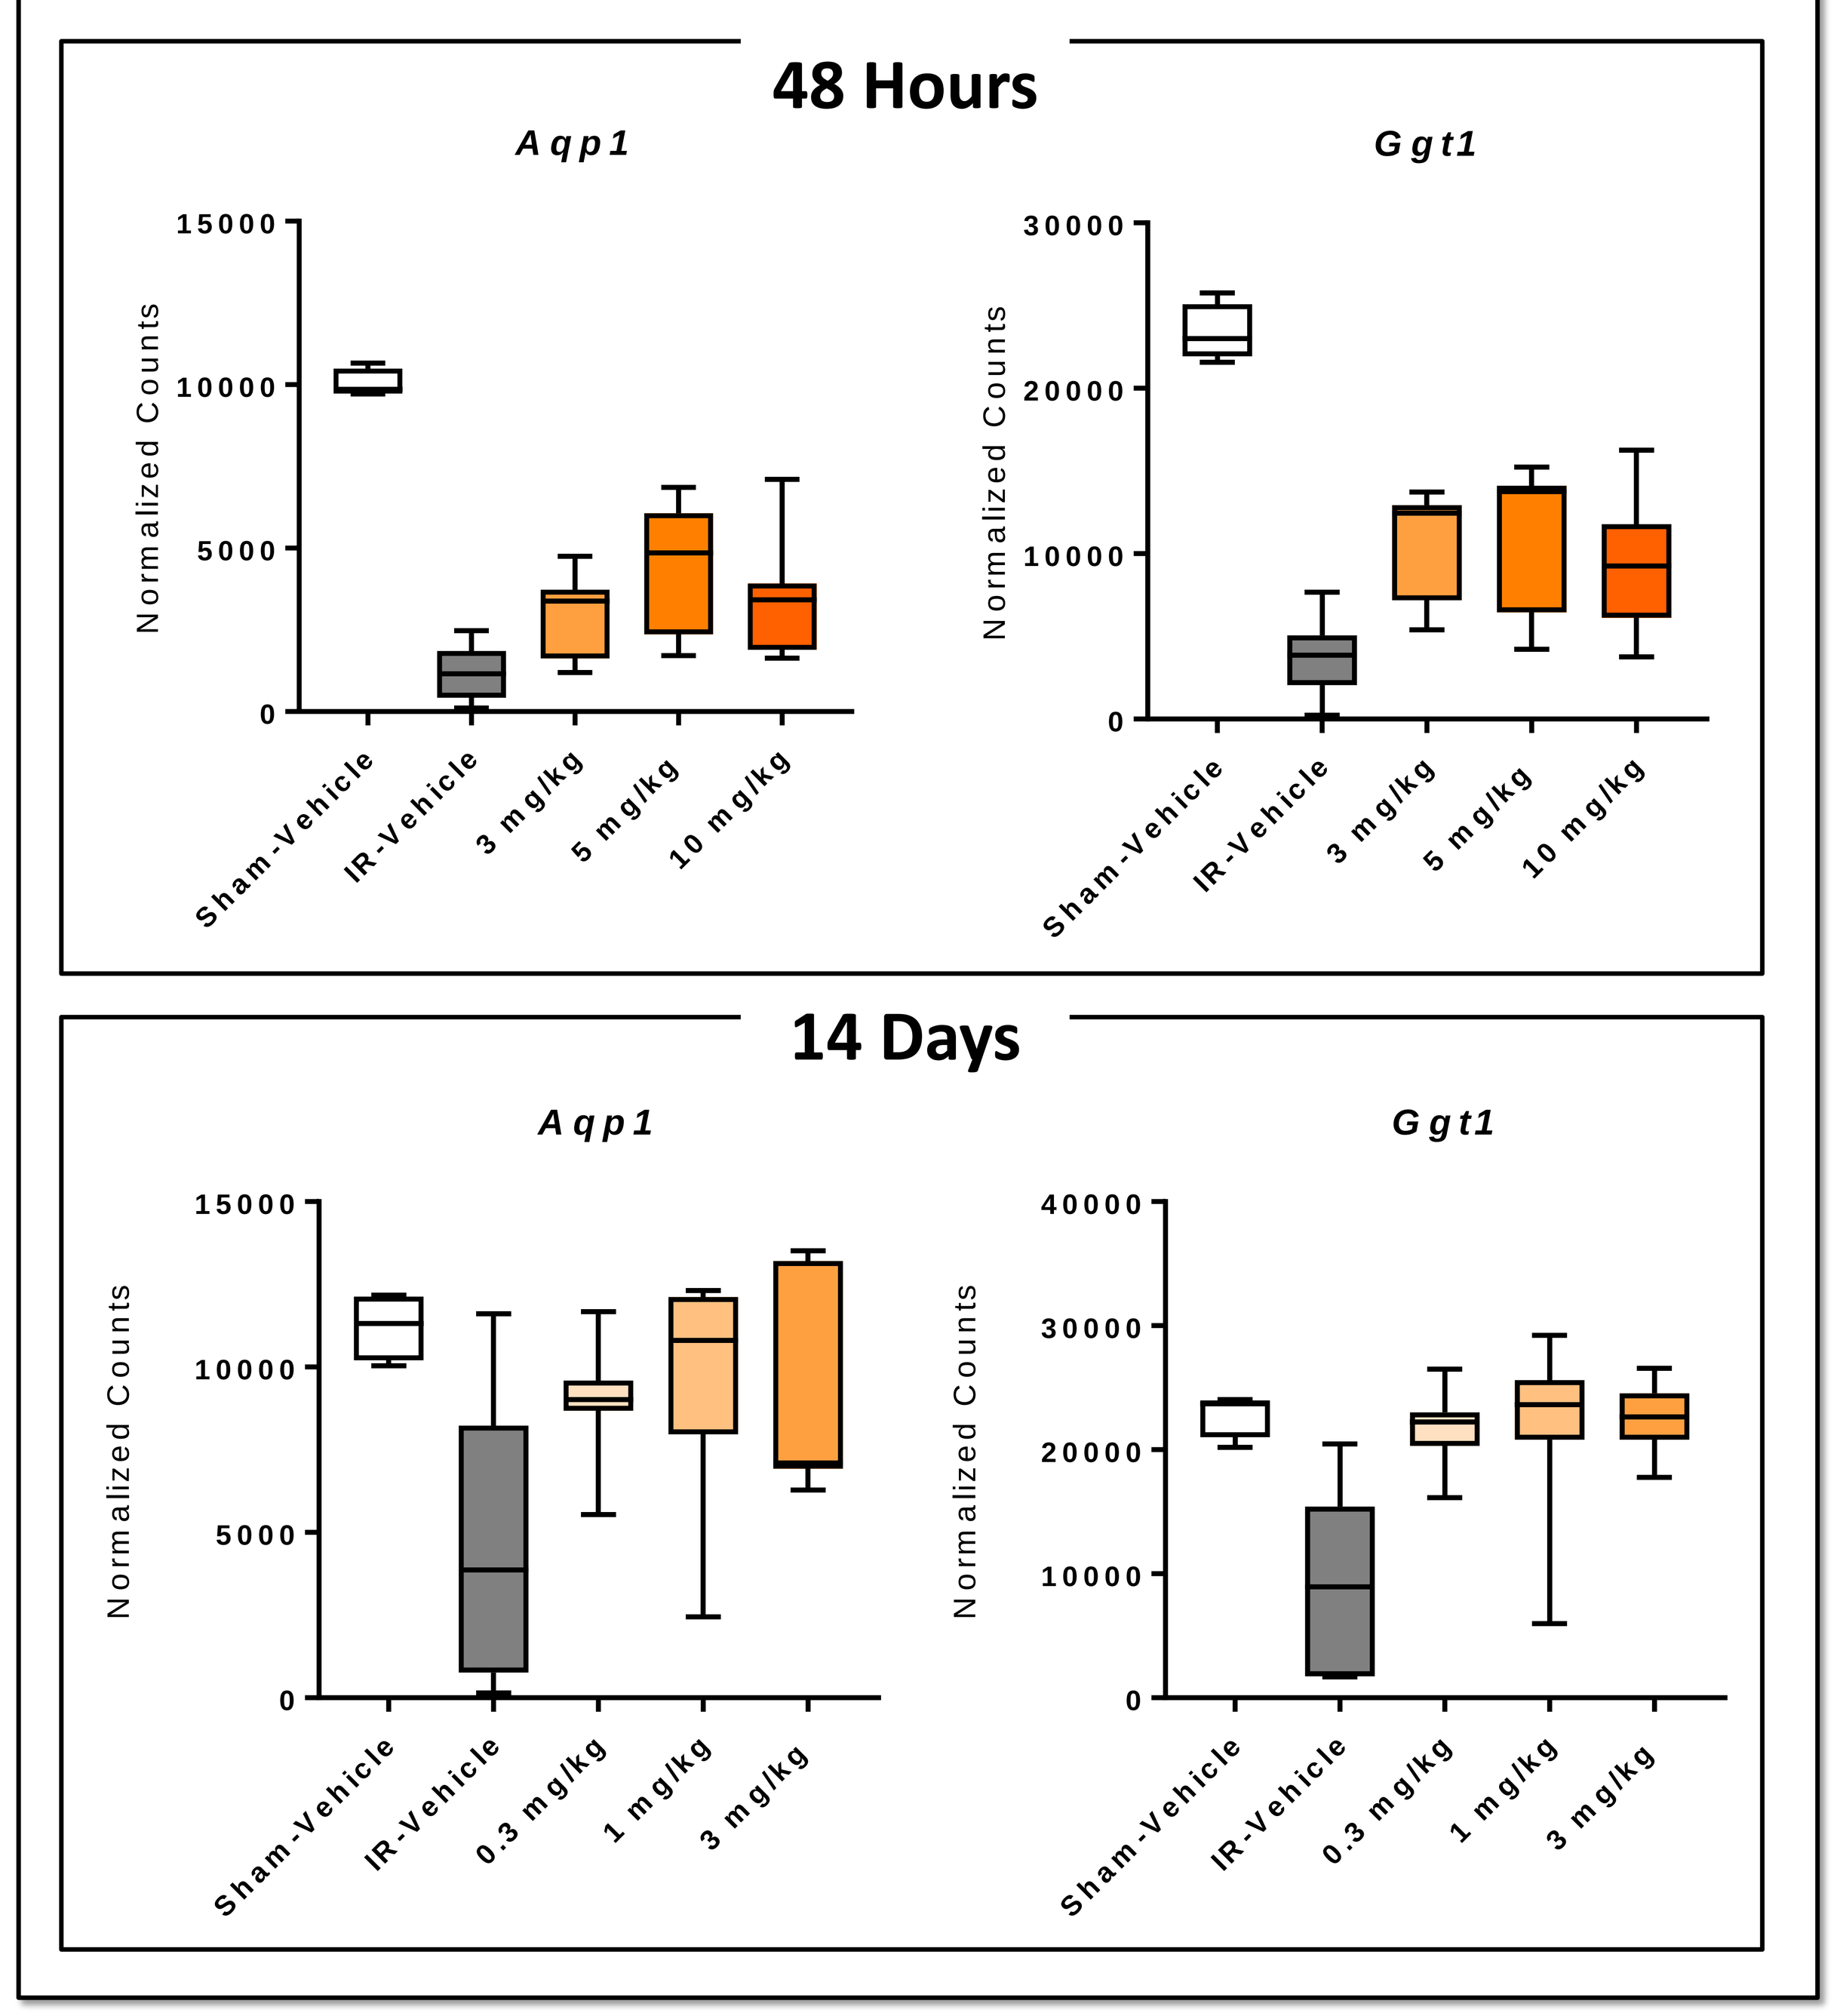
48 Hours
Aqp1
Ggt1
14 Days
Aqp1
Ggt1
Normalized Counts
Sham-Vehicle
IR-Vehicle
3 mg/kg
5 mg/kg
10 mg/kg
0.3 mg/kg
1 mg/kg

Kidney Injury

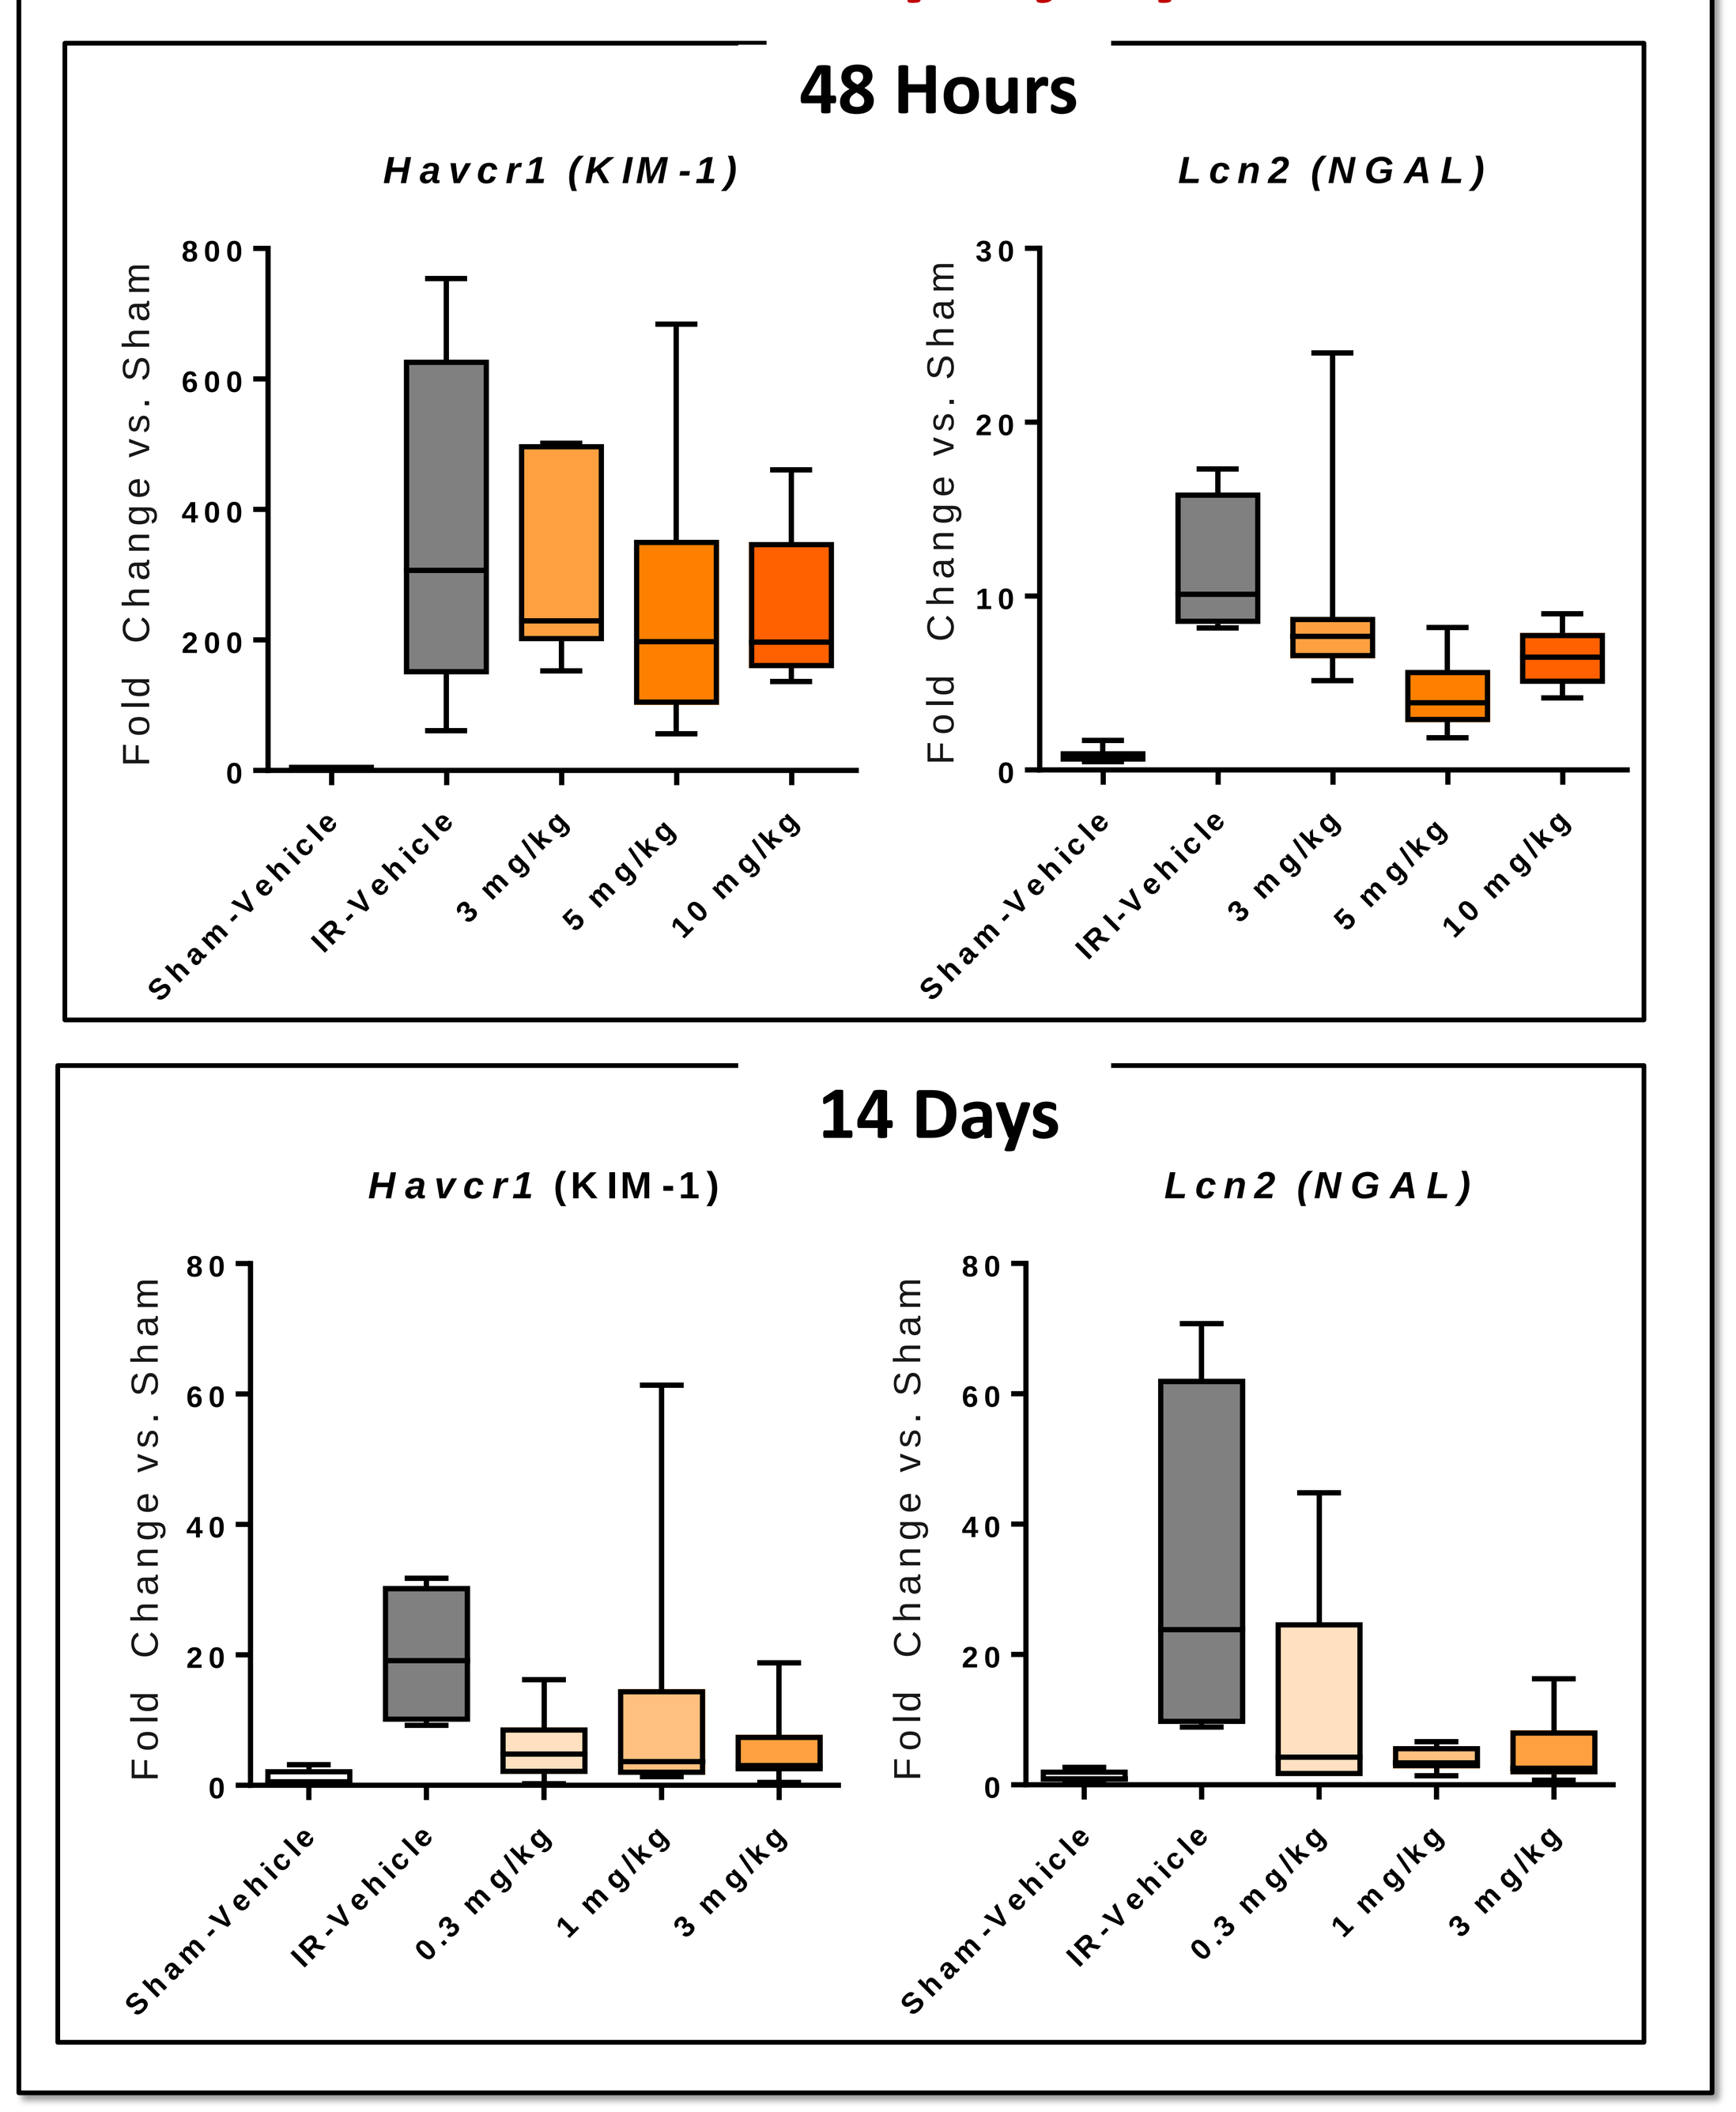
48 Hours
Havcr1 (KIM-1)
Lcn2 (NGAL)
14 Days
Havcr1 (KIM-1)
Lcn2 (NGAL)
Fold Change vs. Sham
Sham-Vehicle
IR-Vehicle
IRI-Vehicle
3 mg/kg
5 mg/kg
10 mg/kg
0.3 mg/kg
1 mg/kg

MTB-2 rescues expression of genes governing mitochondrial function and decreases fibrotic gene expression

Mitochondrial Homeostasis

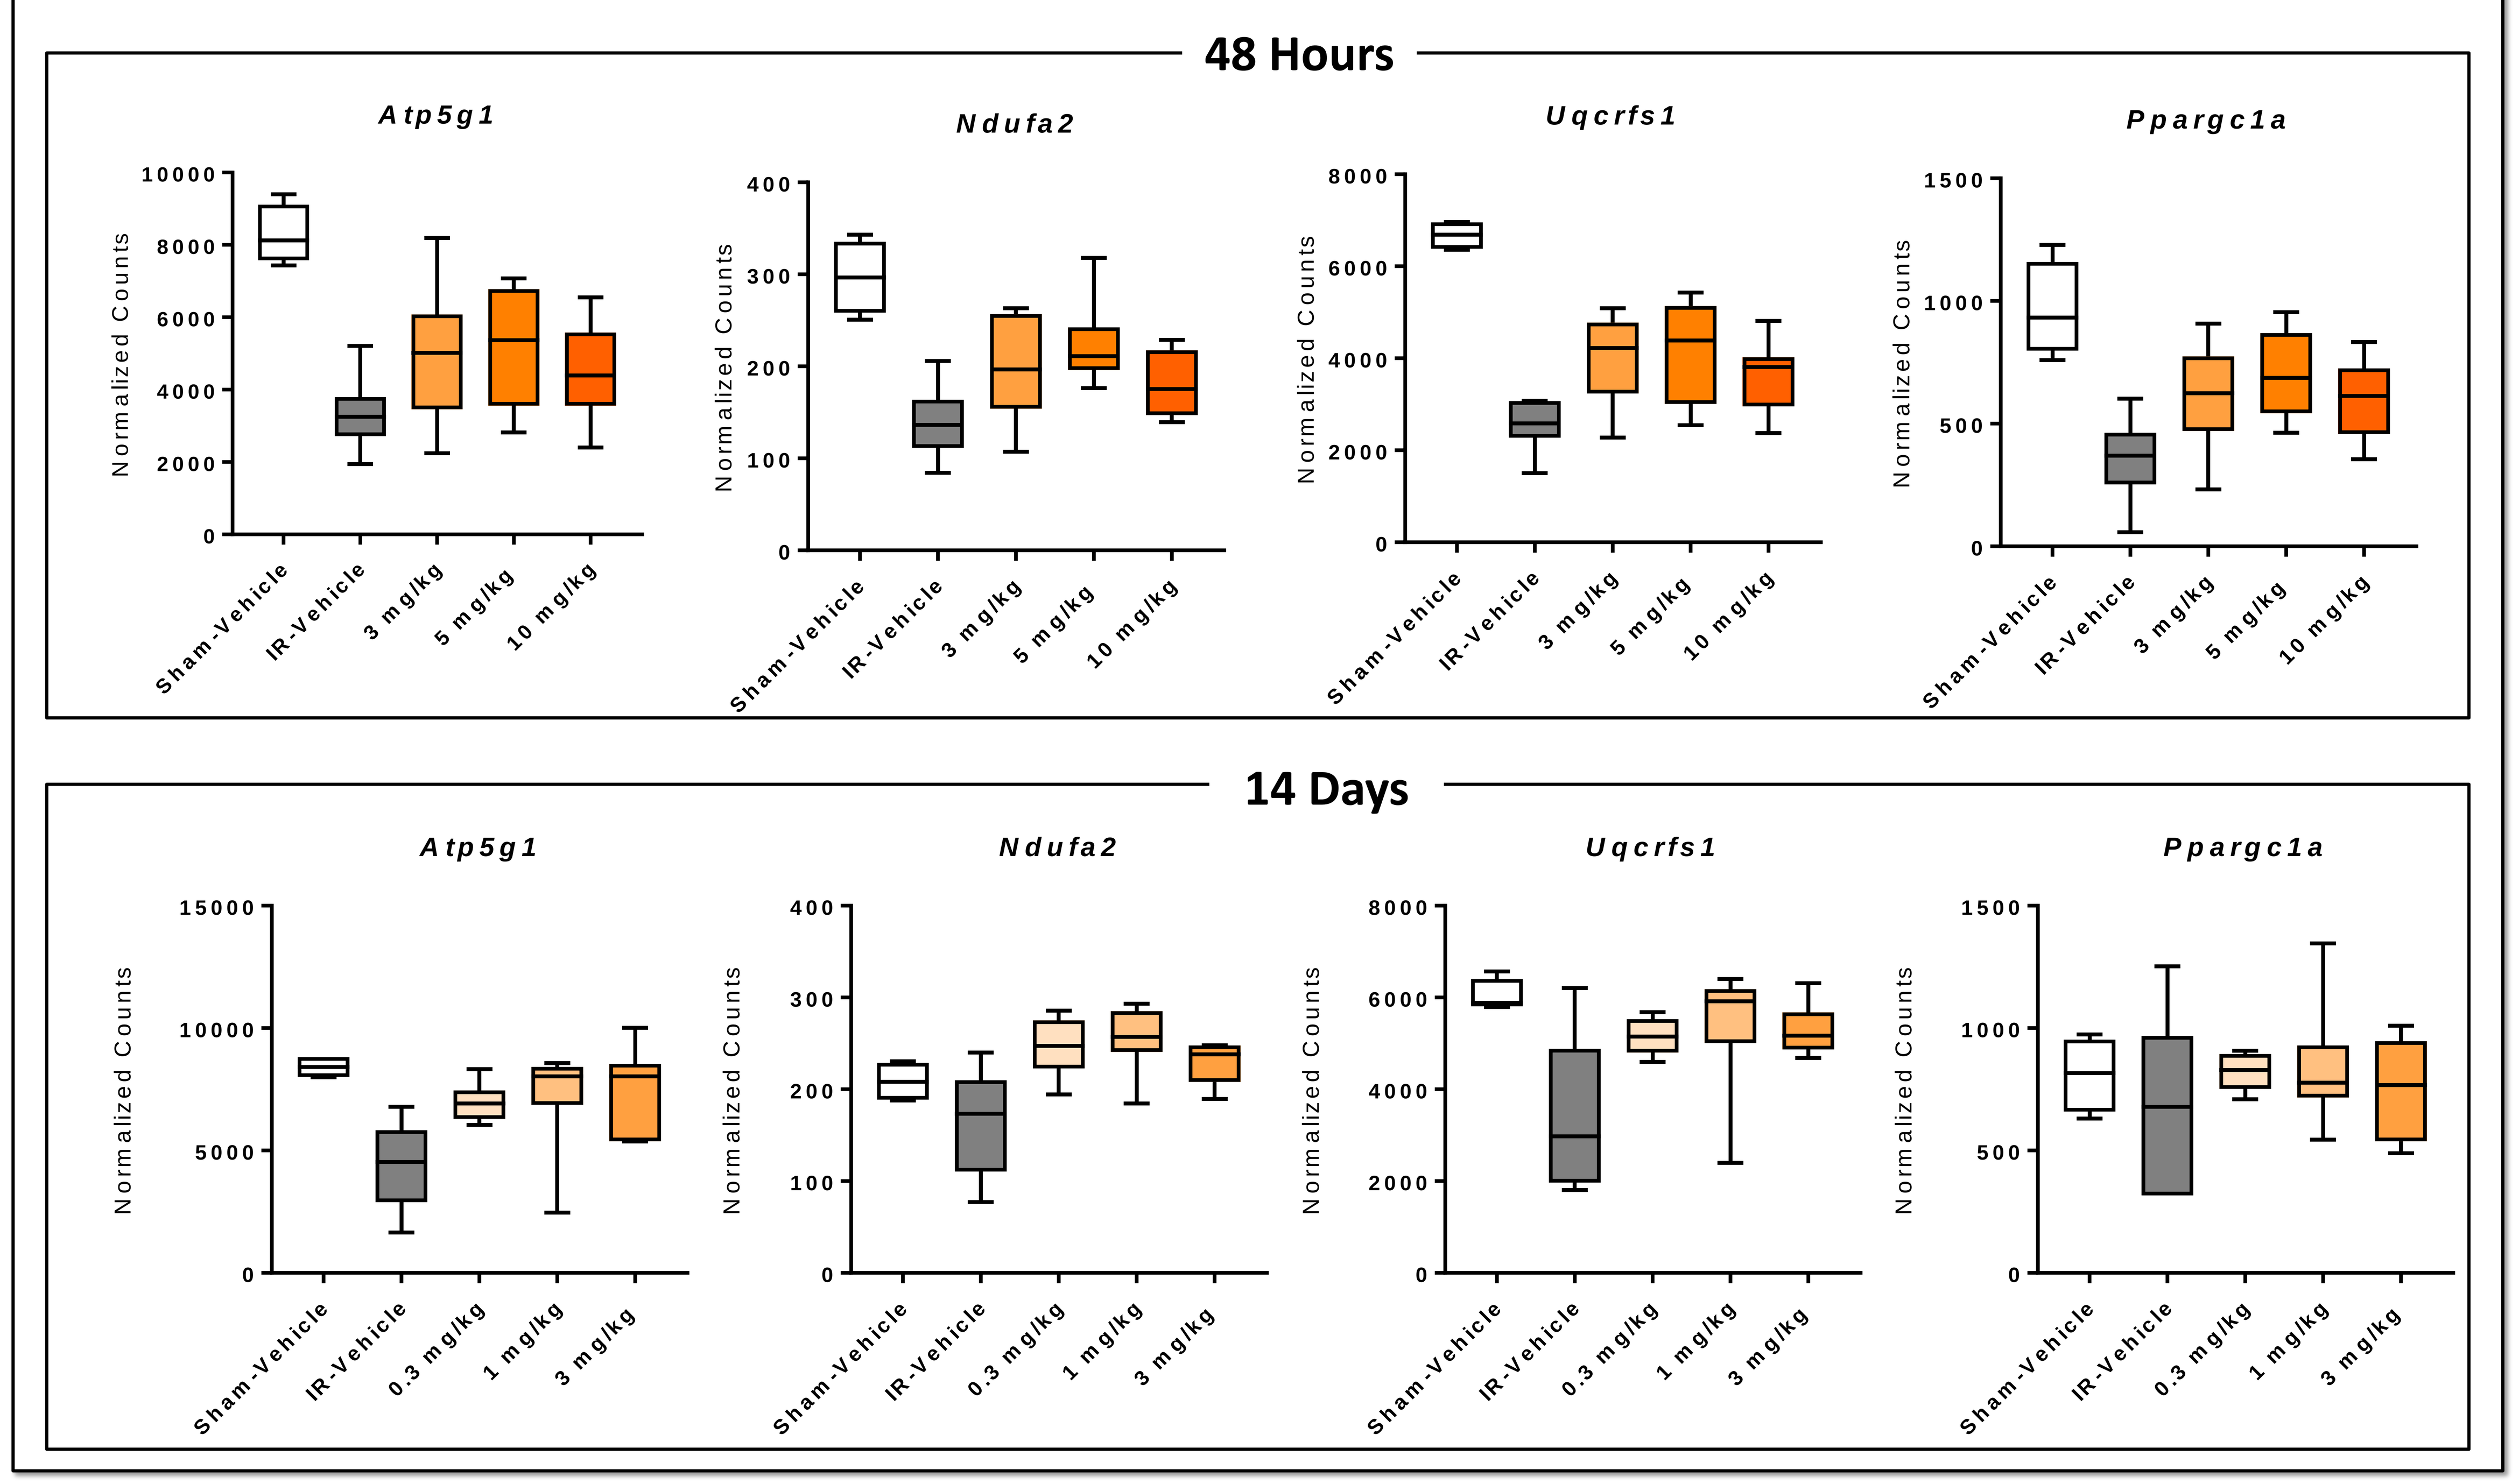
48 Hours
Atp5g1
Ndufa2
Uqcrfs1
Ppargc1a
14 Days
Atp5g1
Ndufa2
Uqcrfs1
Ppargc1a
Normalized Counts
Sham-Vehicle
IR-Vehicle
3 mg/kg
5 mg/kg
10 mg/kg
0.3 mg/kg
1 mg/kg

Fibrosis

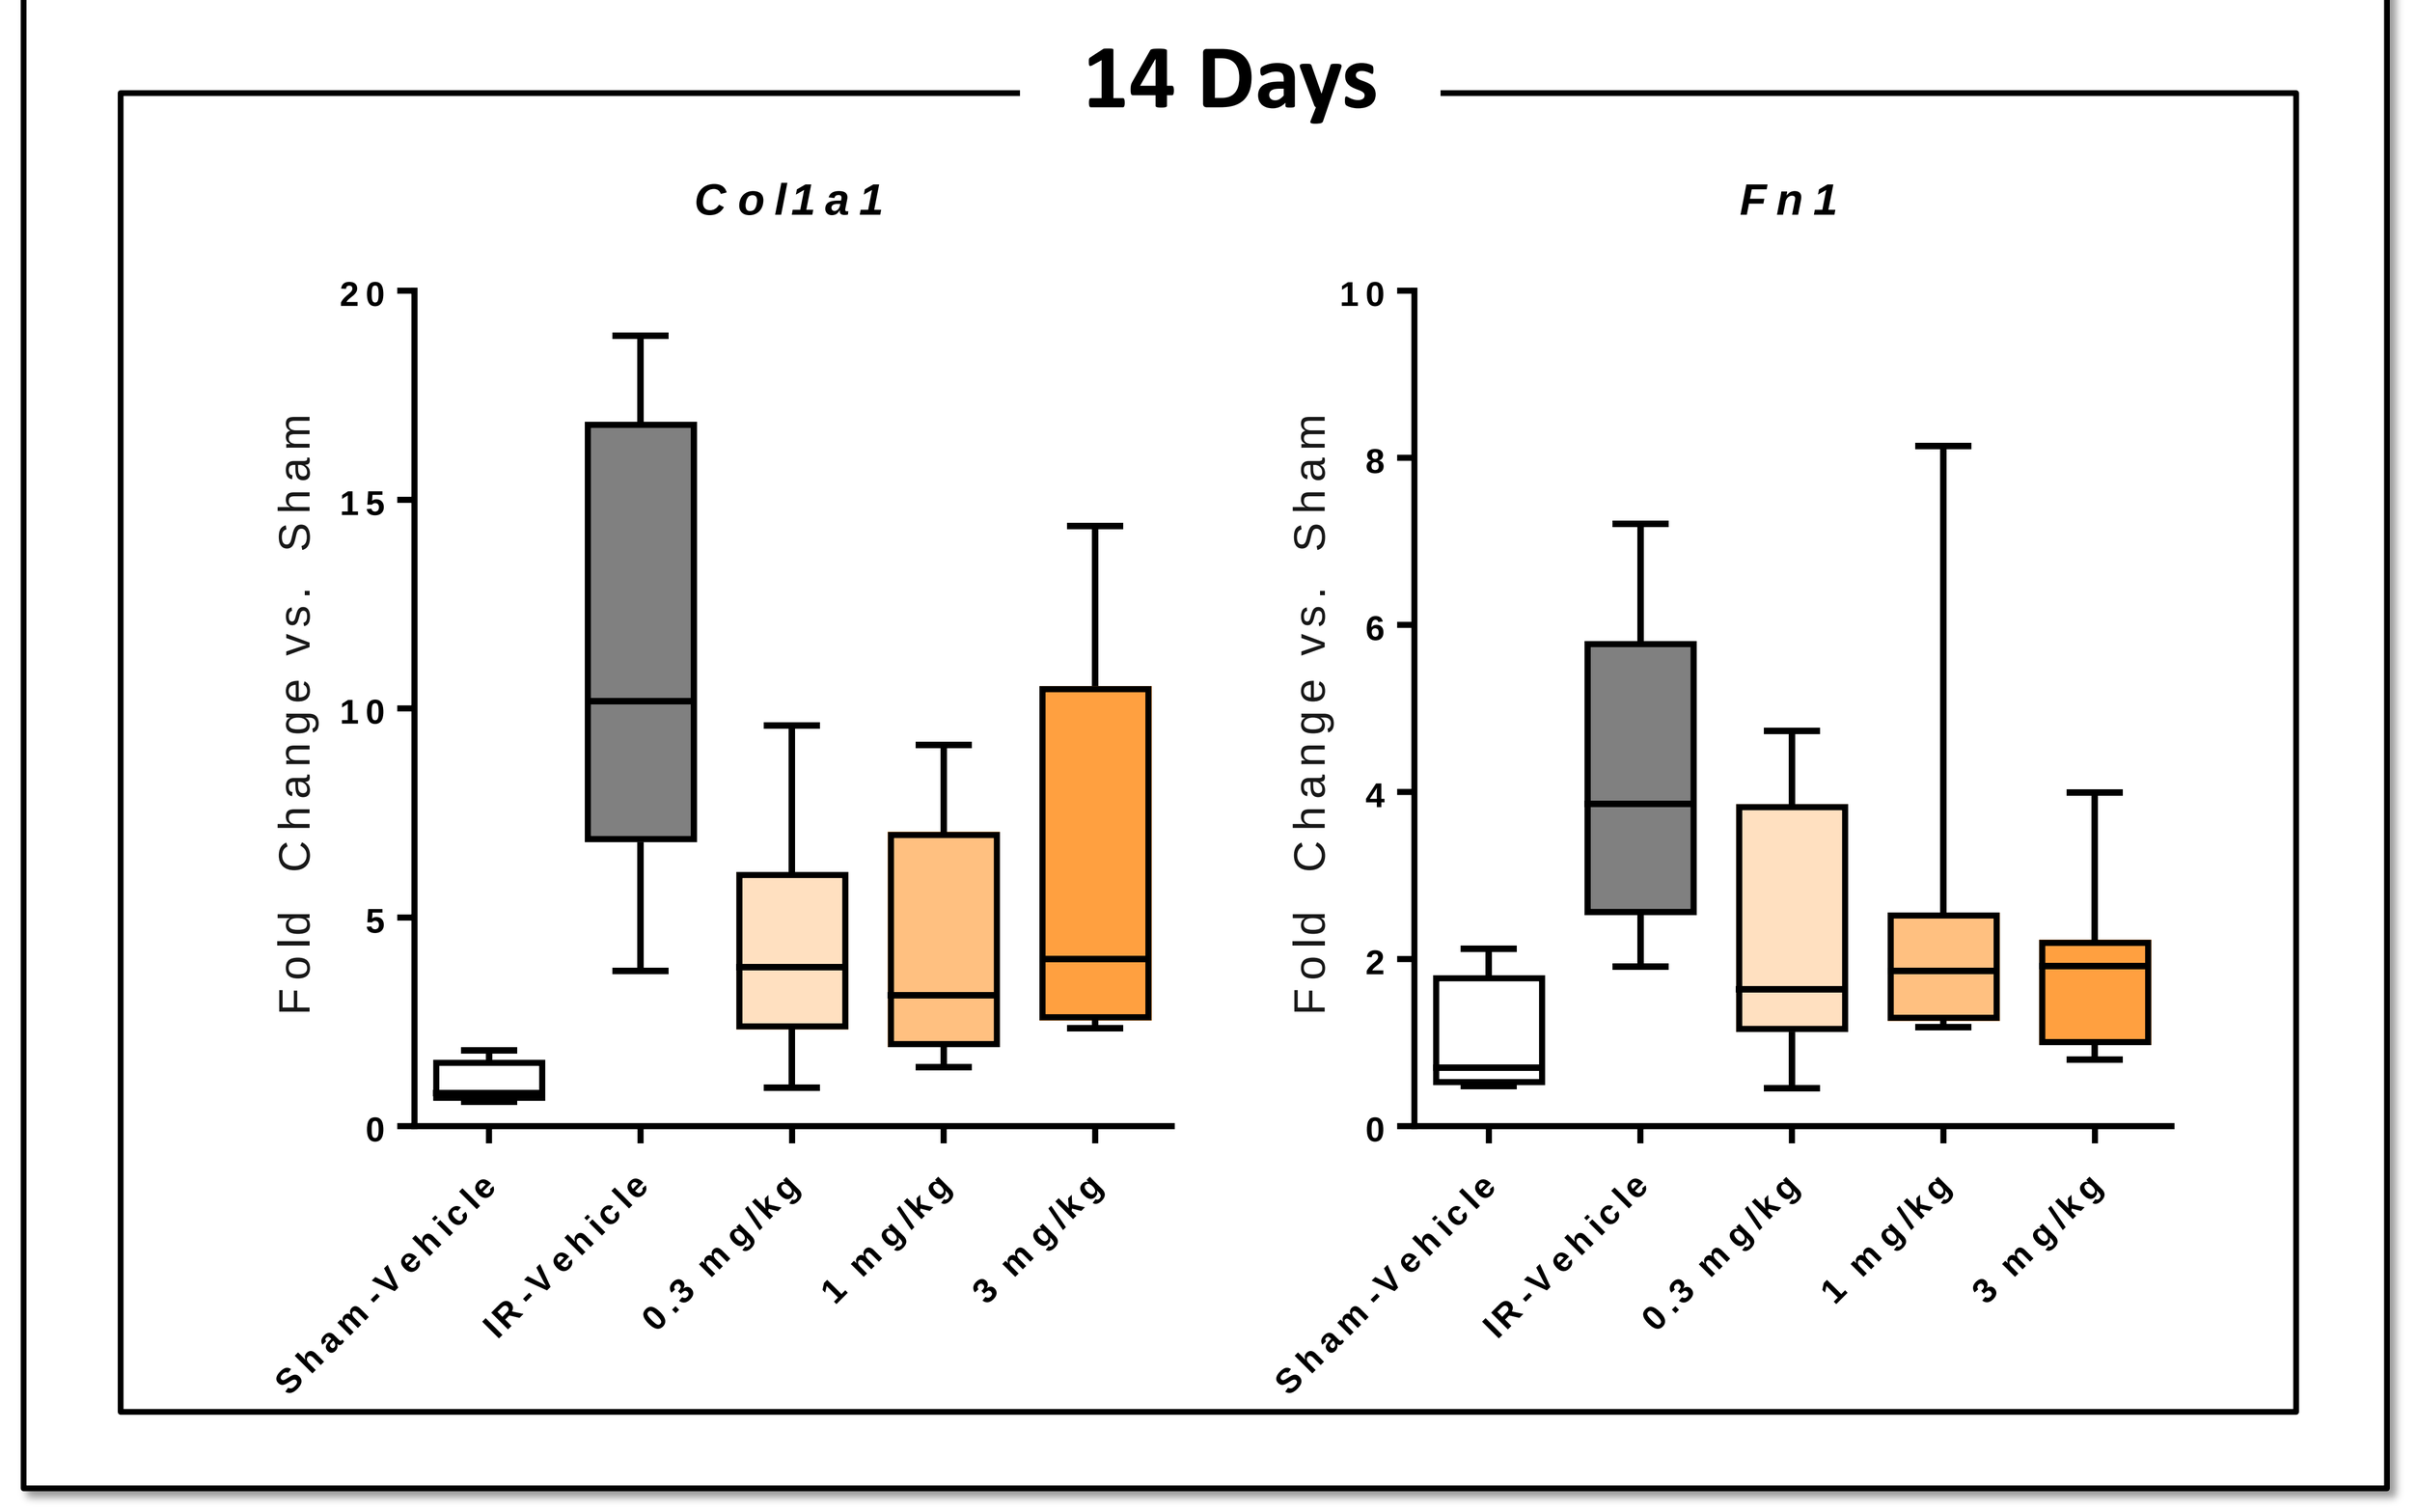
14 Days
Col1a1
Fn1
Fold Change vs. Sham
Sham-Vehicle
IR-Vehicle
0.3 mg/kg
1 mg/kg
3 mg/kg

Conclusions and future directions

Selective PPARd modulation by MTB-2after AKI in rats recovered renal function through preservation of proximal tubular and mitochondrial gene expression and reduction of kidney injury and profibrotic genes. In preclinical safety studies MTB-2 was shown to be safe and well tolerated. The next step will be to test this therapeutic in clinical trials. Developing clinical plan in partnership with Astellas Pharma

mitobridge